THERAPEUTIC BIOMARKER DISCOVERY IN IDIOPATHIC PULMONARY FIBROSIS (IPF) THROUGH PROTEOMIC ANALYSIS OF PRECISION-CUT LUNG SLICE (PCLS) SUPERNATANTS

An M,[1] Ho S,[1] Rao V,[1] Wolters P,[2] Turner S,[1] Schaub J,[1] Decaris M[1]
[1]Pliant Therapeutics, Inc., South San Francisco, CA, USA; [2]Department of Medicine, University of California, San Francisco, CA, USA
Research supported by Pliant Therapeutics, Inc.
Authors' relevant interests: MA, SH, VR, ST, JS, and MD are employed by Pliant Therapeutics, Inc., and own stock. PW was provided with a fee for their service by Pliant Therapeutics, Inc.
Poster no. P1012

RATIONALE

Idiopathic pulmonary fibrosis is a rare fibrotic disease that progressively destroys both lung structure and function and has a poor prognosis.[1,2] Based on conservative estimates from Europe and North America, the estimate incidence is 3–9 cases per 100,000 per year.[3] Identification of therapeutic biomarkers of idiopathic pulmonary fibrosis, which can better predict long-term therapeutic outcomes, is hampered by a lack of translational preclinical models. Precision-cut lung slices generated from explanted lung tissue from patients with idiopathic pulmonary fibrosis offer a physiologically relevant *ex vivo* model system to study fibrosis, preserving the diverse cellular composition and extracellular matrix architecture of fibrotic human lungs.[1,4] Using a highly sensitive proteomics platform, we characterized the proteins secreted by fibrotic precision-cut lung slices in the presence or absence of a transforming growth factor-beta type I receptor kinase inhibitor (ALK5i), as proof-of-concept for biomarker identification in this model system.

PRECISION-CUT LUNG SLICE CULTURES PROVIDE A UNIQUE PLATFORM FOR THE DISCOVERY AND EVALUATION OF TREATMENT-SPECIFIC TISSUE-SECRETED BIOMARKERS

Background: Precision-cut lung slice culture methodology

- Precision-cut lung slices generated from idiopathic pulmonary fibrosis lung explant tissue can be cultured *ex vivo* for 7 days
- Replicate slices can be treated with low-molecular-weight inhibitors to evaluate treatment-mediated effects on protein or messenger ribonucleic acid expression

Figure 1. Schematic diagram showing the procedure to generate precision-cut lung slices

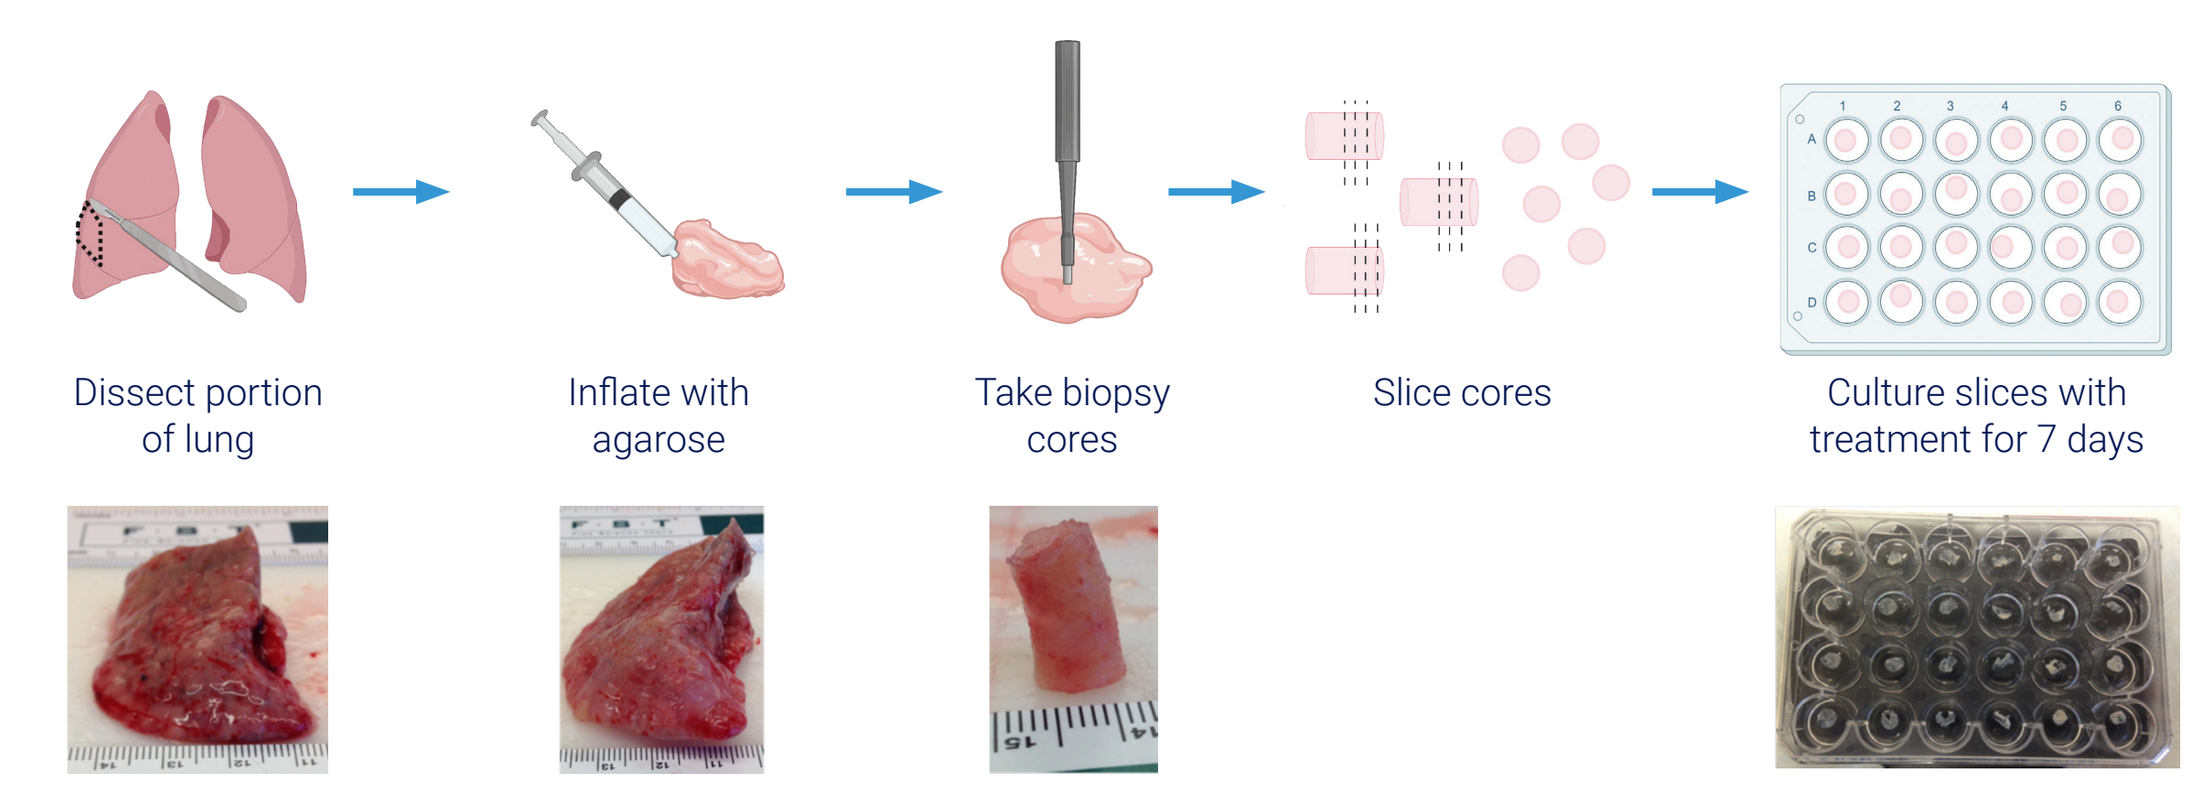
Dissect portion of lung
Inflate with agarose
Take biopsy cores
Slice cores
Culture slices with treatment for 7 days

Experimental design

- Precision-cut lung slices, generated from explanted lung tissue from 5 patients with idiopathic pulmonary fibrosis at the time of transplant, were cultured to assess the impact of transforming growth factor-beta pathway inhibition on secreted proteins
- Precision-cut lung slices were cultured for 7 days with continuous treatment of either 0.1% dimethylsulfoxide as a vehicle or ALK5i (R-268712; 1 μM), a transforming growth factor-beta type I receptor kinase inhibitor. Medium was changed daily
- Hyper reaction monitoring deep proteomic profiling by liquid chromatography–tandem mass spectrometry was performed on pooled conditioned media collected on the last day of culture (Day 7) from the precision-cut lung slices (n=6 pooled wells per donor/treatment)

Figure 2. Proteomic analysis study design

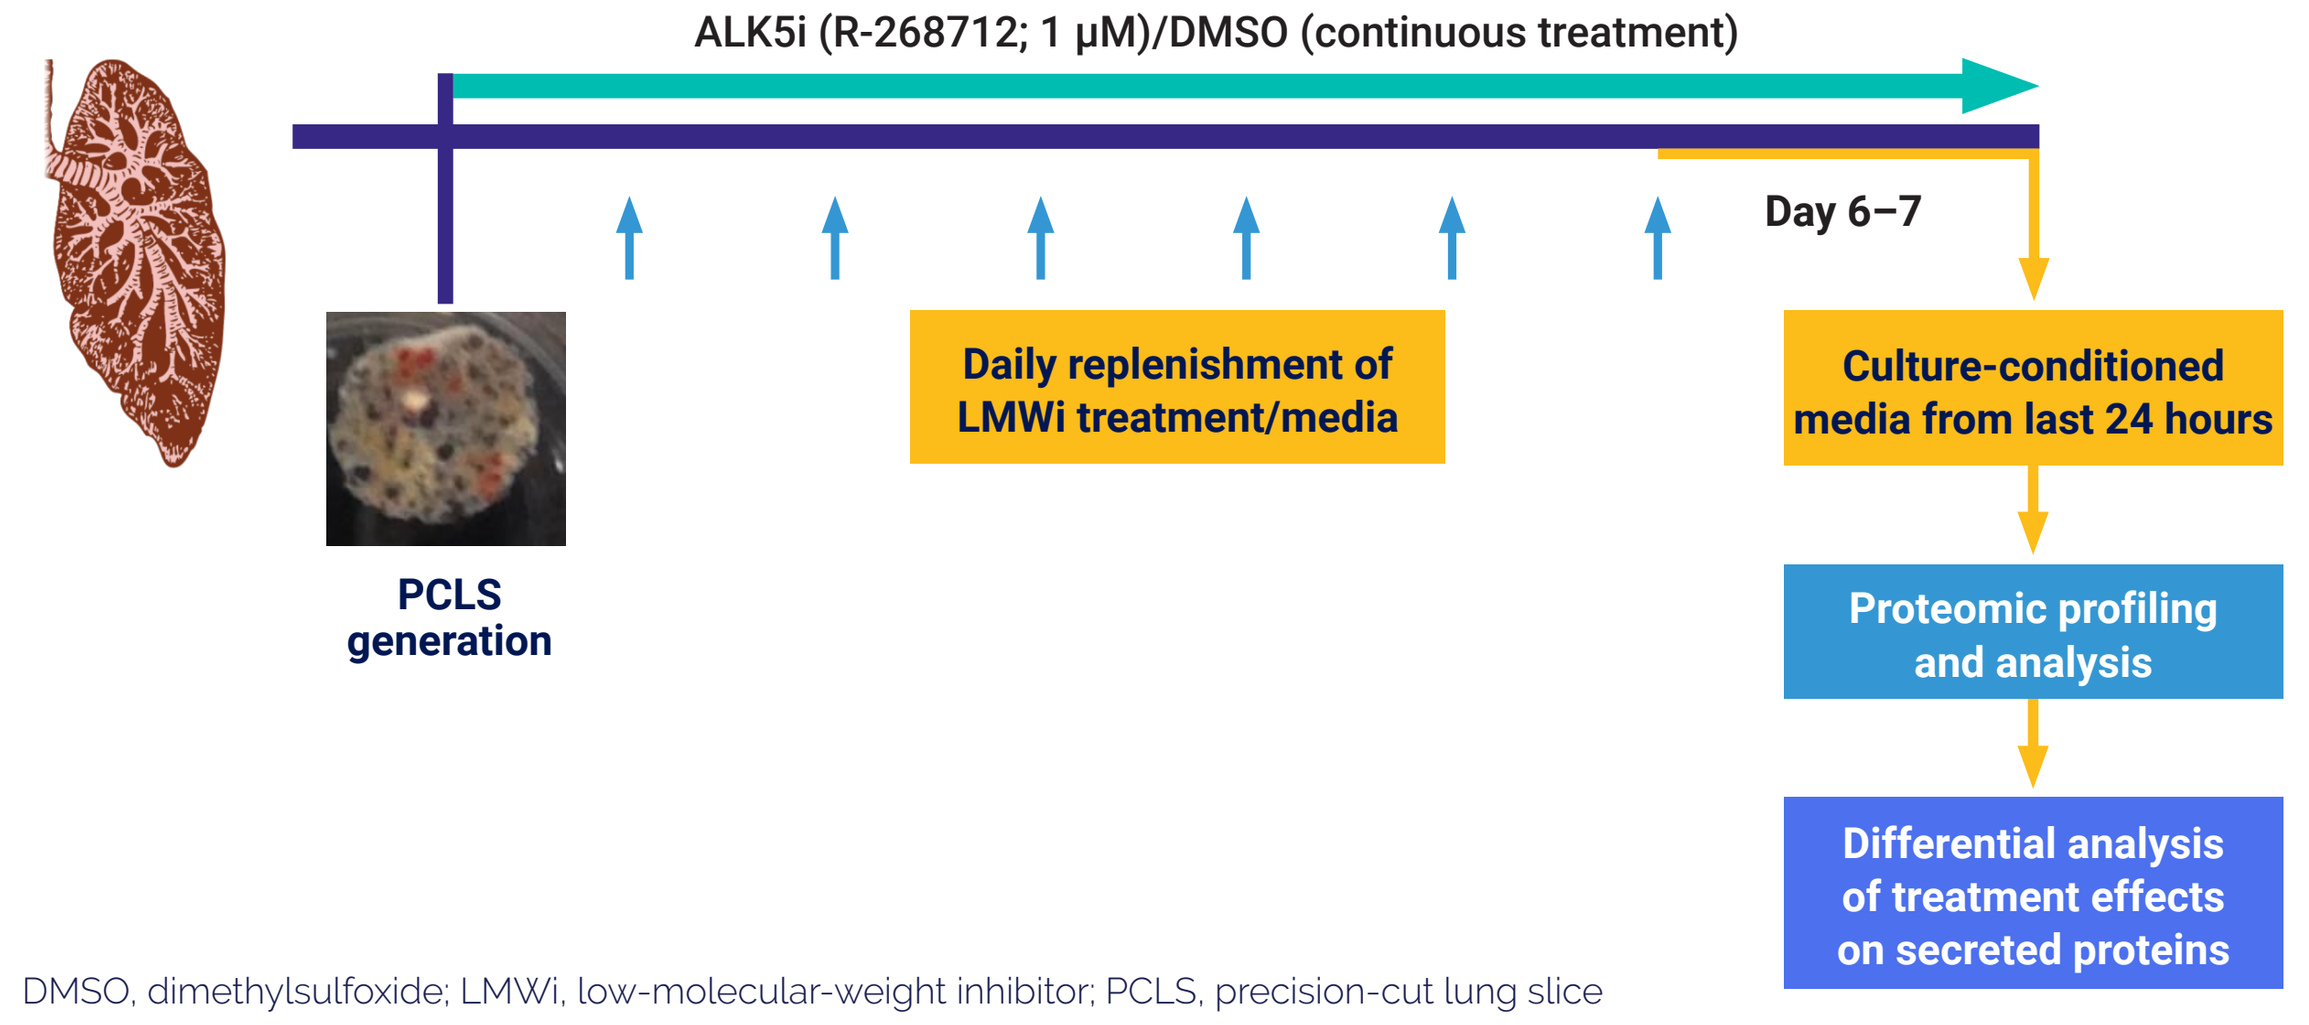
ALK5i (R-268712; 1 μM)/DMSO (continuous treatment)
Day 6–7
PCLS generation
Daily replenishment of LMWi treatment/media
Culture-conditioned media from last 24 hours
Proteomic profiling and analysis
Differential analysis of treatment effects on secreted proteins

DMSO, dimethylsulfoxide; LMWi, low-molecular-weight inhibitor; PCLS, precision-cut lung slice

SUMMARY OF PRECISION-CUT LUNG SLICE DONOR TISSUES

Donor sample summary

Table 1. Characteristics of patients with idiopathic pulmonary fibrosis who donated lung tissue

Sample ID	Diagnosis	Previous patient treatment	Sex	Age
19-H-4	IPF	Nintedanib	M	58
19-H-6	IPF	Nintedanib	M	61
19-H-11	IPF (familial)	Pirfenidone	F	57
19-H-12	IPF	–	M	60
19-H-13	IPF	Nintedanib	M	62

F, female; ID, identification; IPF, idiopathic pulmonary fibrosis; M, male

Figure 3. Picrosirius red staining on adjacent fixed tissue from each donor

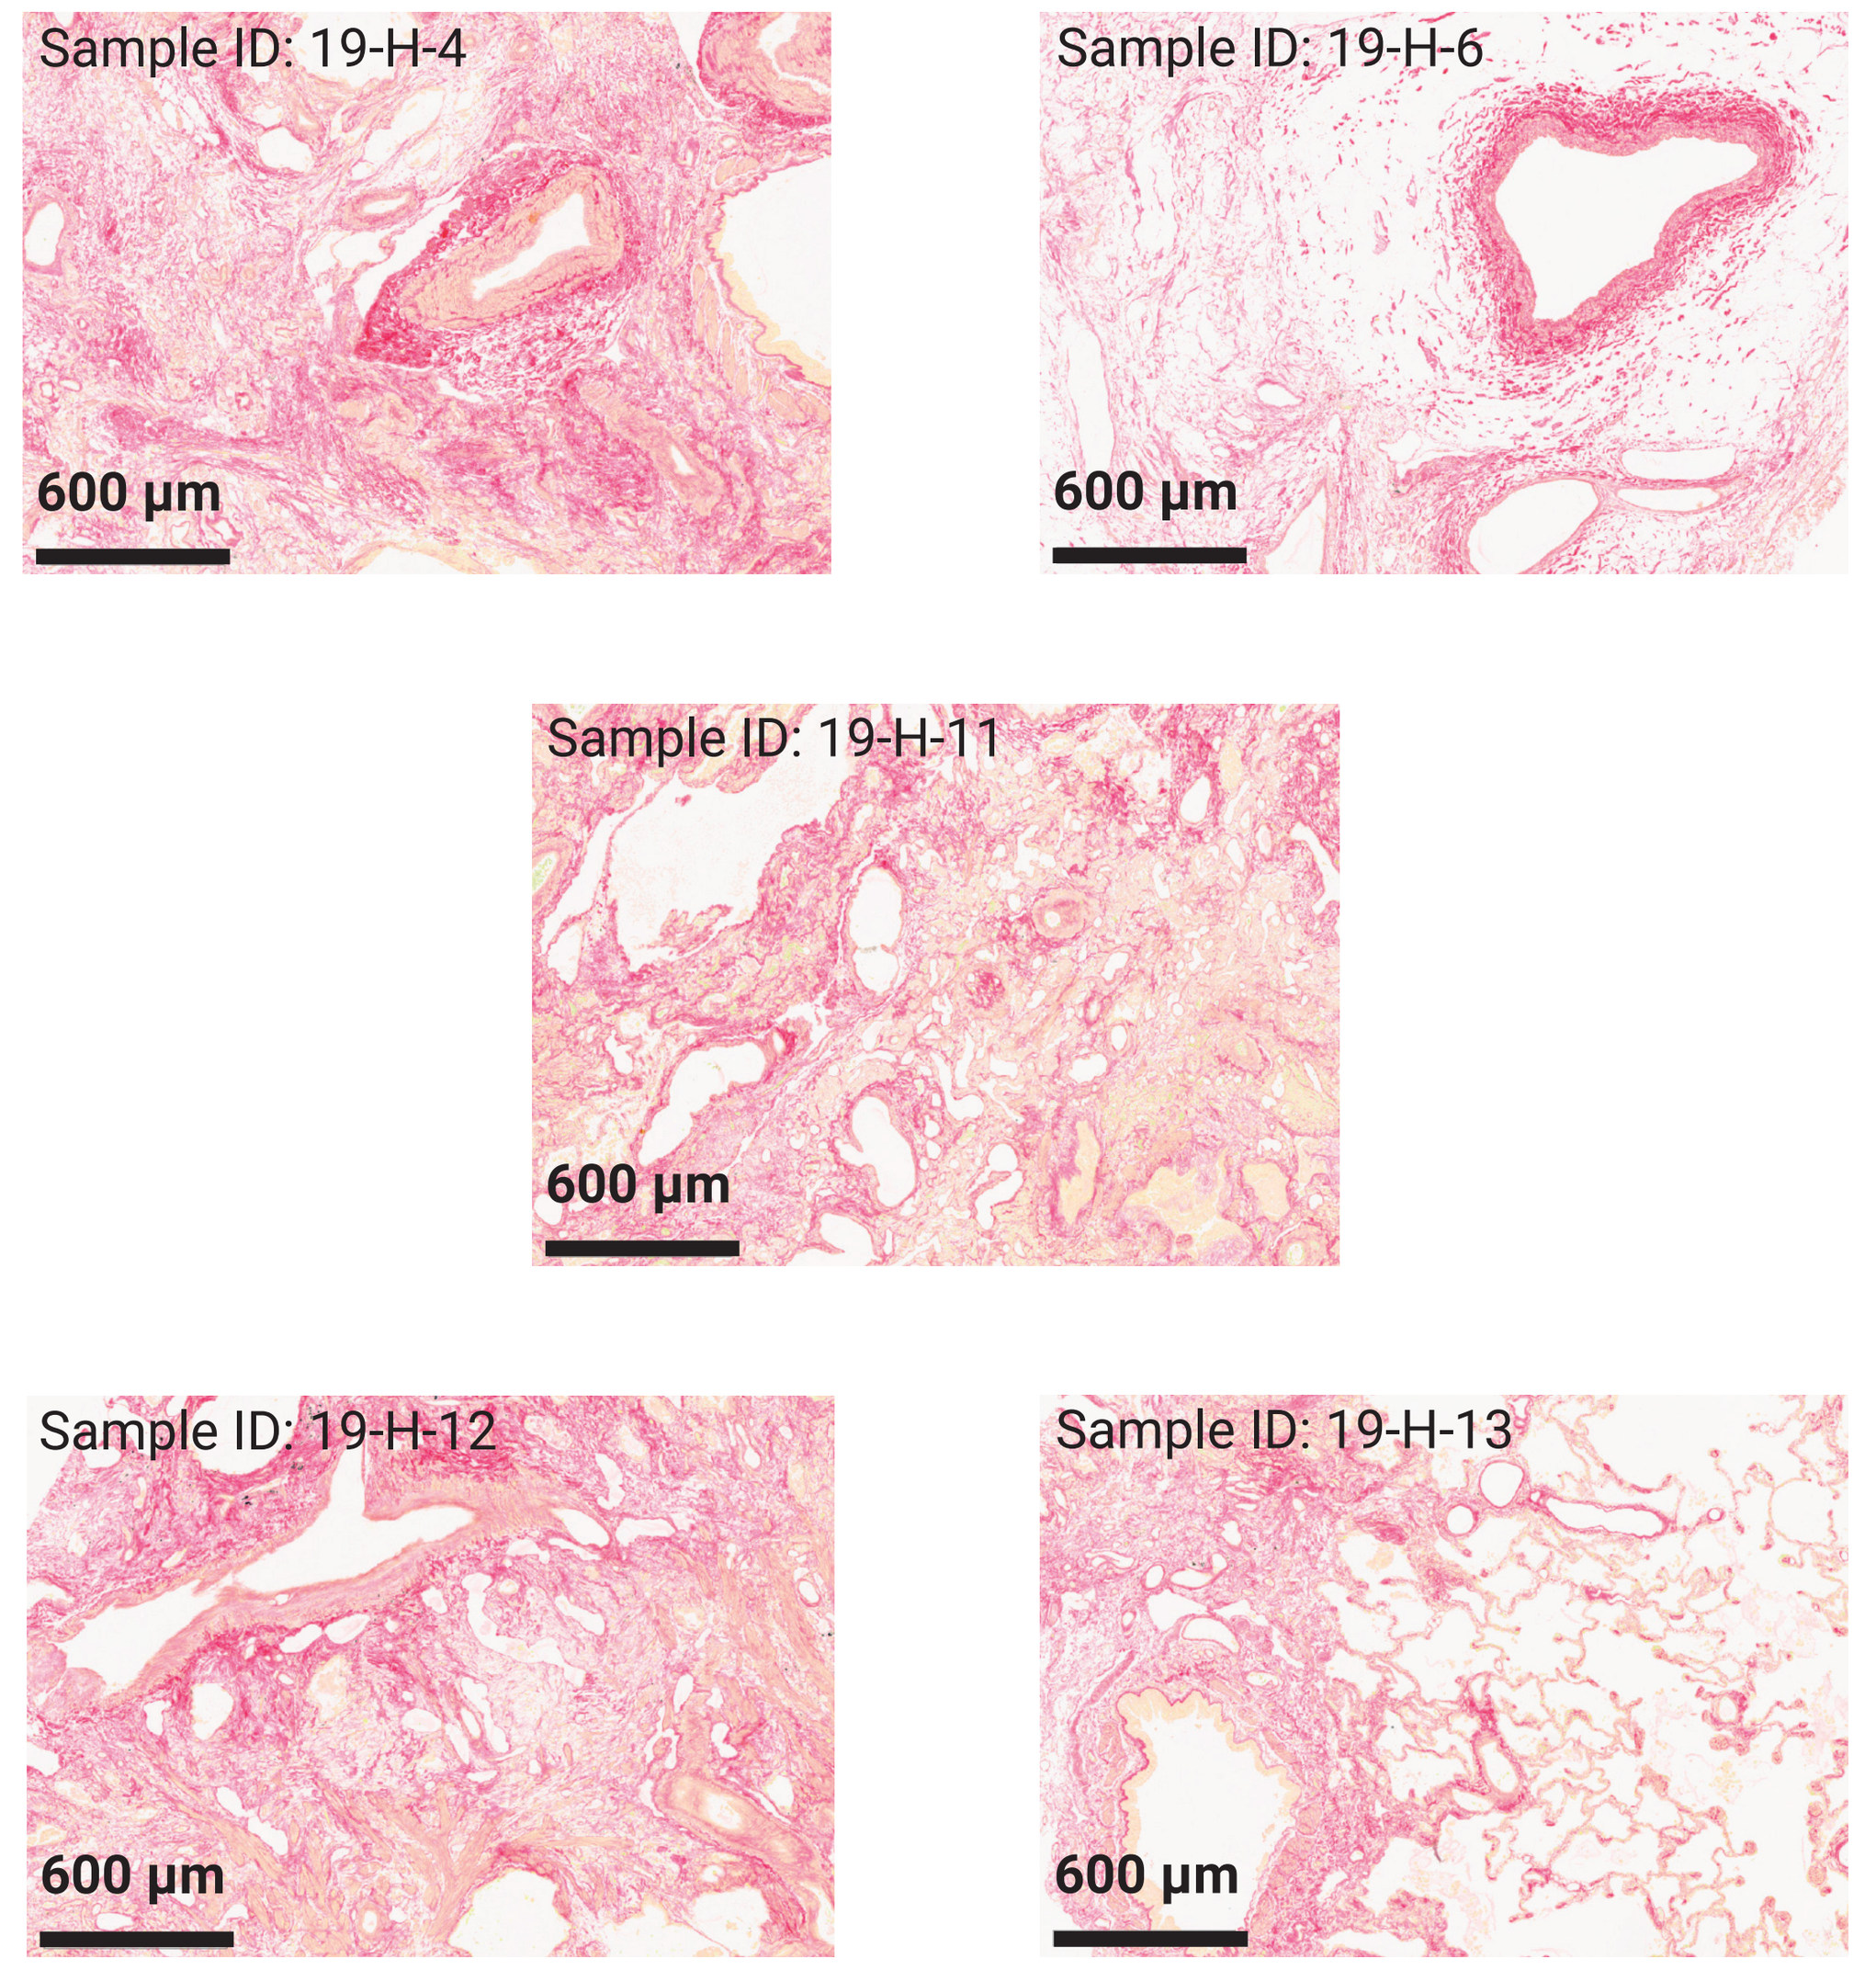
Sample ID: 19-H-4
600 μm
Sample ID: 19-H-6
600 μm
Sample ID: 19-H-11
600 μm
Sample ID: 19-H-12
600 μm
Sample ID: 19-H-13
600 μm

ID, identification

99 PROTEINS WERE EXPRESSED AT SIGNIFICANTLY DIFFERENT LEVELS IN ALK5i-TREATED VS. VEHICLE-TREATED SAMPLES

Quantification of proteins from precision-cut lung slice conditioned media samples by proteomic analysis

- In total, 3347 proteins represented by ~37,000 unique peptides were quantified by hyper reaction monitoring across all precision-cut lung slice conditioned media samples
- On average, there were 2605 proteins per sample; the number of proteins was relatively uniform across samples
- Samples were protein-normalized; subsequent differential analysis was reflective of the relative protein amount across samples, not absolute protein amount

Differential analysis of protein abundance

- Differential protein abundance was calculated for ALK5i-treated vs. vehicle-treated samples
- In total, 99 proteins were expressed at significantly different levels in ALK5i- treated vs. vehicle-treated samples (p<0.05), with an average $\log_2$ fold change >1.5
- The volcano plot shows all proteins, plotted by $\log_2$ fold change and p value for ALK5i-treated vs. vehicle-treated samples. The 99 differential proteins are highlighted in red, with the subset of genes identified by name

Figure 4. Volcano plot showing candidate proteins with differential abundance in conditioned media samples of ALK5i-treated precision-cut lung slices

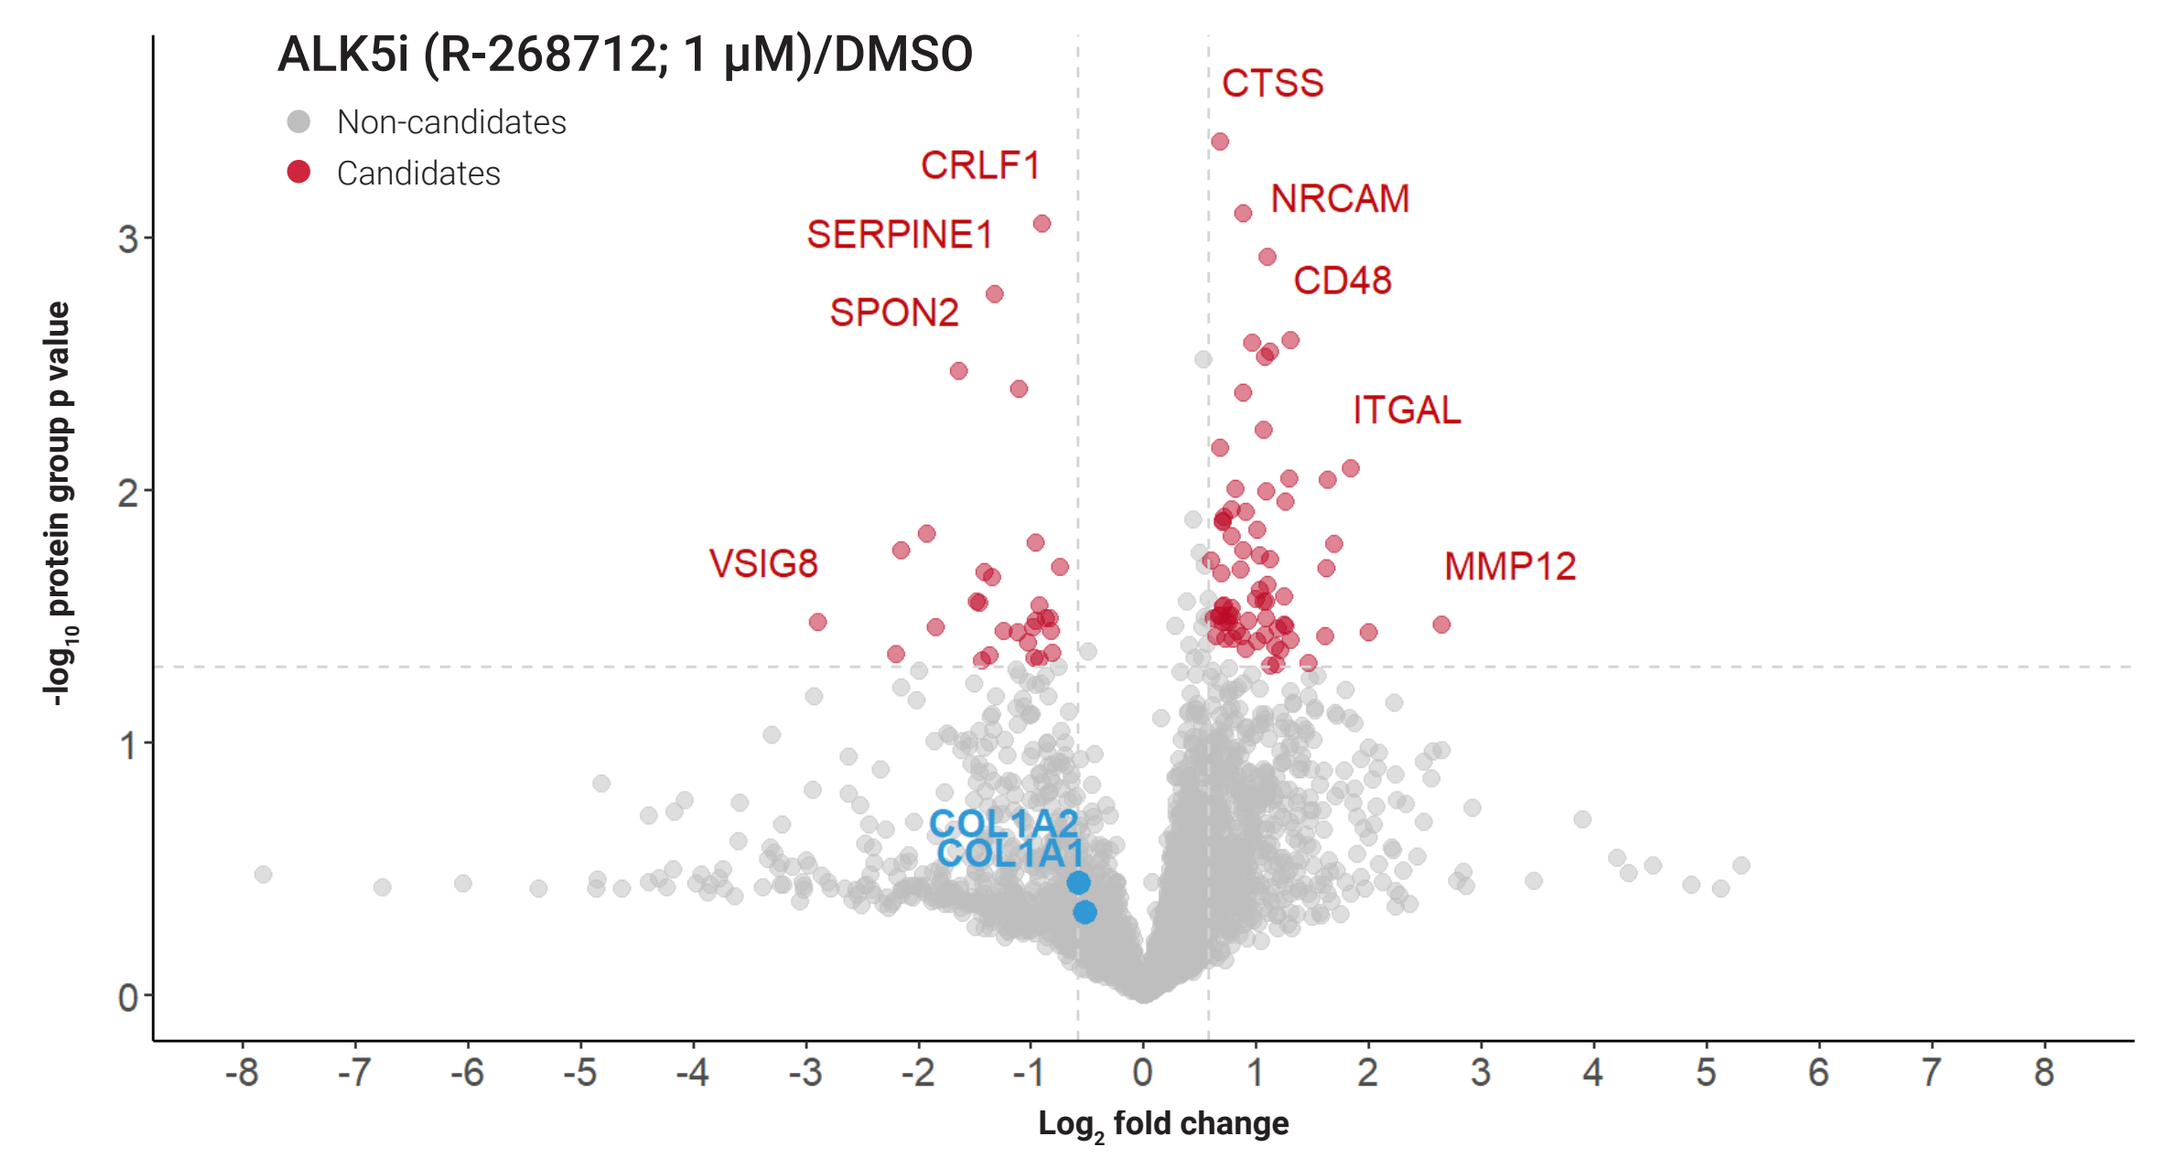
ALK5i (R-268712; 1 μM)/DMSO
Non-candidates
Candidates
CTSS
CRLF1
NRCAM
SERPINE1
CD48
SPON2
ITGAL
VSIG8
MMP12
COL1A2
COL1A1
$-\log_{10}$ protein group p value
$\log_2$ fold change

CD, cluster of differentiation; COL1A1, collagen type I alpha 1 chain; COL1A2, collagen type I alpha 2 chain; CRLF1, cytokine-like factor 1; CTSS, cathepsin S; DMSO, dimethylsulfoxide; ITGAL, integrin subunit alpha L; MMP12, matrix metalloproteinase-12; NRCAM, neuronal cell adhesion molecule; SERPINE1, serpin family E member 1; SPON2, spondin 2; VSIG8, V-set and immunoglobulin domain containing 8

Table 2. Summary of top proteins in precision-cut lung slice conditioned media samples significantly downregulated by ALK5i treatment

Gene ID	Protein description	Average log ratio	Protein p value
Downregulated by treatment			
VSIG8	V-set and immunoglobulin domain-containing protein 8	-2.898	3.34E-02
COL10A1	Collagen type X alpha 1 chain	-2.197	4.44E-02
LTBP2	Latent-transforming growth factor-beta-binding protein 2	-2.151	1.73E-02
FAP	Prolyl endopeptidase FAP	-1.926	1.49E-02
SPOCK1	Testican-1	-1.846	3.48E-02
SPON2	Spondin-2	-1.646	3.37E-03
ANGPTL4	Angiopoietin-like protein 4	-1.481	2.76E-02
ADAMTS7	A disintegrin and metalloproteinase with thrombospondin motifs 7	-1.461	2.78E-02
PLOD2	Procollagen-lysine, 2-oxoglutarate 5-dioxygenase 2	-1.434	4.73E-02
COL11A2	Collagen type XI alpha 2 chain	-1.416	2.11E-02
COL7A1	Collagen type VII alpha 1 chain	-1.368	4.50E-02
LOXL1	Lysyl oxidase homolog 1	-1.343	2.20E-02
SERPINE1	Plasminogen activator inhibitor-1	-1.328	1.67E-03
PRSS23	Serine protease 23	-1.240	3.61E-02
APOH	Beta-2-glycoprotein 1	-1.125	3.65E-02
OLFML2B	Olfactomedin-like protein 2B	-1.106	3.96E-03
DDX21	Nucleolar RNA helicase 2	-1.026	4.01E-02
LAMA-1	Laminin subunit alpha-1	-0.983	3.49E-02
GAP43	Neuromodulin	-0.977	4.64E-02
MRC2	C-type mannose receptor 2	-0.965	1.61E-02
COL5A2	Collagen type V alpha 2 chain	-0.955	3.28E-02
ROCK2	Rho-associated protein kinase 2	-0.923	4.65E-02
BGN	Biglycan	-0.920	2.87E-02
CRLF1	Cytokine receptor-like factor 1	-0.908	8.86E-04
PXDN	Peroxidasin homolog	-0.867	3.21E-02
ANTXR1	Anthrax toxin receptor 1	-0.833	3.22E-02
PLXDC2	Plexin domain-containing protein 2	-0.819	3.63E-02
POSTN	Periostin	-0.815	4.41E-02
GSTM3	Glutathione S-transferase Mu 3	-0.739	2.02E-02

FAP, fibroblast activation protein; ID, identification; RNA, ribonucleic acid

Table 3. Summary of top proteins in precision-cut lung slice conditioned media samples significantly upregulated by ALK5i treatment

Gene ID	Protein description	Average log ratio	Protein p value
Upregulated by treatment			
MMP12	Macrophage metalloelastase	2.647	3.40E-02
TNFSF13B	Tumor necrosis factor ligand superfamily member 13B	1.998	3.67E-02
ITGAL	Integrin alpha-L	1.841	8.18E-03
NUDT9	ADP-ribose pyrophosphatase, mitochondrial	1.685	1.64E-02
PTX3	Pentraxin-related protein	1.630	9.11E-03
SECTM1	Secreted and transmembrane protein 1	1.618	2.05E-02
AMY1A	Alpha-amylase 1	1.613	3.76E-02
LIPA	Lysosomal acid lipase/cholesteryl ester hydrolase	1.464	4.85E-02
CD48	CD48 antigen	1.304	2.54E-03
IFITM3	Interferon-induced transmembrane protein 3	1.302	3.90E-02
VPS36	Vacuolar protein-sorting-associated protein 36	1.297	9.03E-03
SLC34A2	Sodium-dependent phosphate transport protein 2B	1.260	1.10E-02
C1QA	Complement C1q subcomponent subunit A	1.259	3.46E-02
C1QC	Complement C1q subcomponent subunit C	1.243	2.65E-02
PCSK9	Proprotein convertase subtilisin/kexin type 9	1.242	3.41E-02
LRRN4	Leucine-rich repeat neuronal protein 4	1.217	4.29E-02
HLA-A	HLA class I histocompatibility antigen, A alpha chain	1.192	3.52E-02
GBP2	Guanylate-binding protein 2	1.180	4.88E-02
TGFB2	Transforming growth factor-beta-2 proprotein	1.168	4.15E-02
IST1	IST1 homolog	1.127	4.96E-02
RAB27B	Ras-related protein Rab-27B	1.118	1.87E-02
SMPDL3A	Acid sphingomyelinase-like phosphodiesterase 3a	1.116	2.85E-03
NRCAM	Neuronal cell adhesion molecule	1.099	1.20E-03
ITGB2	Integrin beta-2	1.094	2.37E-02
HLA-C	HLA class I histocompatibility antigen, C alpha chain	1.087	1.01E-02
FERMT3	Fermitin family homolog 3	1.086	3.22E-02
PTPN6	Tyrosine-protein phosphatase non-receptor type 6	1.085	2.76E-02
A2M	Alpha-2-macroglobulin	1.080	2.96E-03
SARG	Specifically androgen-regulated gene protein	1.079	3.75E-02
CD47	Leukocyte surface antigen CD47	1.066	2.75E-02

ADP, adenosine diphosphate; CD, cluster of differentiation; HLA, human leukocyte antigen; ID, identification

DOWNREGULATION OF COLLAGEN SUBTYPES IN CONDITIONED MEDIA SAMPLES OF ALK5i-TREATED PRECISION-CUT LUNG SLICES

- Differential abundance of collagen subtypes are indicated on a heat map of all proteins, sorted by $\log_2$ fold change in ALKi5-treated vs. vehicle-treated precision-cut lung slices
- Upon treatment, the majority of detectable collagen subtypes demonstrated a trend for relative downregulation

Figure 5. Heatmap of collagen subtypes sorted by $\log_2$ fold change in ALK5i-treated vs. vehicle-treated precision-cut lung slices

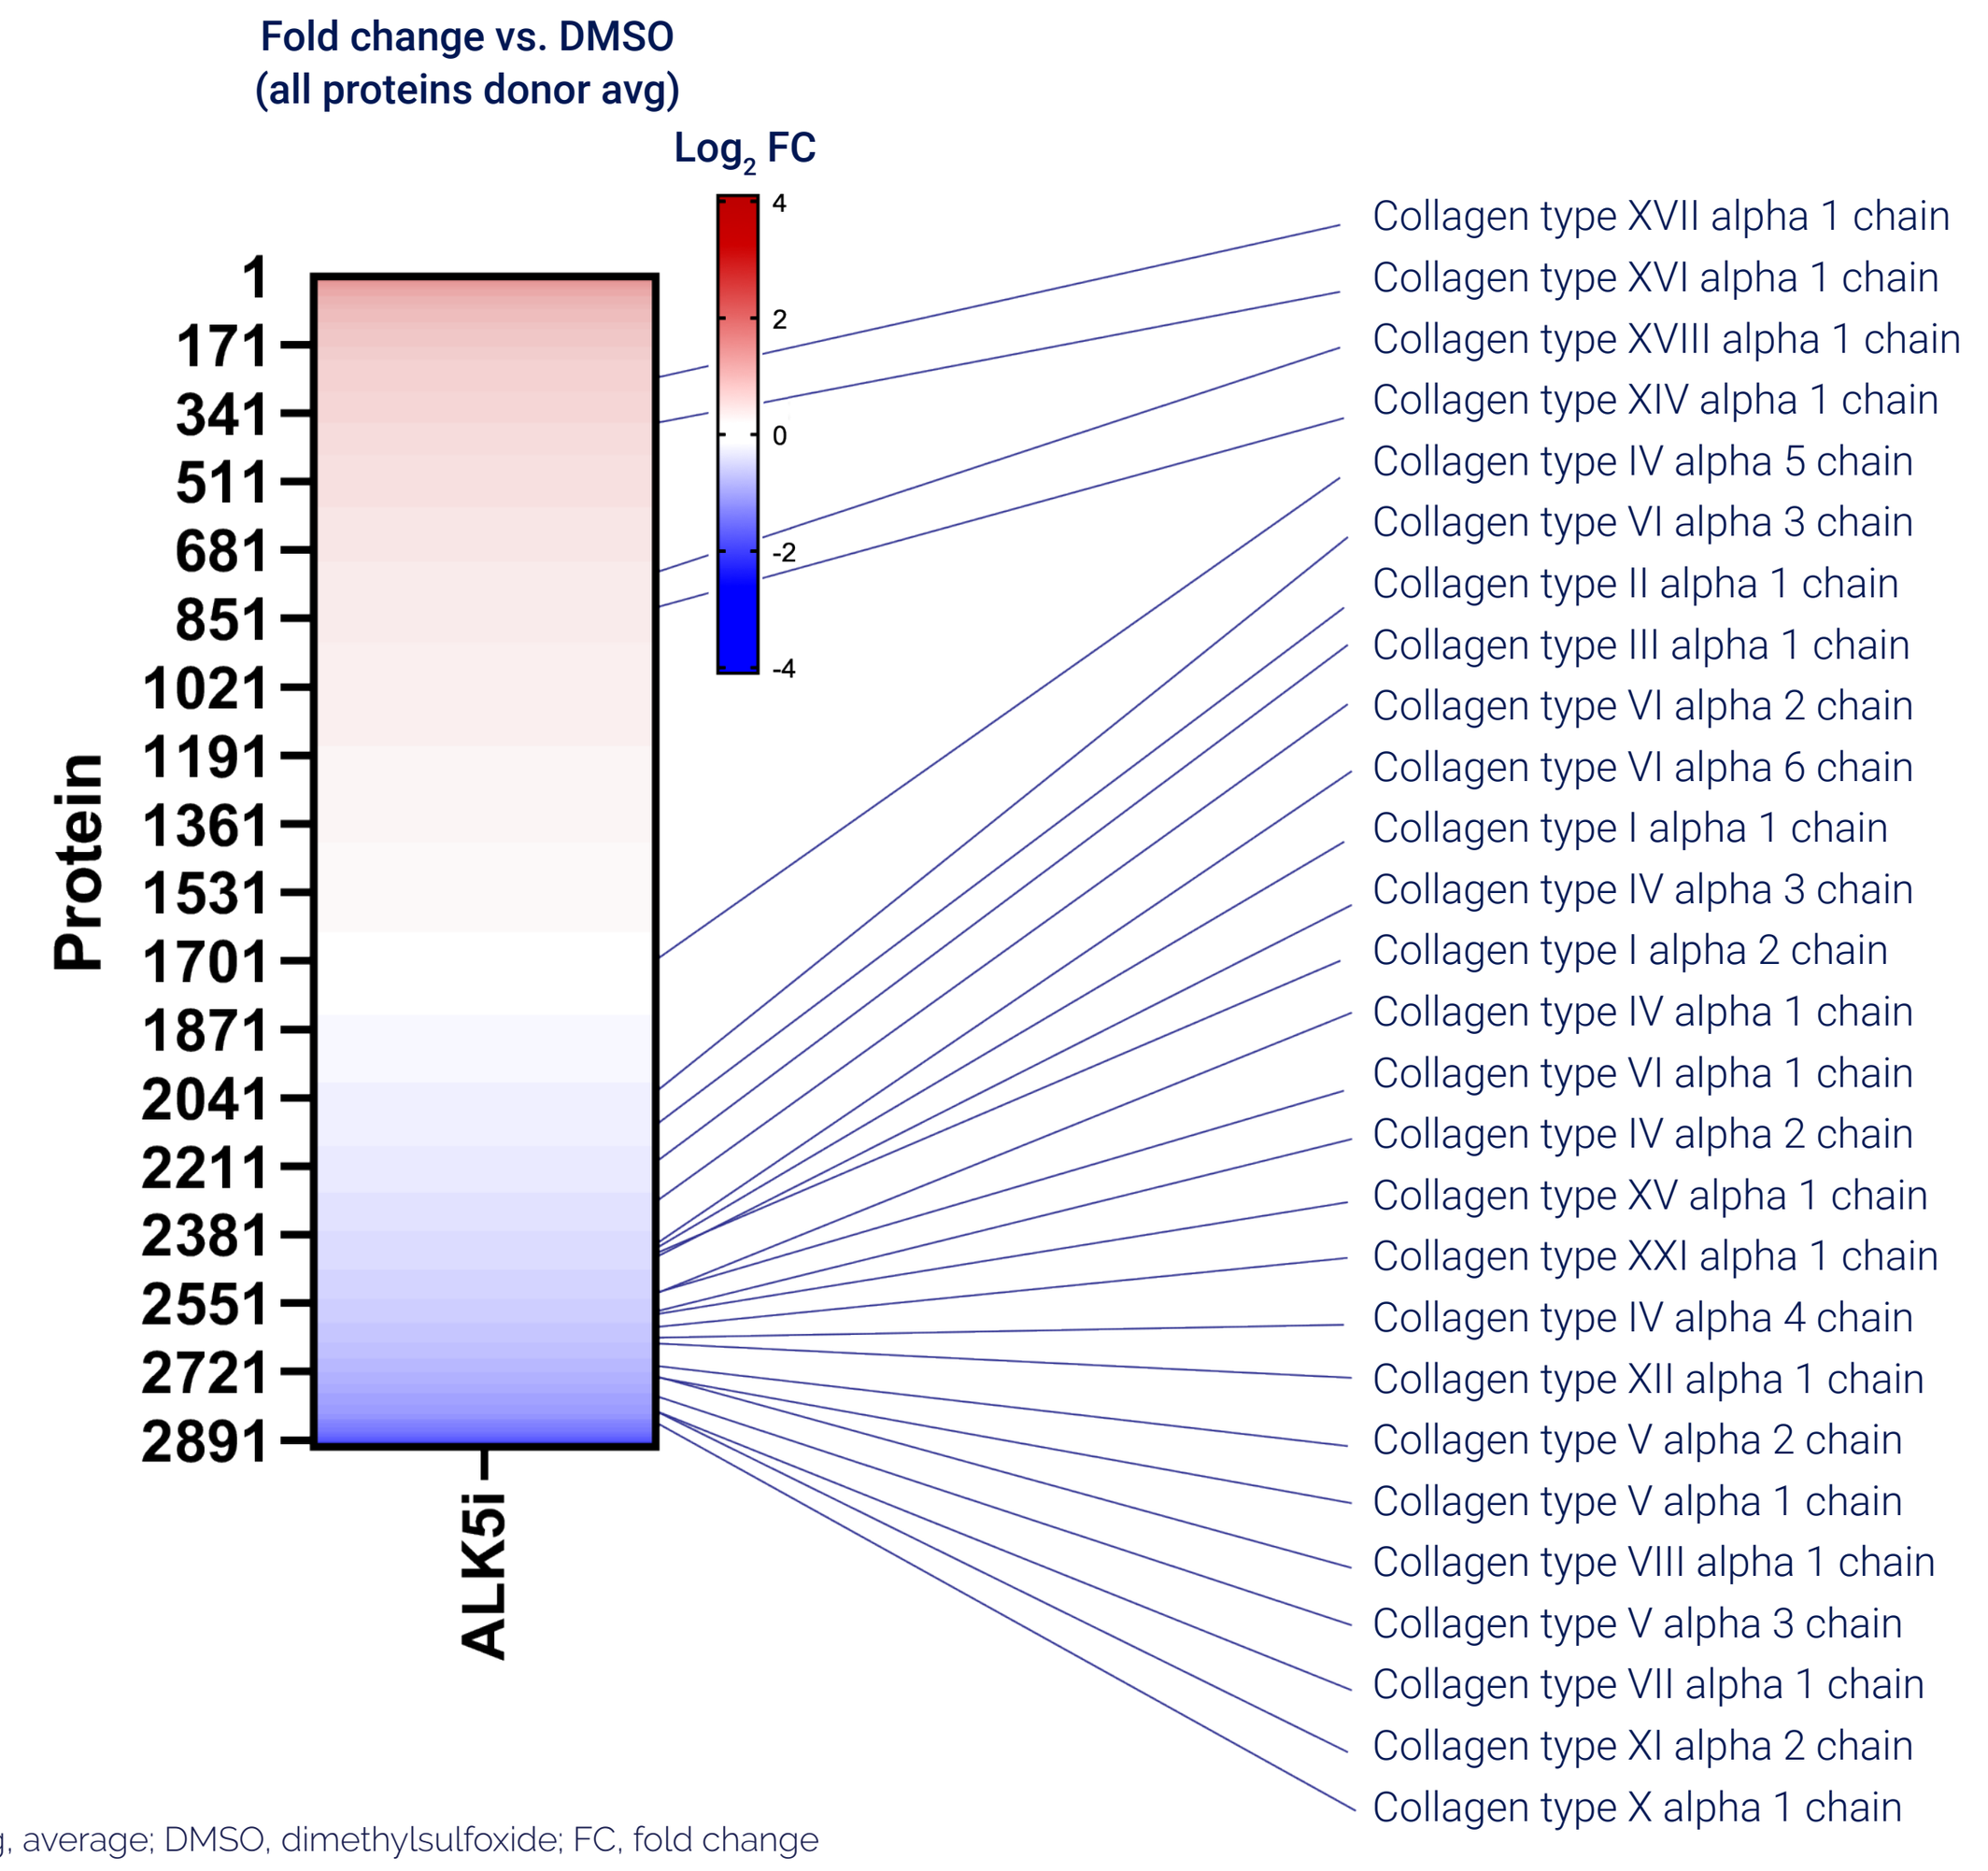
Fold change vs. DMSO
(all proteins donor avg)
$\log_2$ FC
Protein
ALK5i
Collagen type XVII alpha 1 chain
Collagen type XVI alpha 1 chain
Collagen type XVIII alpha 1 chain
Collagen type XIV alpha 1 chain
Collagen type IV alpha 5 chain
Collagen type VI alpha 3 chain
Collagen type II alpha 1 chain
Collagen type III alpha 1 chain
Collagen type VI alpha 2 chain
Collagen type VI alpha 6 chain
Collagen type I alpha 1 chain
Collagen type IV alpha 3 chain
Collagen type I alpha 2 chain
Collagen type IV alpha 1 chain
Collagen type VI alpha 1 chain
Collagen type IV alpha 2 chain
Collagen type XV alpha 1 chain
Collagen type XXI alpha 1 chain
Collagen type IV alpha 4 chain
Collagen type XII alpha 1 chain
Collagen type V alpha 2 chain
Collagen type V alpha 1 chain
Collagen type VIII alpha 1 chain
Collagen type V alpha 3 chain
Collagen type VII alpha 1 chain
Collagen type XI alpha 2 chain
Collagen type X alpha 1 chain

Avg, average; DMSO, dimethylsulfoxide; FC, fold change

MODULATION OF CLINICAL BIOMARKERS AFTER ALK5i TREATMENT OF PRECISION-CUT LUNG SLICES FROM IDIOPATHIC PULMONARY FIBROSIS LUNG EXPLANTS

- Multiple idiopathic pulmonary fibrosis clinical biomarkers were quantified using this approach in precision-cut lung slices conditioned media samples
- Plasminogen activator inhibitor-1 was significantly (p=0.0079) reduced in the ALK5i-treated vs. vehicle-treated samples, while periostin showed a strong trend toward downregulation (not significant, Mann–Whitney t-test)

Figure 6. Quantification of idiopathic pulmonary fibrosis biomarkers in precision-cut lung slices conditioned media samples

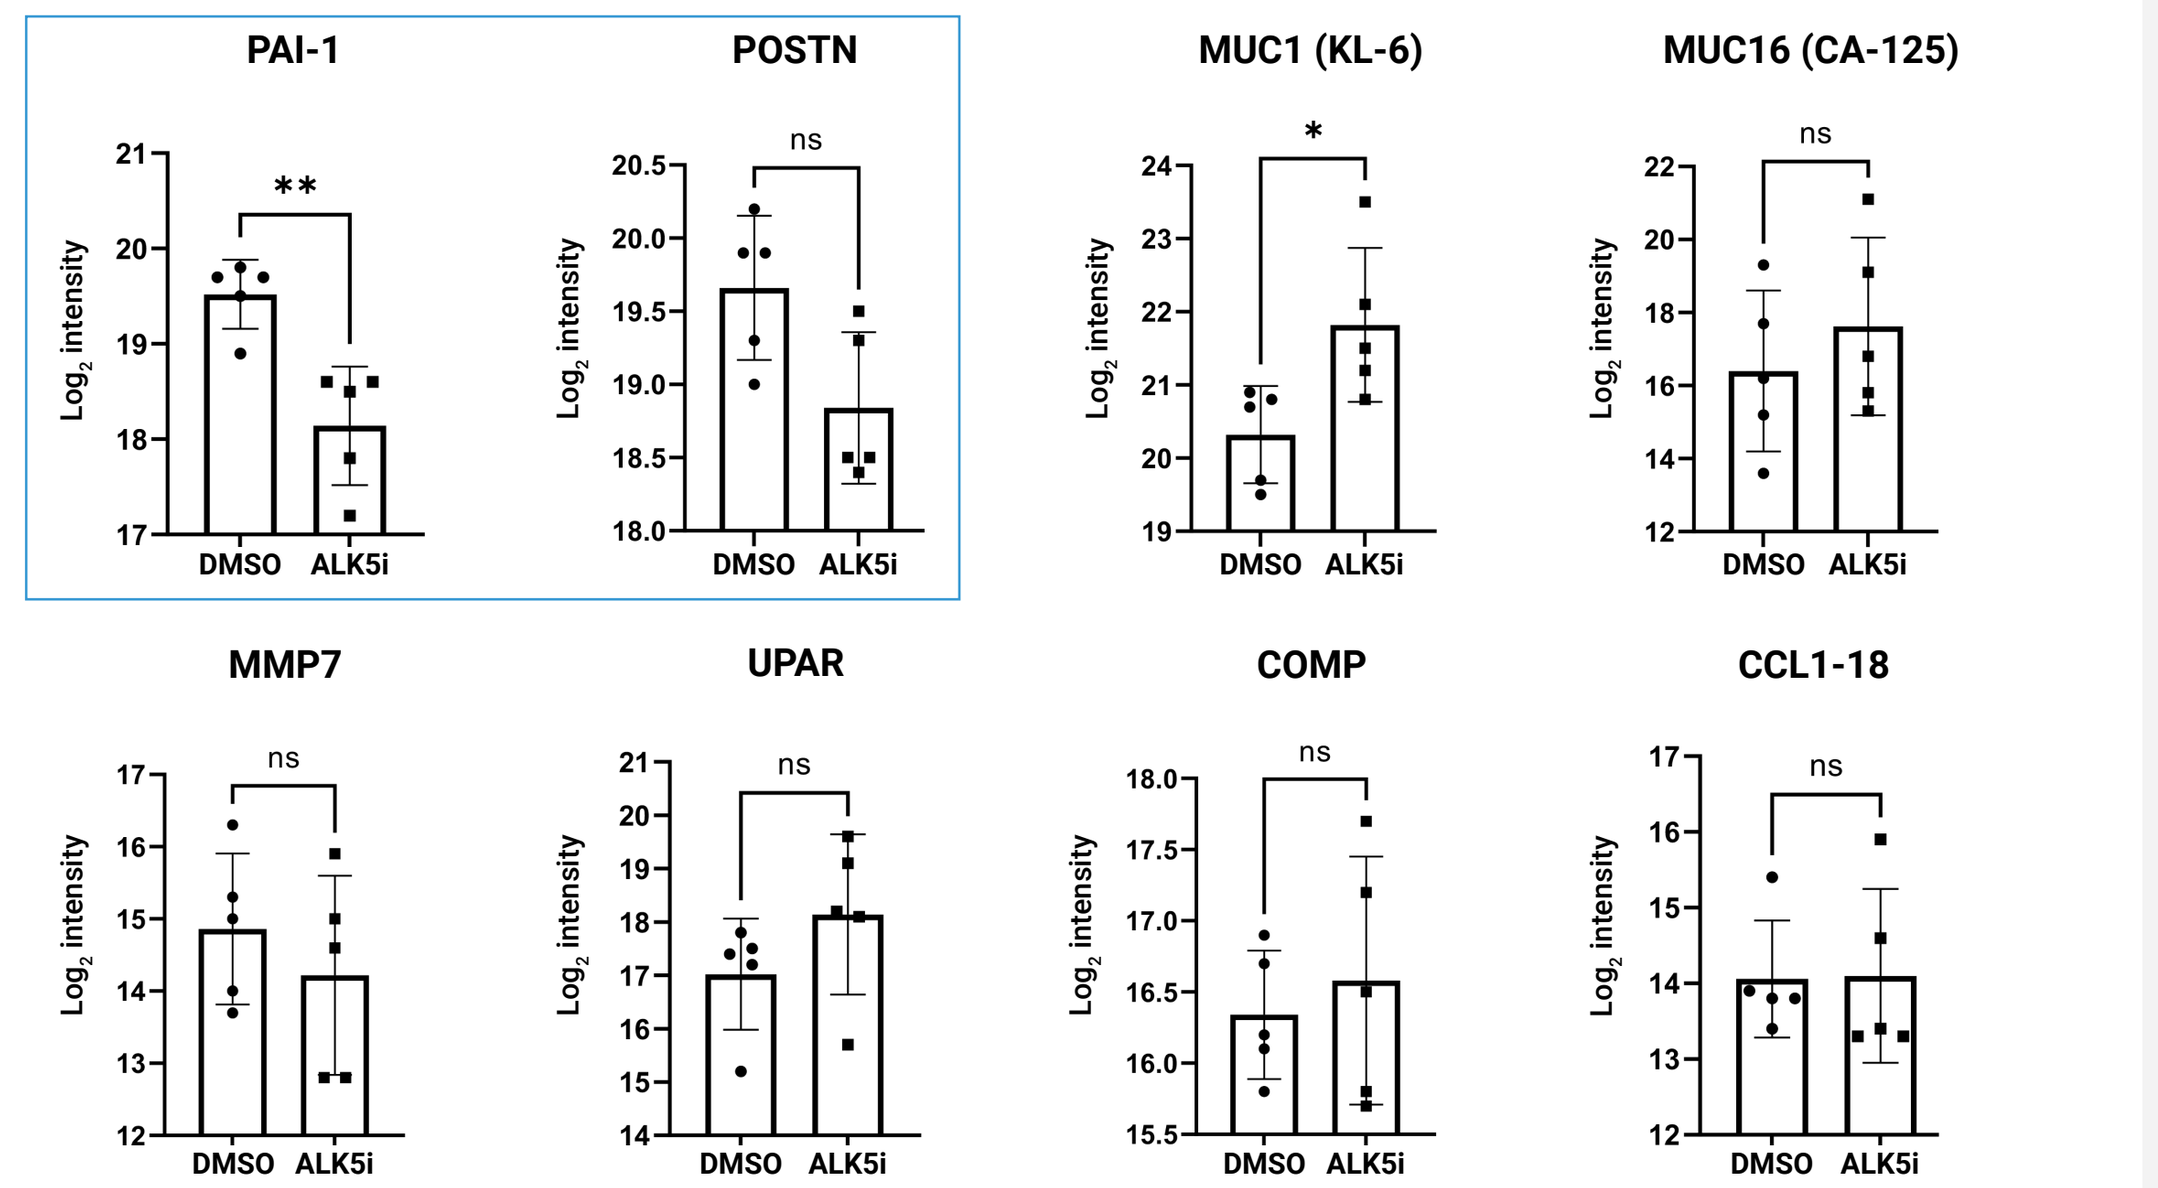
PAI-1
POSTN
MUC1 (KL-6)
MUC16 (CA-125)
MMP7
UPAR
COMP
CCL1-18
$\log_2$ intensity
DMSO
ALK5i
**
*
ns

*p<0.05; **p<0.01
Error bars represent standard deviation
CA, cancer antigen; CCL1-18, CC-chemokine ligand 18; COMP, cartilage oligomeric matrix protein; DMSO, dimethylsulfoxide; KL-6, Krebs von den Lungen-6; MMP7, matrix metalloproteinase-7; MUC1, mucin 1; MUC16, mucin 16; ns, not significant; PAI-1, p lasminogen activator inhibitor-1; POSTN, periostin; UPAR, urokinase-type plasminogen activator receptor

STABLE ISOTOPE LABELING EXPERIMENTS CONFIRMED THAT SECRETION OF NEWLY SYNTHESIZED PROTEINS OCCURRED DURING CULTURE

- To evaluate the presence of protein turnover in precision-cut lung slice cultures, lung slices were cultured in the presence of heavy water, and conditioned media samples were collected on Day 7 for tandem mass spectrometric analysis of mass isotopomer abundances of trypsin-generated peptides
- A substantial fraction of heavy water-labeled peptides were quantified for several matrix proteins
- Peptides measured in conditioned media samples from 7-day cultured precision-cut lung slices contained stable isotopes, indicating that they were newly synthesized during *ex vivo* culture

Table 4. Percentage of heavy water-labeled proteins synthesized during precision-cut lung slice culture

Select D_2O-labeled proteins	Protein synthesized during PCLS culture (D_2O labeled), %	Unlabeled control, %
Collagen type I alpha 1 chain	30	6
Fibronectin	55	0
Collagen type III alpha 1 chain	17	5
Lumican	30	3
Collagen type I alpha 2 chain	32	6
Metalloproteinase inhibitor 1	49	0
Collagen type VI alpha 1 chain	64	4

D_2O, heavy water; PCLS, precision-cut lung slice

CONCLUSIONS

- Proteomic profiling of conditioned media samples from *ex vivo* cultured fibrotic lung tissue was performed to identify putative biomarker candidates for idiopathic pulmonary fibrosis
- We identified 99 proteins present at significantly different levels between precision-cut lung slices treated with transforming growth factor-beta type I receptor kinase inhibitor (ALK5i) vs. vehicle
- This study demonstrates the utility of the precision-cut lung slice culture methodology, combined with deep proteomic profiling, to identify idiopathic pulmonary fibrosis biomarkers modulated by drug treatment of lung tissue from patients with idiopathic pulmonary fibrosis

References: 1. Decaris ML, et al. *Respir Res* 2021;22(1):265; 2. Maher TM, et al. *Respir Res* 2021;22(1):197; 3. Hutchinson JH, et al. *Eur Respir J* 2015;46(3):795–806; 4. Alsafadi HN, et al. *Am J Physiol Lung Cell Mol Physiol* 2017;312(6):L896–L902

Contact information: man@pliantrx.com

Poster presented at the American Thoracic Society (ATS) International Conference, May 13–18, 2022. Copyright © 2022 Pliant Therapeutics, Inc. | All Rights Reserved

Acknowledgments: Editorial assistance was provided by Alpharmaxim Healthcare Communications and funded by Pliant Therapeutics, Inc.