PLN-74809, AN ORAL, DUAL-SELECTIVE $\alpha_v\beta_6/\alpha_v\beta_1$ INHIBITOR IN PHASE 2 CLINICAL TRIALS FOR IDIOPATHIC PULMONARY FIBROSIS (IPF), SUSTAINABLY REDUCES TRANSFORMING GROWTH FACTOR-BETA (TGF-β) ACTIVITY IN THE LUNGS OF HEALTHY PARTICIPANTS WITH ONCE-DAILY DOSING

Cosgrove G, Decaris M, Wong S, Park E,* Turner S, Adegbite E,* Lefebvre E
Pliant Therapeutics, Inc., South San Francisco, CA, USA
Research supported by Pliant Therapeutics, Inc.
Authors' relevant interests: All authors were employed by Pliant Therapeutics, Inc., and owned stock at the time of the study. *Former employees; did not contribute to poster development
Poster no. 707

BACKGROUND

- Integrins $\alpha_v\beta_6$ and $\alpha_v\beta_1$ are cell-surface proteins that regulate activation of transforming growth factor-beta (TGF-β) signaling in the fibrotic human lung
 - Integrins $\alpha_v\beta_6$ on lung epithelial cells and $\alpha_v\beta_1$ on lung fibroblasts activate latent TGF-β signaling, resulting in SMAD2/3 phosphorylation[1,2]
- Elevated levels of integrins $\alpha_v\beta_6$ and $\alpha_v\beta_1$ are detectable in the lungs of patients with interstitial lung disease compared with healthy participants[3–5]
 - High levels of integrin $\alpha_v\beta_6$ detected within lung tissue biopsies[6] and plasma[7] are predictors of worse survival rates for patients with interstitial lung disease
- PLN-74809 is an oral, once-daily, dual-selective inhibitor of $\alpha_v\beta_6$ and $\alpha_v\beta_1$ integrins in development for the treatment of idiopathic pulmonary fibrosis, with orphan drug designation granted by the United States Food and Drug Administration[8]
- Localized TGF-β inhibition in the fibrotic lung, achieved by targeting $\alpha_v\beta_6$ and $\alpha_v\beta_1$, integrins with PLN-74809, may provide a novel approach for treating idiopathic pulmonary fibrosis, without affecting TGF-β signaling systemically
- The PLN-74809-P1-03 study previously demonstrated significantly reduced TGF-β signaling 6 hours post-dose after 7 days (40 mg once daily; p<0.05)[9]
- In lung tissue explanted from patients with idiopathic pulmonary fibrosis (precision-cut lung slices), PLN-74809 significantly reduced *COL1A1* mRNA expression when tested alone (42%; p<0.001), while nintedanib and pirfenidone alone had no effect on *COL1A1* expression when tested at their approximate clinical maximum observed drug concentration (C_{max}) levels of 75 nM and 50 μM, respectively[3]

STUDY DESIGN

- Alveolar macrophages collected by bronchoalveolar lavage enable the evaluation of $\alpha_v\beta_6$-dependent TGF-β signaling in human lungs and determination of $\alpha_v\beta_6$-inhibitor pharmacokinetics/pharmacodynamics (**Figure 1**)[8]
- PLN-74809-109 is an ongoing, double-blind, placebo-controlled, pharmacokinetic/pharmacodynamic Phase 1b study in 48 healthy participants, consisting of two parts:
 - In Part 1, plasma concentrations of PLN-74809 and pSMAD2/SMAD2 ratio in bronchoalveolar lavage cells were assessed in 24 participants, following 7 days of once-daily dosing with PLN-74809 80 mg, PLN-74809 160 mg, or placebo (**Figure 2**)
 - In Part 2, 24 participants will receive a daily dose of PLN-74809 320 mg or placebo
- Key inclusion criteria included participants being healthy adults 18–55 years of age, non-smokers, and having no known lung disease
- The primary objective was to assess the pharmacodynamic changes in biomarkers of TGF-β activation in alveolar macrophages collected from bronchoalveolar lavage following dosing of PLN-74809 for 7 days
- Secondary objectives included assessment of the plasma pharmacokinetics of PLN-74809, the safety and tolerability of PLN-74809 following dosing for 7 days, and the pharmacokinetic/pharmacodynamic relationship based on the pSMAD2/SMAD2 ratio
- Bronchoalveolar lavage fluid was collected prior to dosing (Day -1) and at 6 and 24 hours post-dose on Day 7
 - Bronchoalveolar lavage procedures on Day 7 alternated between the right middle lobe and lingula
 - Bronchoalveolar lavage cell pSMAD2/total SMAD2 ratio was quantified by electrochemiluminescence assay and bronchoalveolar lavage white blood cell (leukocyte) differentials were performed
 - Participants with no Day 7 bronchoalveolar lavage fluid samples could be replaced
- Plasma samples for pharmacokinetic analysis were collected at pre-dose and 0.5, 1, 2, 3, 4, 6, 12, and 24 hours post-dose on Days 1 and 7

Figure 1. Integrins $\alpha_v\beta_6$ and $\alpha_v\beta_1$ play crucial roles in TGF-β activation associated with the formation of fibrotic lung tissue in patients with IPF

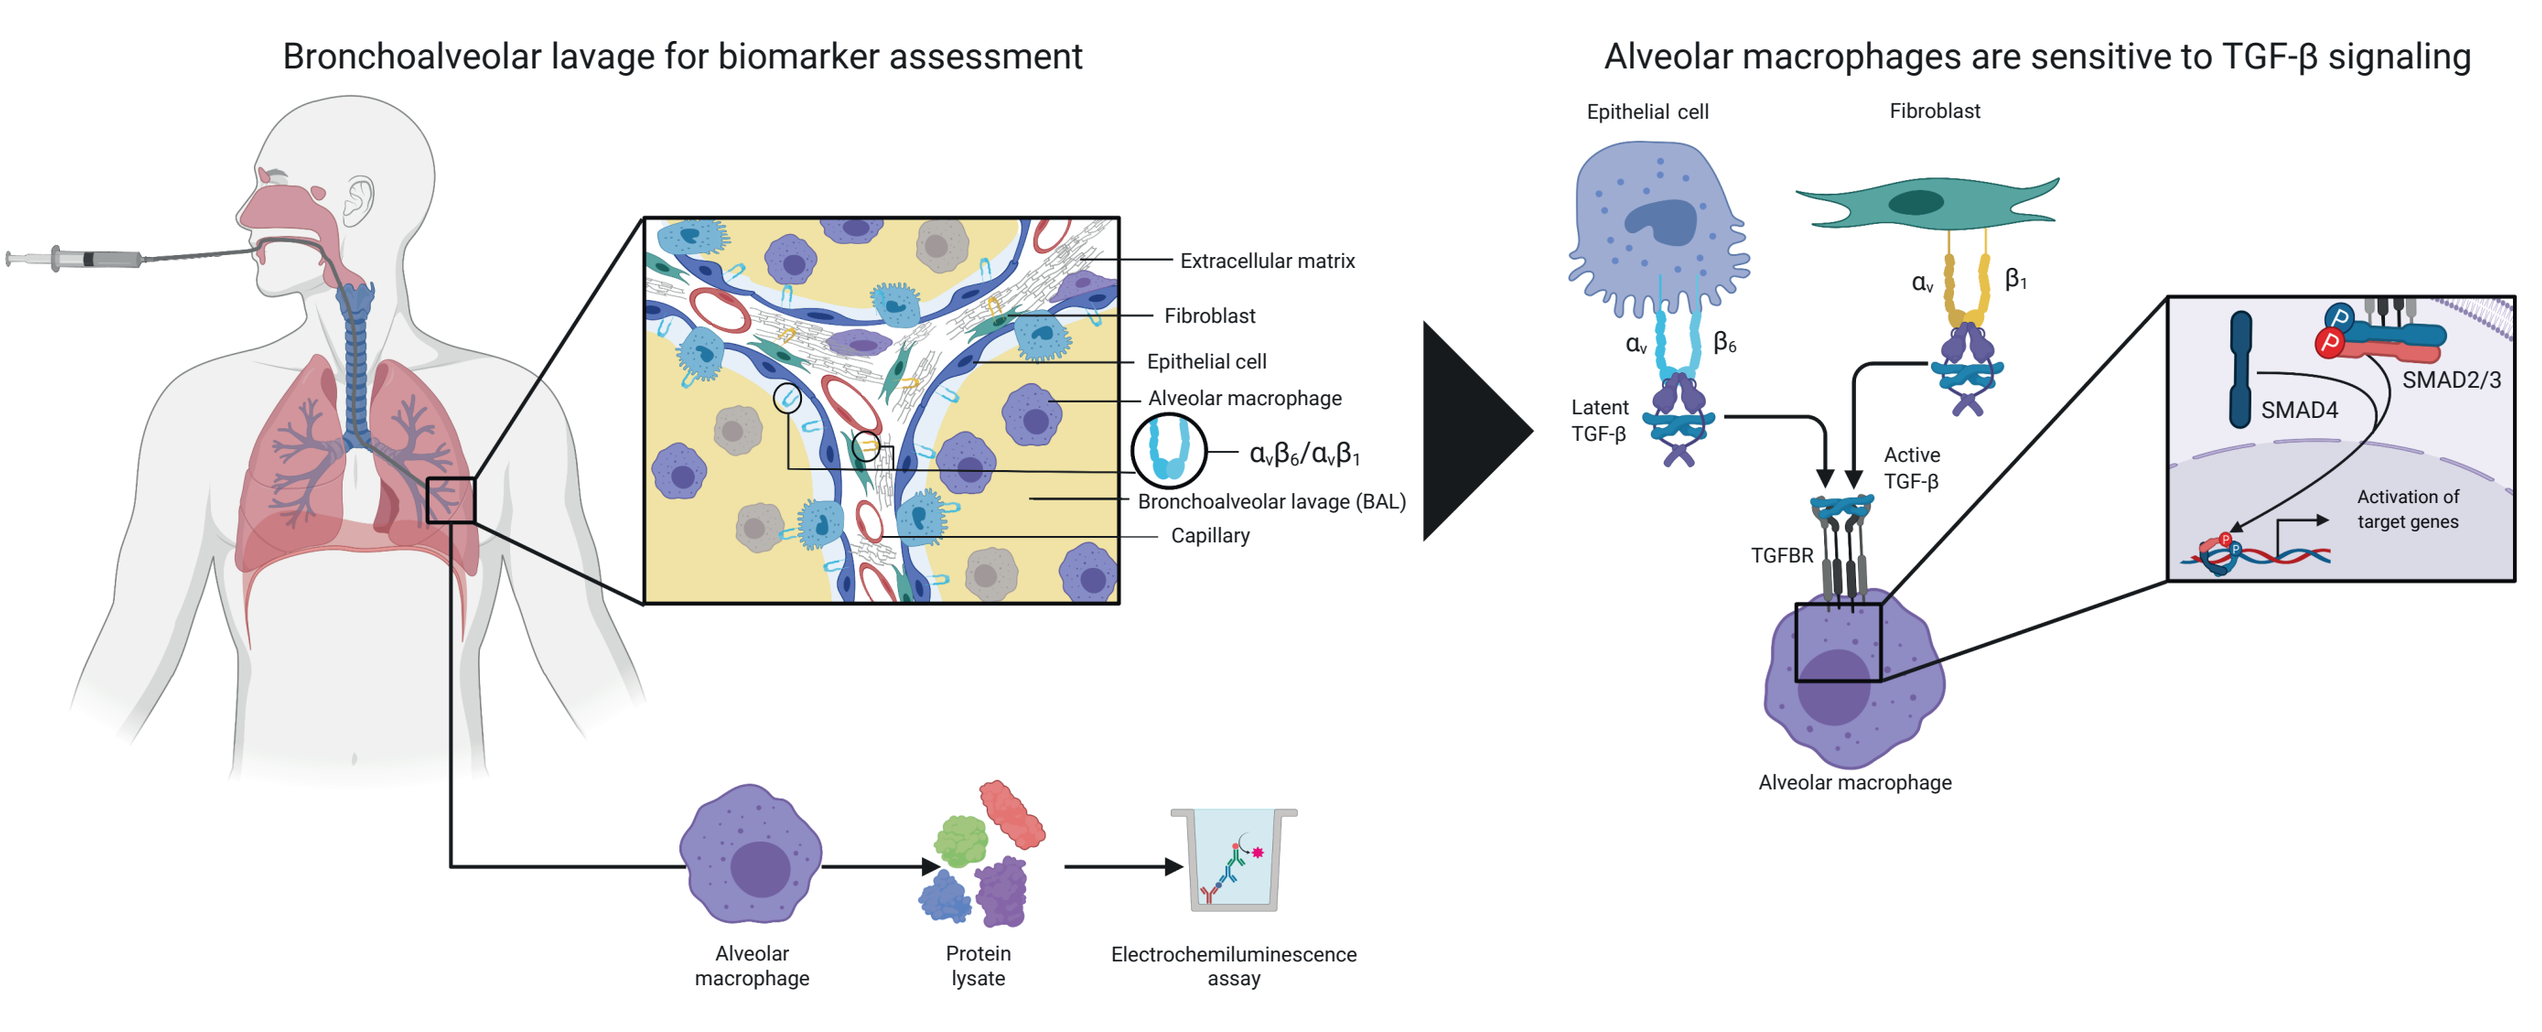
Bronchoalveolar lavage for biomarker assessment
Extracellular matrix
Fibroblast
Epithelial cell
Alveolar macrophage
αvβ6/αvβ1
Bronchoalveolar lavage (BAL)
Capillary
Alveolar macrophage
Protein lysate
Electrochemiluminescence assay
Alveolar macrophages are sensitive to TGF-β signaling
Epithelial cell
Fibroblast
Latent TGF-β
Active TGF-β
TGFBR
SMAD4
SMAD2/3
Activation of target genes
Alveolar macrophage

IPF, idiopathic pulmonary fibrosis; SMAD, family of proteins similar to the gene products of the Drosophila gene 'mothers against decapentaplegic' *(Mad)* and the *C. elegans gene Sma*; TGF-, transforming growth factor-beta; TGFBR, TGF- receptor

Figure 2. PLN-74809-109 (Part 1) study design

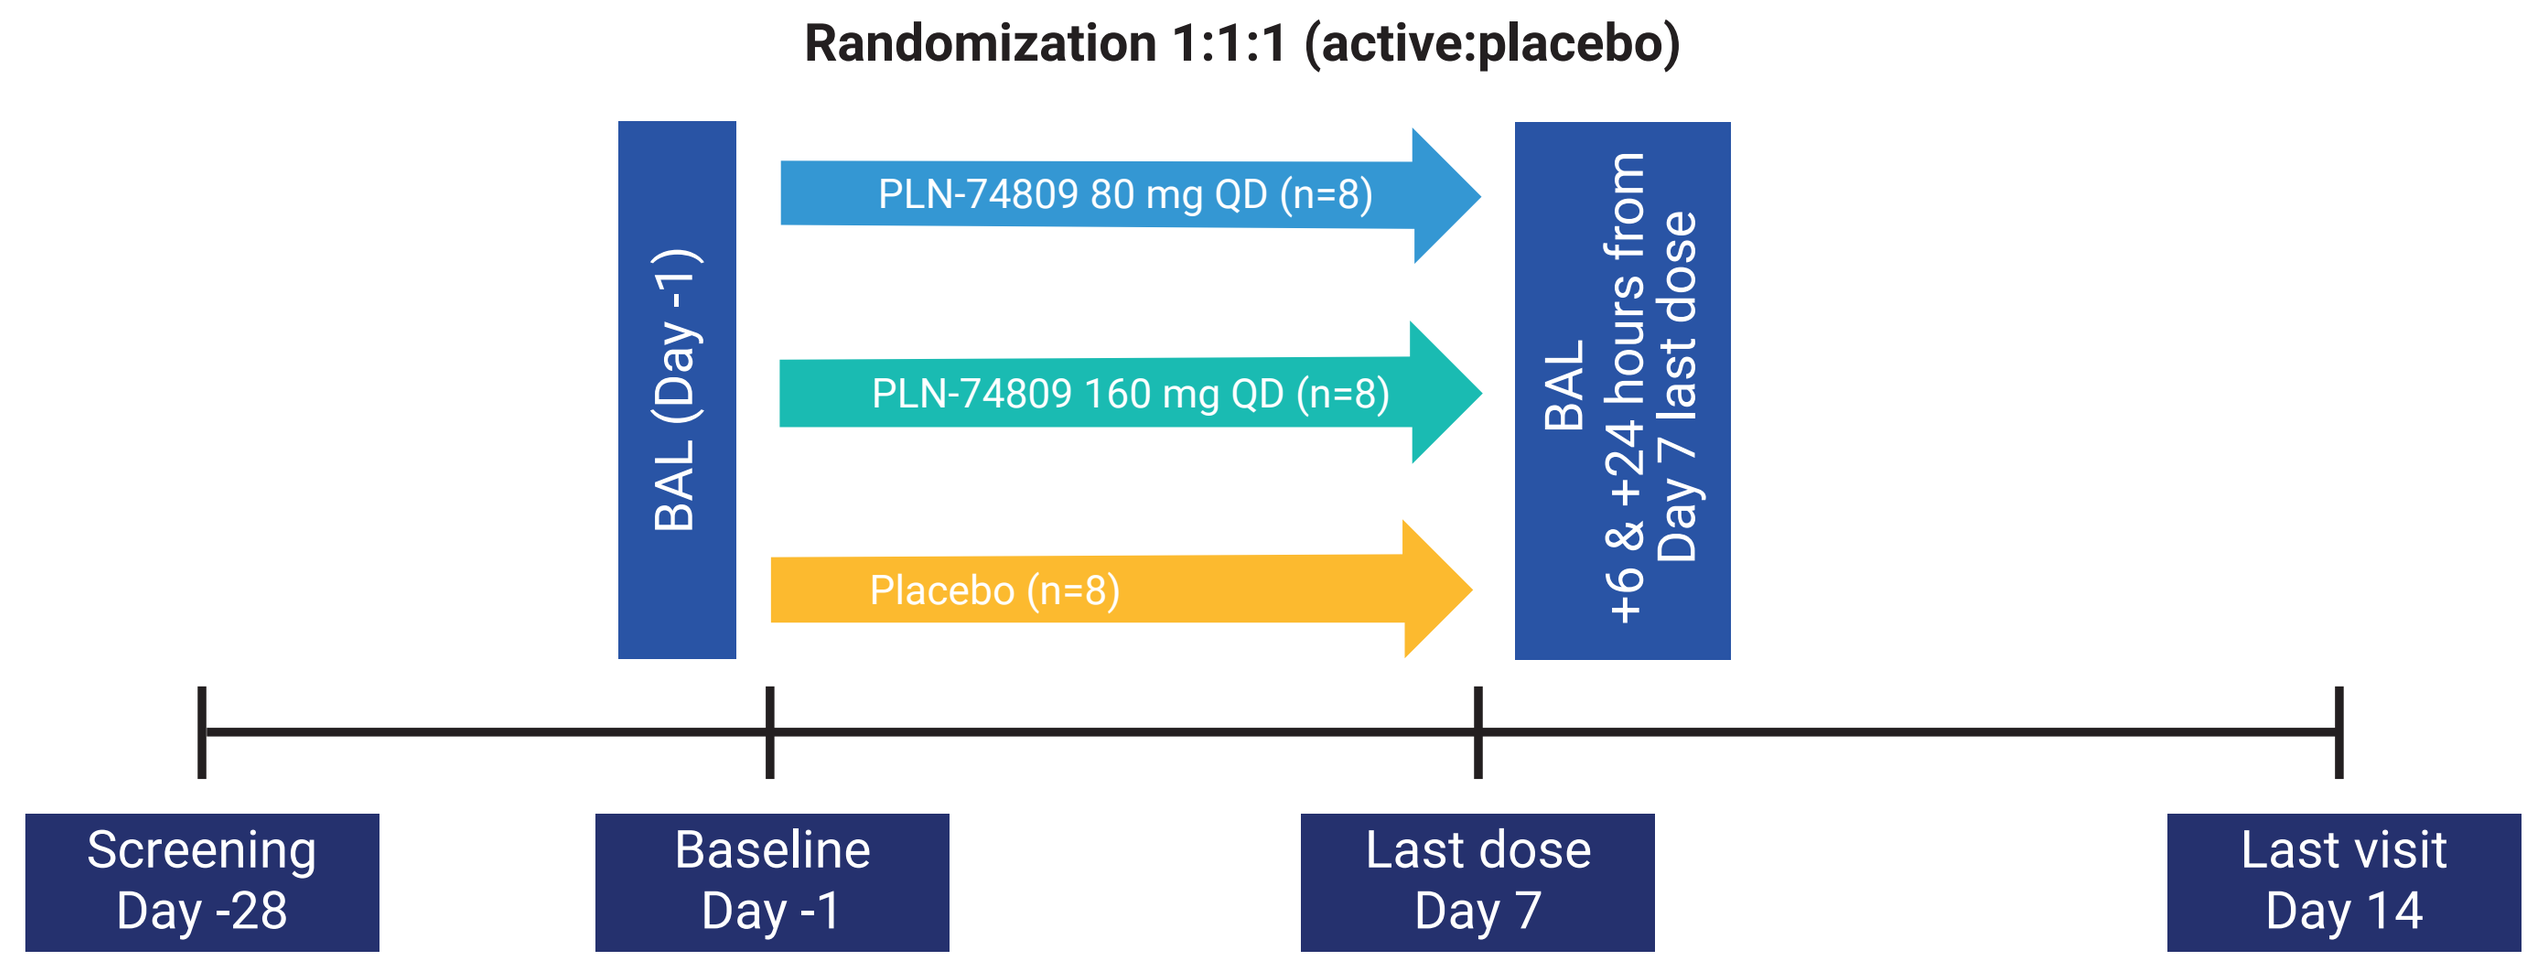
Randomization 1:1:1 (active:placebo)
BAL (Day -1)
PLN-74809 80 mg QD (n=8)
PLN-74809 160 mg QD (n=8)
Placebo (n=8)
BAL +6 & +24 hours from Day 7 last dose
Screening Day -28
Baseline Day -1
Last dose Day 7
Last visit Day 14

BAL, bronchoalveolar lavage; QD, once daily

Figure 3. Percent change in alveolar pSMAD2/SMAD2 ratio from baseline to 6 and 24 hours post-dose on Day 7 following 7 days of once-daily dosing

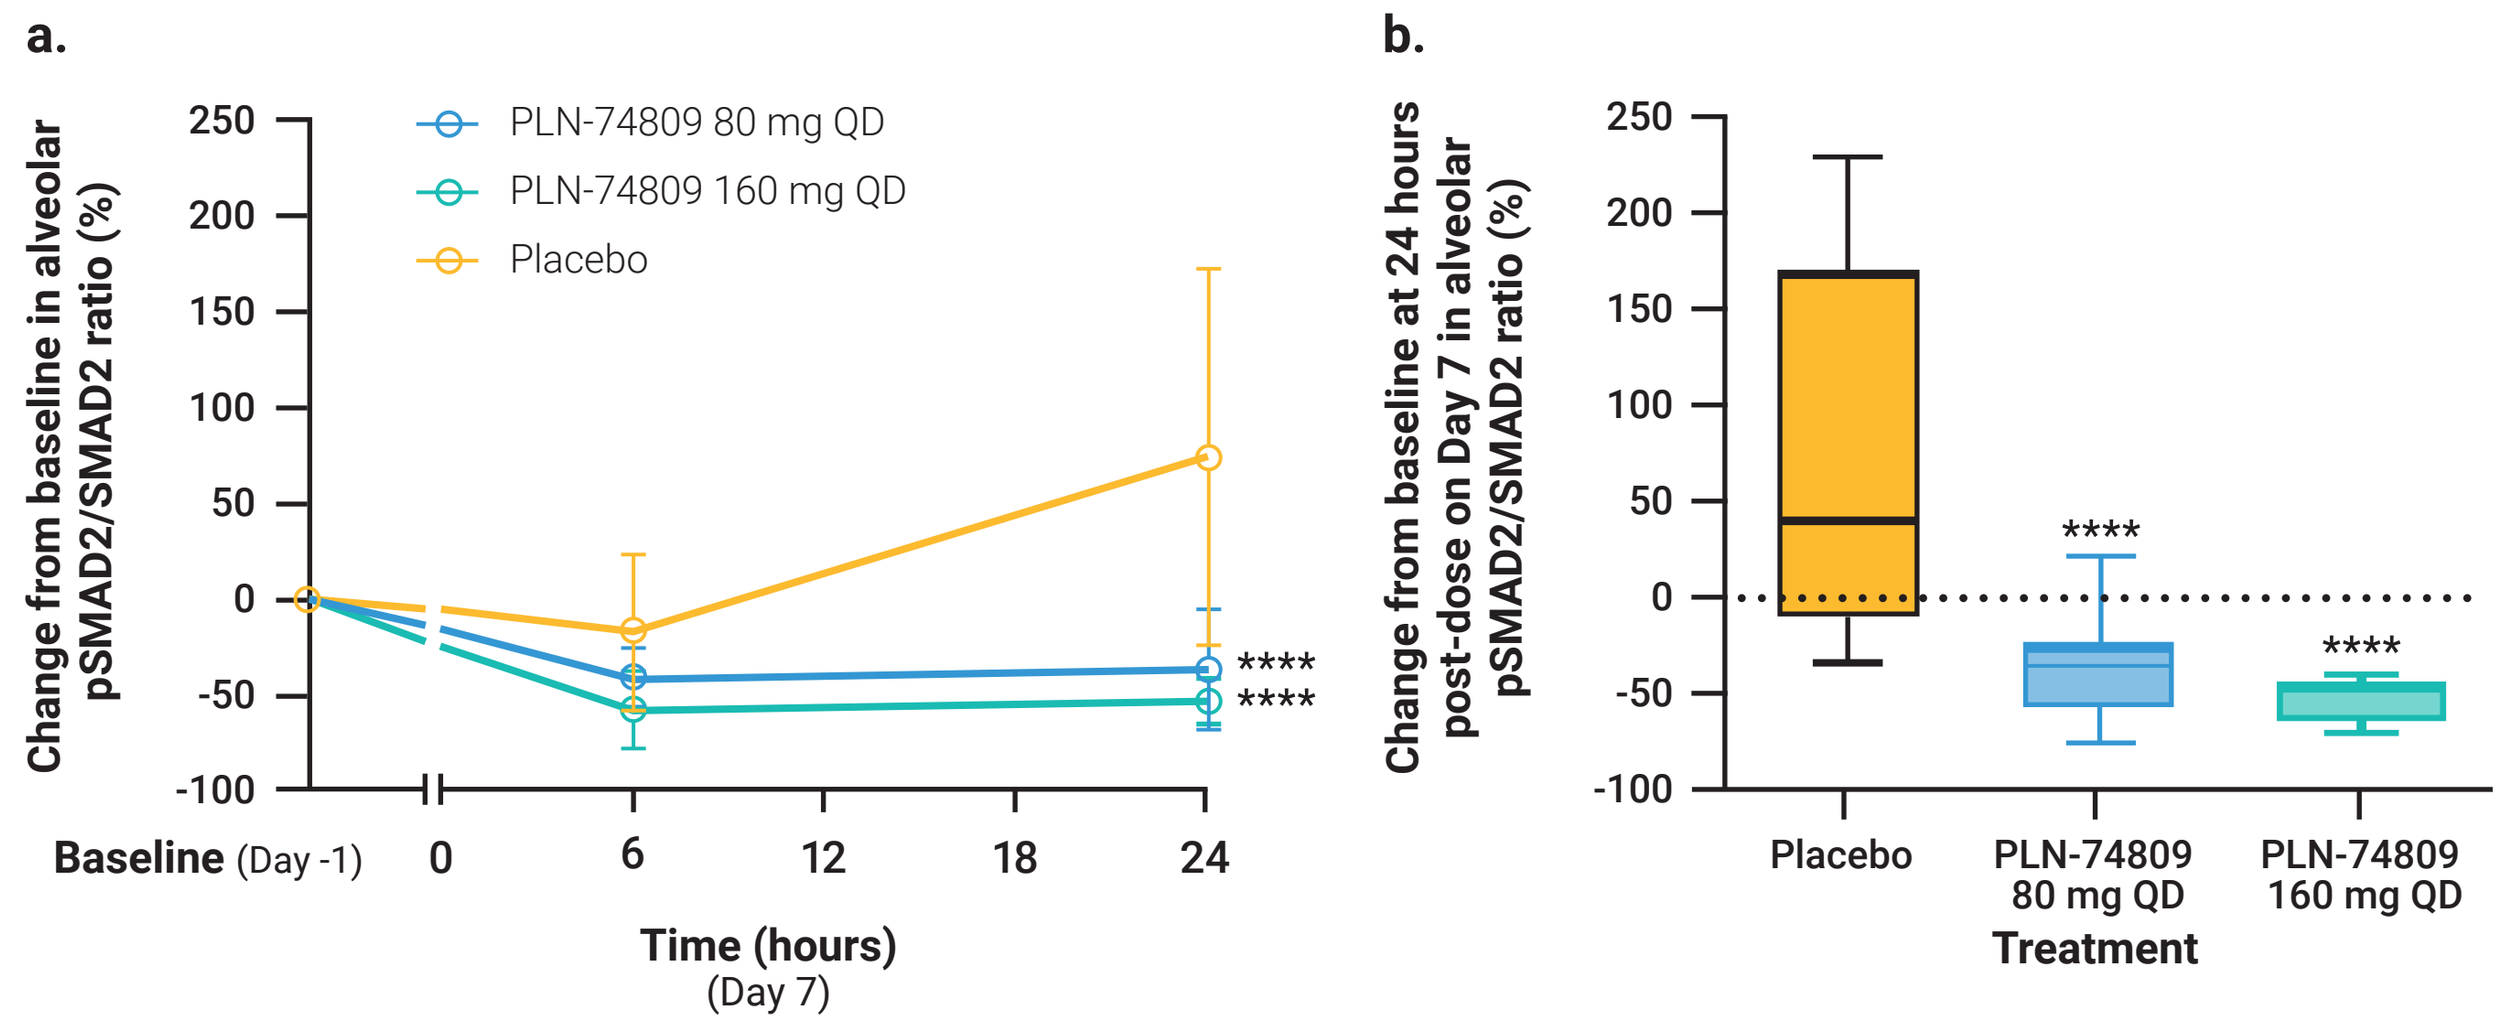
a.
Change from baseline in alveolar pSMAD2/SMAD2 ratio (%)
PLN-74809 80 mg QD
PLN-74809 160 mg QD
Placebo

Baseline (Day -1)
Time (hours) (Day 7)
b.
Change from baseline at 24 hours post-dose on Day 7 in alveolar pSMAD2/SMAD2 ratio (%)
Placebo
PLN-74809 80 mg QD
PLN-74809 160 mg QD
Treatment

****p<0.0001 (vs. baseline [Figure 3a]; vs. placebo [Figure 3b])
p, phosphorylation; QD, once daily; SMAD, family of proteins similar to the gene products of the Drosophila gene 'mothers against decapentaplegic' *(Mad)* and the *C. elegans* gene *Sma*

Figure 4. Dose- and concentration-dependent reduction in TGF-β signaling following treatment with PLN-74809 at 80 mg and 160 mg QD support PK/PD relationship

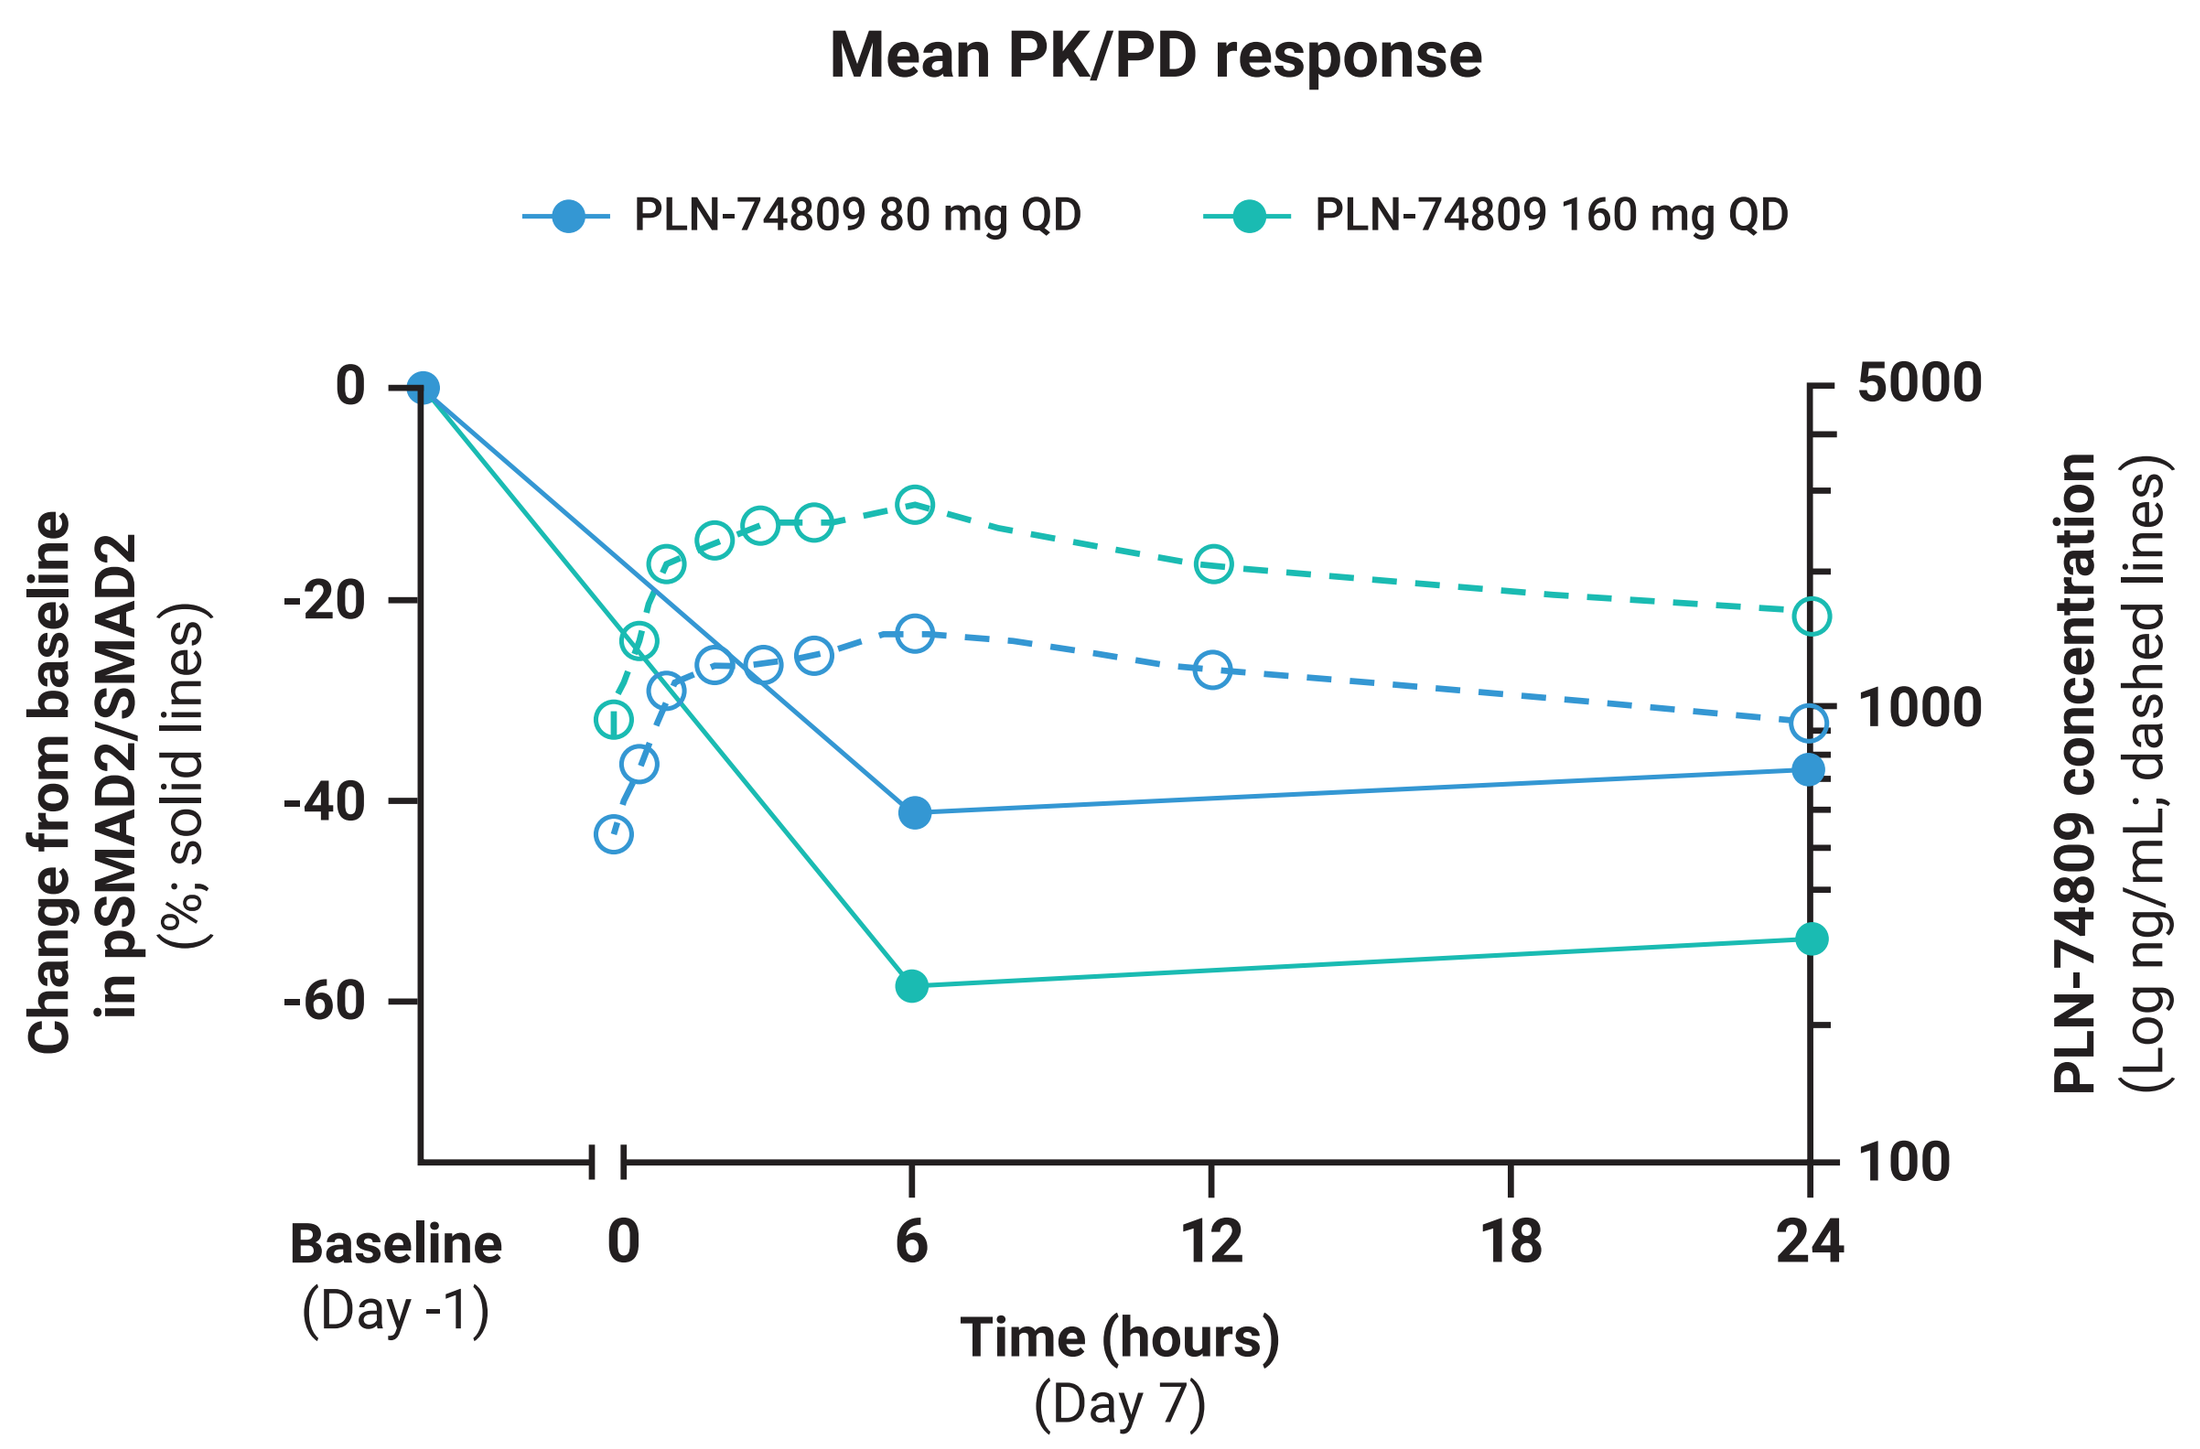
Mean PK/PD response
PLN-74809 80 mg QD
PLN-74809 160 mg QD
Change from baseline in pSMAD2/SMAD2 (%; solid lines)
PLN-74809 concentration (Log ng/mL; dashed lines)
Baseline (Day -1)
Time (hours) (Day 7)

p, phosphorylation; PD, pharmacodynamic; PK, pharmacokinetic; QD, once daily; SMAD, family of proteins similar to the gene products of the Drosophila gene 'mothers against decapentaplegic' *(Mad)* and the *C. elegans gene Sma;* TGF-β, transforming growth factor-beta

Table 1. Drug-related treatment-emergent adverse events by severity during Part 1 of the PLN-74809-109 study

	Participants, n (%)		
TEAE severity	PLN-74809 80 mg QD (n=8)	PLN-74809 160 mg QD (n=8)	Placebo (n=9[a])
Mild	1 (12.5)	0 (0.0)	0 (0.0)
Moderate	0 (0.0)	0 (0.0)	1 (11.1)
Severe	0 (0.0)	0 (0.0)	0 (0.0)

[a]One participant on placebo reported AEs of moderate dyspnea exertional and mild pleuritic pain following the Day -1 bronchoalveolar lavage procedure, completed the study but did not undergo Day 7 bronchoalveolar lavage procedures, and was subsequently replaced
AE, adverse event; QD, once daily; TEAE, treatment-emergent adverse event

Figure 5. Percentages of bronchoalveolar lavage leukocyte populations in study participants, following treatment with placebo or PLN-74809 80 mg or 160 mg

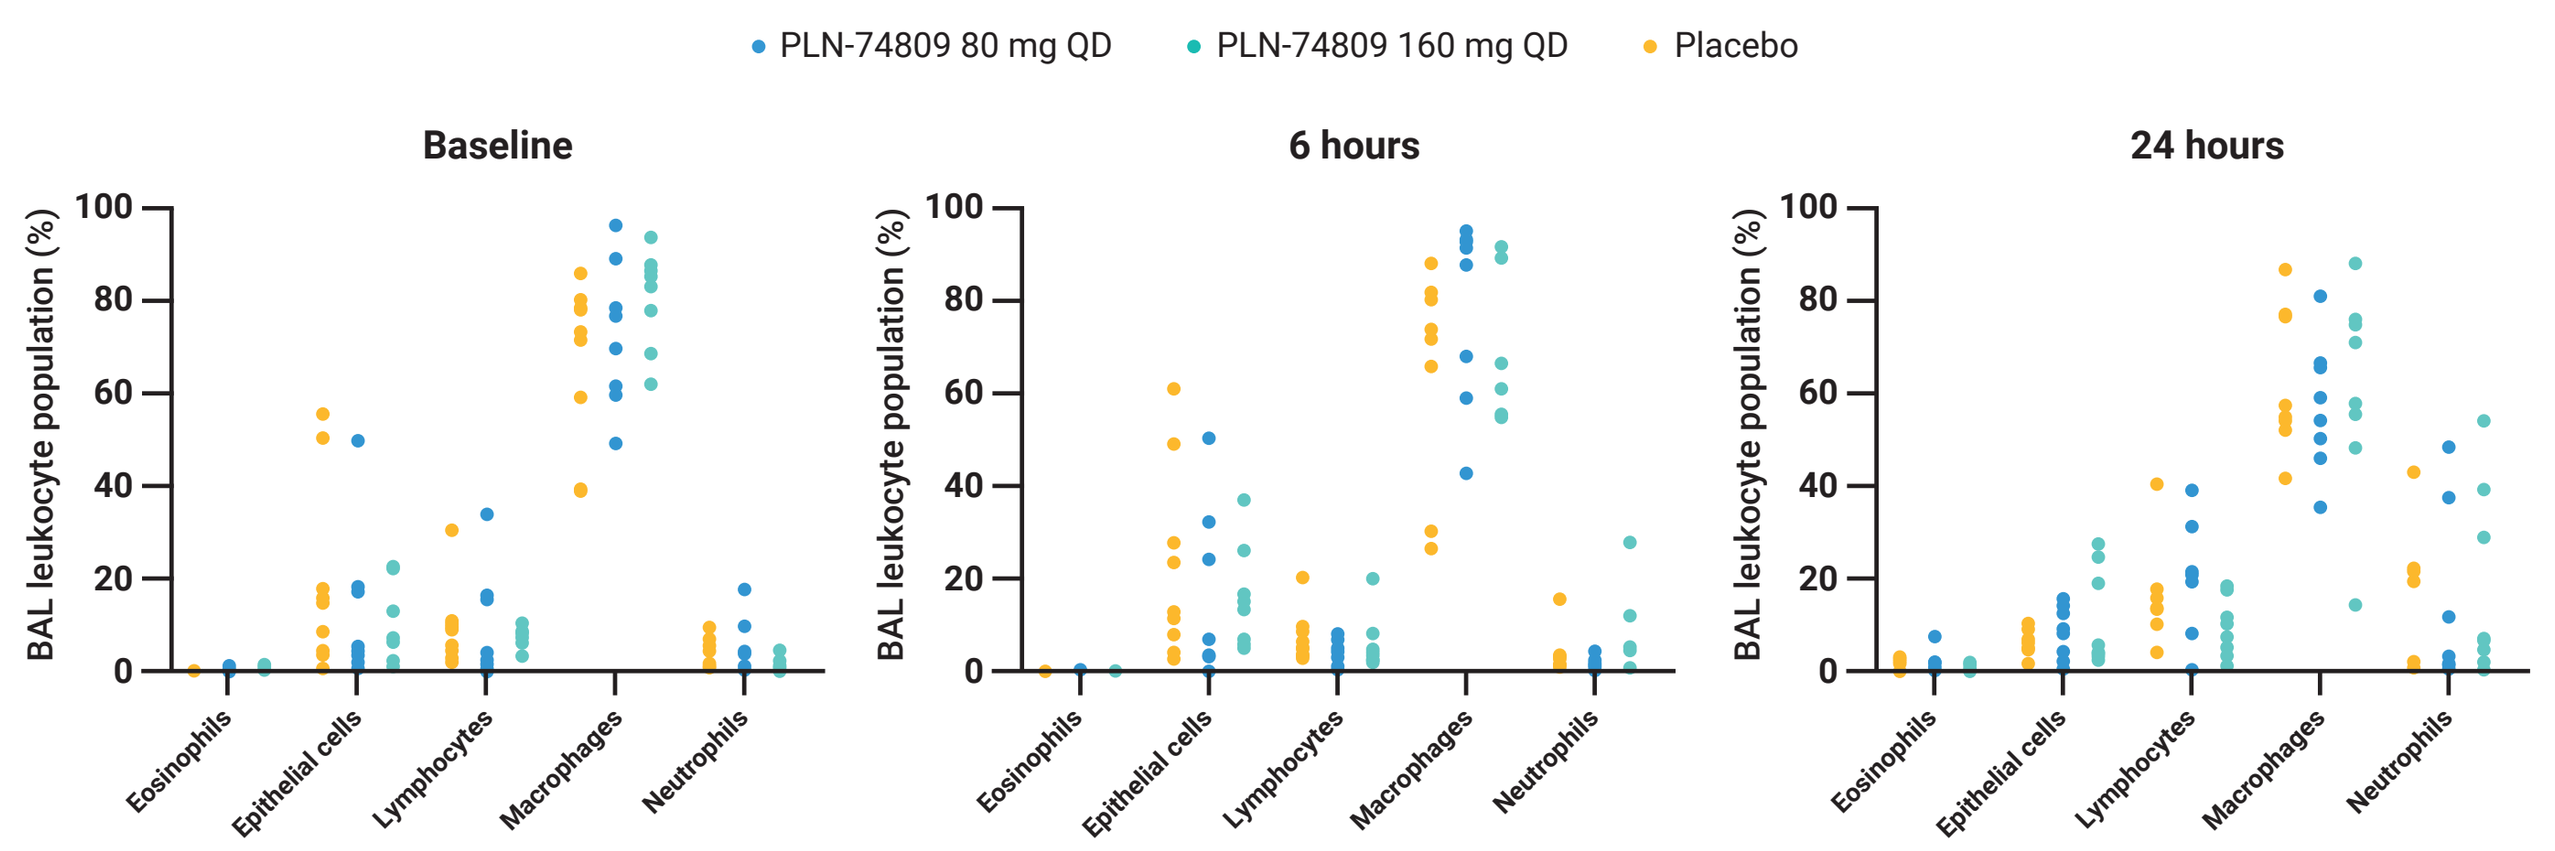
PLN-74809 80 mg QD
PLN-74809 160 mg QD
Placebo
Baseline
6 hours
24 hours
BAL leukocyte population (%)
Eosinophils
Epithelial cells
Lymphocytes
Macrophages
Neutrophils

BAL, bronchoalveolar lavage; QD, once daily

- Safety assessments included open-ended adverse event inquiry, physical examination with vital signs, clinical chemistry, hematology, and 12-lead electrocardiogram
- Plasma pharmacokinetic parameters were computed from the individual plasma concentrations of PLN-74809 using a non-compartmental approach
- Exploratory objectives included measurement of changes in differential cell count in bronchoalveolar lavage fluid following dosing of PLN-74809 for 7 days

RESULTS

- Reduced pSMAD2/SMAD2 ratios were observed at 6 and 24 hours following the final dose of PLN-74809 on Day 7, indicating that once-daily dosing with PLN-74809 (80 mg or 160 mg) led to dose- and concentration-dependent inhibition of TGF-β signaling (**Figure 3a**)
 - Compared with baseline, PLN-74809 reduced pSMAD2/SMAD2 ratio at 6 and 24 hours post-dose by:
 - 41.2% and 36.6%, respectively (80 mg)
 - 57.8% and 53.2%, respectively (160 mg)
- At 24 hours post-dose (Day 7), the reductions in pSMAD2/SMAD2 ratio for PLN-74809 80 mg and 160 mg were statistically significant compared with placebo (p<0.0001 for both comparisons), reflective of sustained reduction in TGF-β activation (**Figure 3b**)
- Assessment of pharmacokinetics/pharmacodynamics following treatment with PLN-74809 (80 mg or 160 mg) for 7 days demonstrated a dose- and concentration-dependent relationship between PLN-74809 plasma concentration and decreased pSMAD2/SMAD2 ratios (TGF-β signaling) in bronchoalveolar lavage cells (**Figure 4**)

Safety

- PLN-74809 was generally well tolerated, with few drug-related adverse events reported. No serious adverse events were reported
- All treatment-emergent PLN-74809-related adverse events were mild; none were moderate or severe (**Table 1**)
 - Mild headache was the only drug-related adverse event reported (80 mg)
- No clinically significant changes in physical examination, vital signs, chemistry or hematology laboratory studies were noted between the placebo or PLN-74809 cohorts (80 mg or 160 mg)
- Analysis of leukocyte populations within bronchoalveolar lavage cells showed no notable dose-dependent changes following 7 days of once-daily dosing with placebo or PLN-74809 80 mg or 160 mg, suggesting that there was no treatment-associated inflammation in human lungs resulting from exposure to PLN-74809 (**Figure 5**)
 - As expected, alveolar macrophages formed the majority of leukocytes present within bronchoalveolar lavage cells (79%)
 - There were no notable differences in the total cell number or distribution across the treatment groups (**Figure 5**)

CONCLUSIONS

- **Once-daily treatment with PLN-74809 80 mg or 160 mg led to sustained, dose- and concentration-dependent inhibition of TGF-β signaling in the lungs of healthy participants**
- **PLN-74809 was generally well tolerated with only mild drug-related adverse events**
- **The strong pharmacokinetic/pharmacodynamic relationship provides additional support for the doses of PLN-74809 being evaluated in the multinational Phase 2a INTEGRIS-IPF study (PLN-74809-IPF-202; NCT04396756) currently underway**

References: 1. Henderson NC, Sheppard D. *Biochim Phys Acta* 2013;1832(7):891–896; 2. Reed NI, et al. *Sci Transl Med* 2015;7(288):288ra79; 3. Decaris ML, et al. *Respir Res* 2021;22(1):265; 4. Kimura RH, et al. *Nat Commun* 2019;10(1):4673; 5. Horan GS, et al. *Am J Respir Crit Care Med* 2008;177(1):56–65; 6. Saini G, et al. *Eur Respir J* 2015;46(2):486–494; 7. Bowman WS, et al. *Lancet Respir Med* 2022 Jan 18 [Online ahead of print]. DOI: 10.1016/S2213-2600(21)00503-8; 8. Pliant Therapeutics, Inc. [Press release]. 6 August 2018. https://ir.pliantrx.com/news-releases/news-release-details/pliant-therapeutics-receives-fda-orphan-drug-designation-lead. Accessed May 5, 2022; 9. Gorina E, et al. *Am J Respir Crit Care Med* 2020;201:A4554

Contact information: gcosgrove@pliantrx.com

Poster presented at the American Thoracic Society (ATS) International Conference, May 13–18, 2022. Copyright © 2022 Pliant Therapeutics, Inc. | All Rights Reserved

Acknowledgments: Editorial assistance was provided by Alpharmaxim Healthcare Communications and funded by Pliant Therapeutics, Inc. Please see Poster no. 419 (PLN-74809, A Dual-Selective Inhibitor of $\alpha_v\beta_6$ and $\alpha_v\beta_1$, Is Well Tolerated in Over 280 Healthy Participants; B22 – Emerging Management of Fibrotic ILDs) for additional safety and tolerability details