Dual alpha-v/beta-6 and alpha-v/beta-1 integrin inhibitor bexotegrast attenuates profibrogenic gene expression of myofibroblasts in human liver explant tissue with biliary fibrosis

Steve Ho, Chris Her, Richard Ahn, Vikram Rao, Jessie Lau, Mahru An, Scott Turner, Martin Decaris, Johanna Schaub | Pliant Therapeutics, Inc., South San Francisco, CA, USA

POSTER # WED-215

BACKGROUND

Bexotegrast (PLN-74809), a dual inhibitor of TGF-β-activating integrins $\alpha_v\beta_6$/ $\alpha_v\beta_1$ currently in clinical development for the treatment of primary sclerosing cholangitis (PSC), has previously been shown to reduce fibrosis in the Mdr2$^{-/-}$ mouse model of biliary fibrosis. To examine the effects of bexotegrast on individual cell populations in the fibrotic human liver, we combined the precision-cut liver slice (PCLivS) platform, a physiologically relevant *ex vivo* model system, with 10x Genomics single nuclei RNA sequencing (snRNA-seq) to characterize the response of unique cell populations in fibrotic PSC and primary biliary cholangitis (PBC) PCLivS to bexotegrast.

METHODS

Precision-Cut Liver Slices

Liver explants were collected from patients with PSC (n = 3) and PBC (n = 1) at the time of transplant. All livers were transported on ice in UW with less than 24 hours of cold ischemia time. PCLivS were generated and cultured for 2 days in the presence of bexotegrast or vehicle (DMSO). A TGF-β receptor I kinase inhibitor (ALK5i; R-268712) that blocks TGF-β signaling, a well-known driver of fibrotic scar formation, was also evaluated as a control.

Figure 1. Generation and Culture of PCLivS

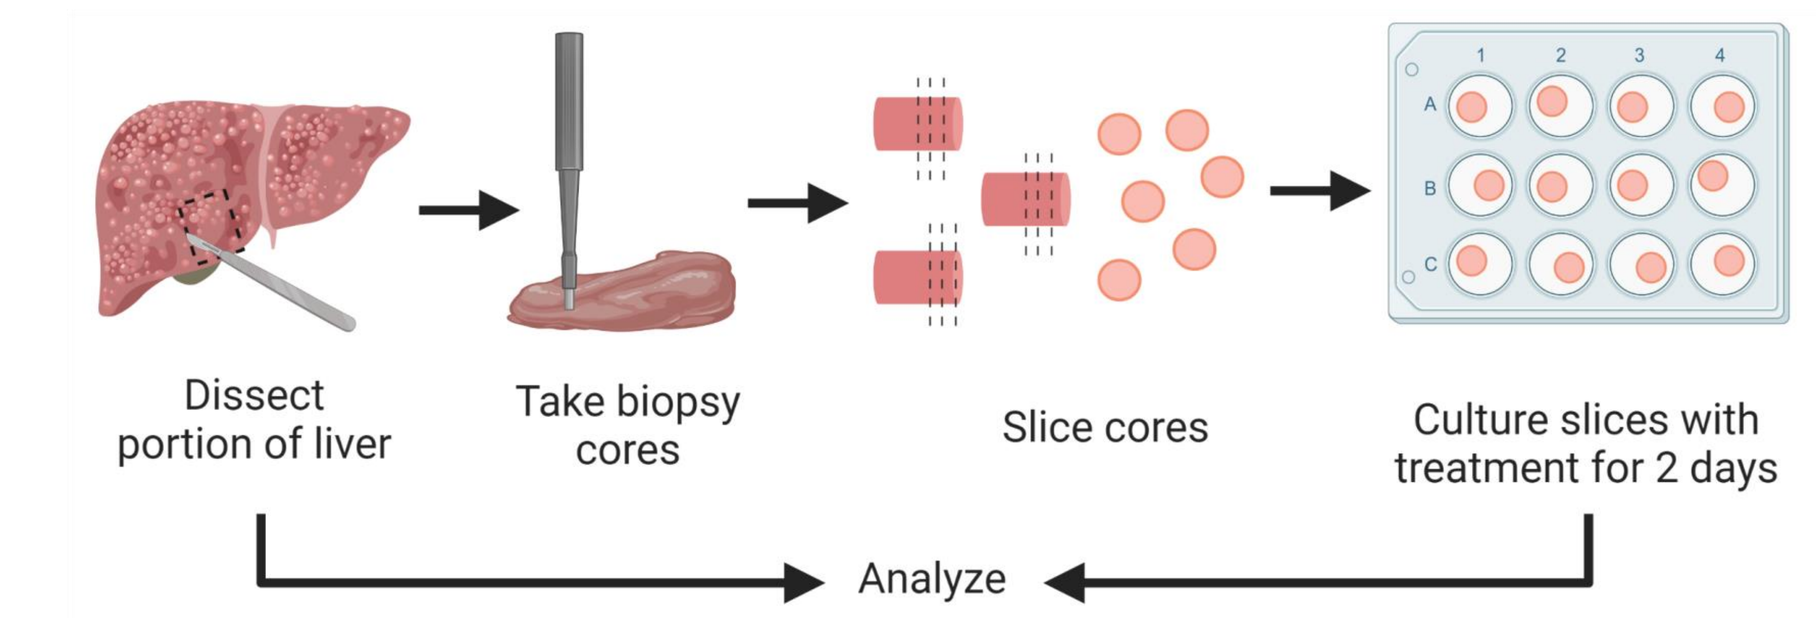
Dissect portion of liver
Take biopsy cores
Slice cores
Culture slices with treatment for 2 days
Analyze

Single Nuclei Isolation

Nuclei were isolated from 2 pooled slices per treatment using a combination of detergent-based lysis, mechanical disruption, and filtration. The crude nuclei suspension underwent density gradient centrifugation to remove debris and enrich for nuclei.

Figure 2. Isolation of Single Nuclei

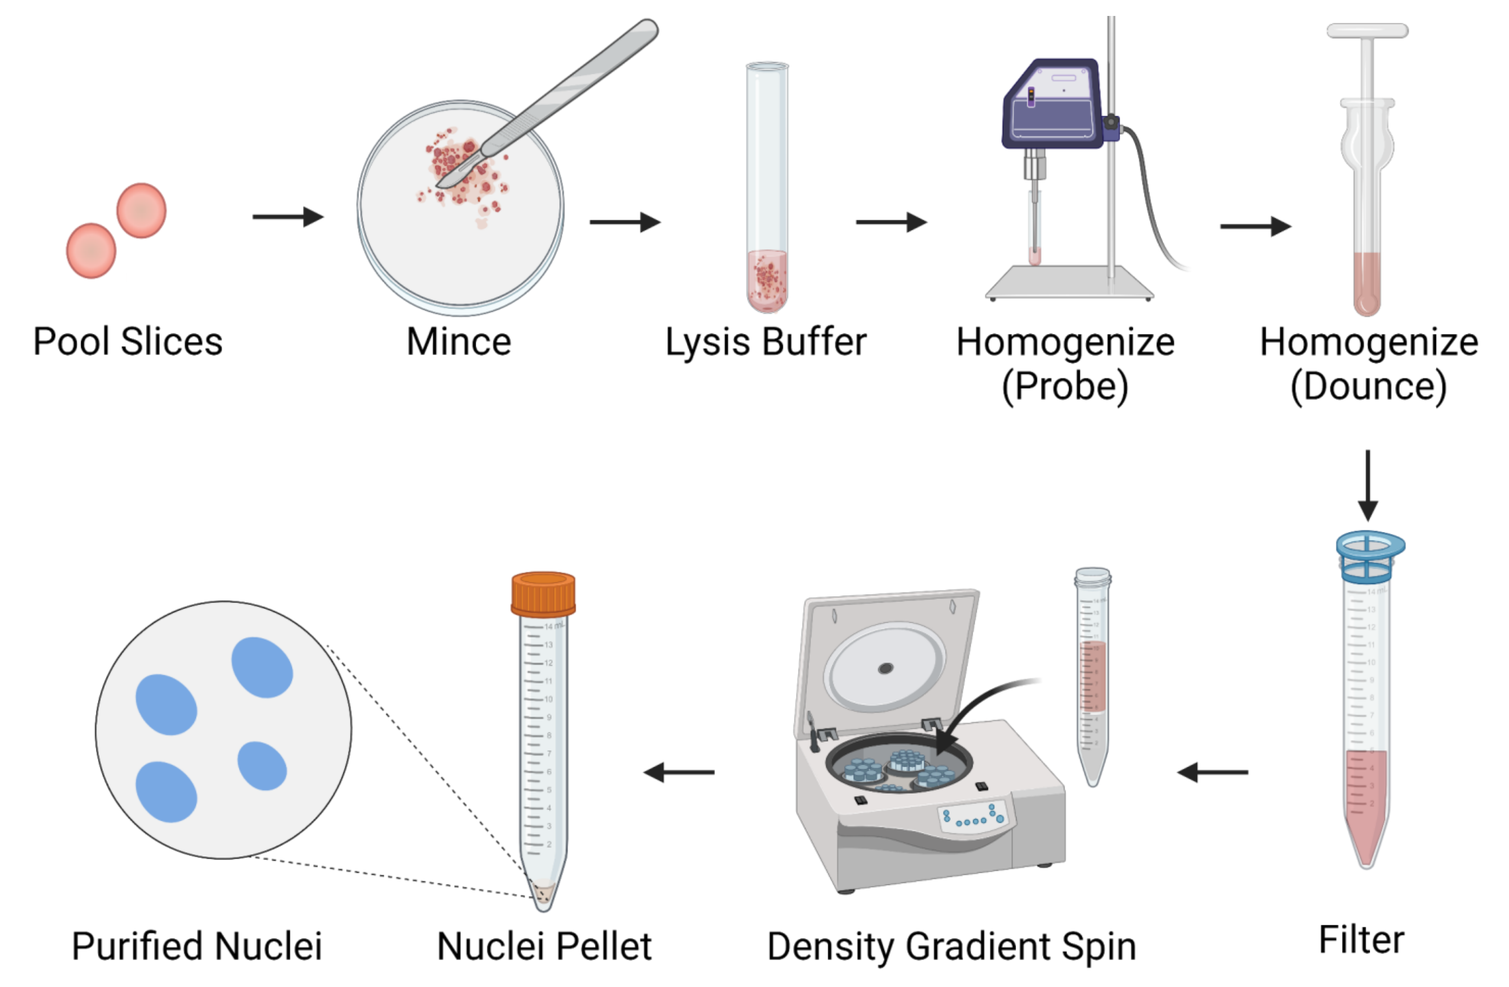
Pool Slices
Mince
Lysis Buffer
Homogenize (Probe)
Homogenize (Dounce)
Filter
Density Gradient Spin
Nuclei Pellet
Purified Nuclei

snRNA-seq and Analysis

Nuclei were processed for single nuclear barcoding using 10x Chromium Next GEM 3' HT kits. Resulting libraries were sequenced, processed using CellRanger, and analyzed using Seurat. Custom annotation of cell types was performed using gene markers established from recently published data sets[1,2]. Differential gene expression was determined using a non-parametric Wilcoxon rank sum test. Pathway enrichment analysis was performed with Enrichr[3].

RESULTS

Characterization of Livers for PCLivS

Table 1. Donor Information

Donor ID	Disease	Age	Sex
H3	PSC	37	M
H6	PSC	23	M
H16	PSC	56	F
H17	PBC	51	F

- PCLivS were generated from 4 donors with biliary fibrosis
- Picrosirius red staining shows fibrosis in donor explants

Figure 3. Fibrosis in Liver Donors

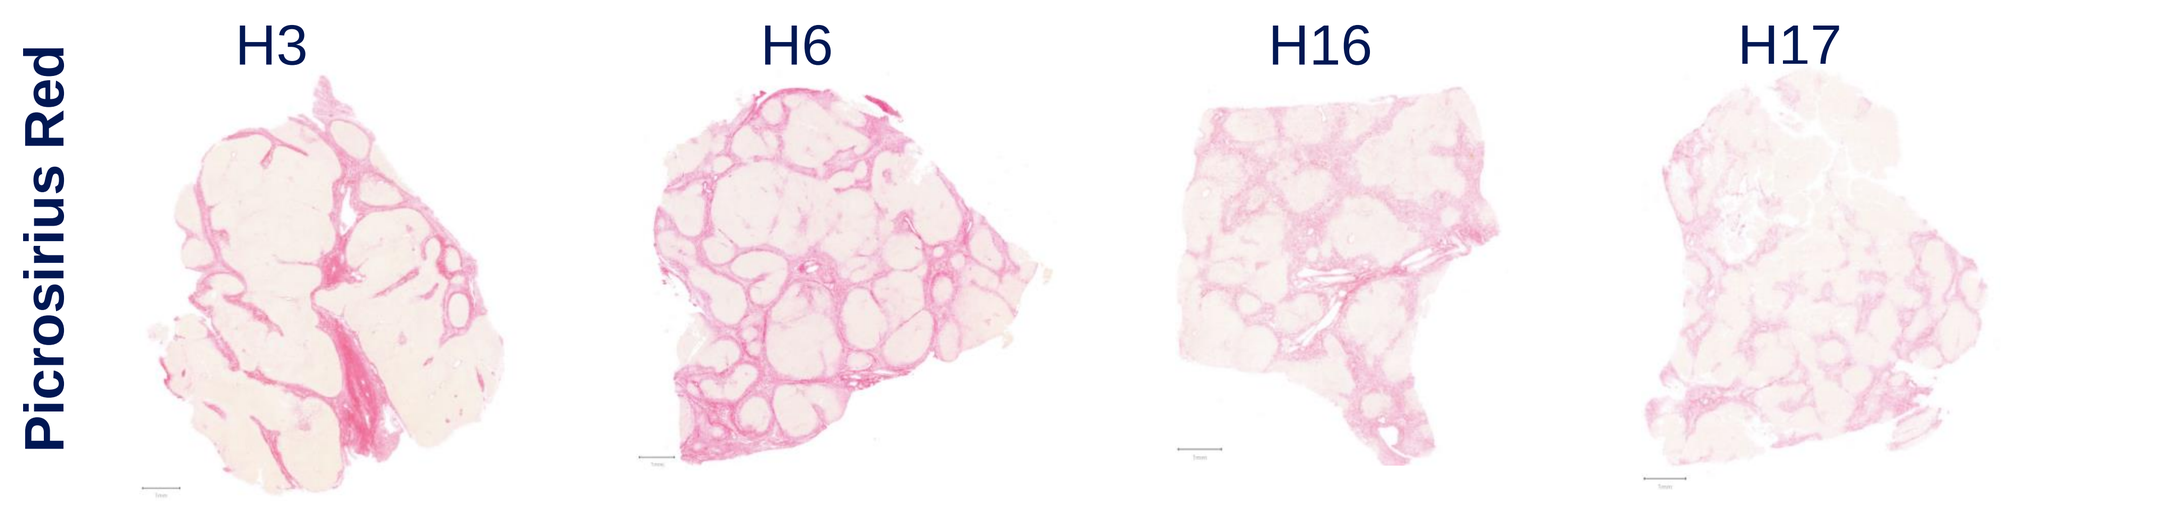
Picrosirius Red
H3
H6
H16
H17

Picrosirius red staining for collagen in liver explant tissue from 4 donors with biliary fibrosis
Scale bars = 1 mm.

Identification of Cells Present in PCLivS

Figure 4. snRNA-seq Analysis Identified Major Hepatic Cell Populations

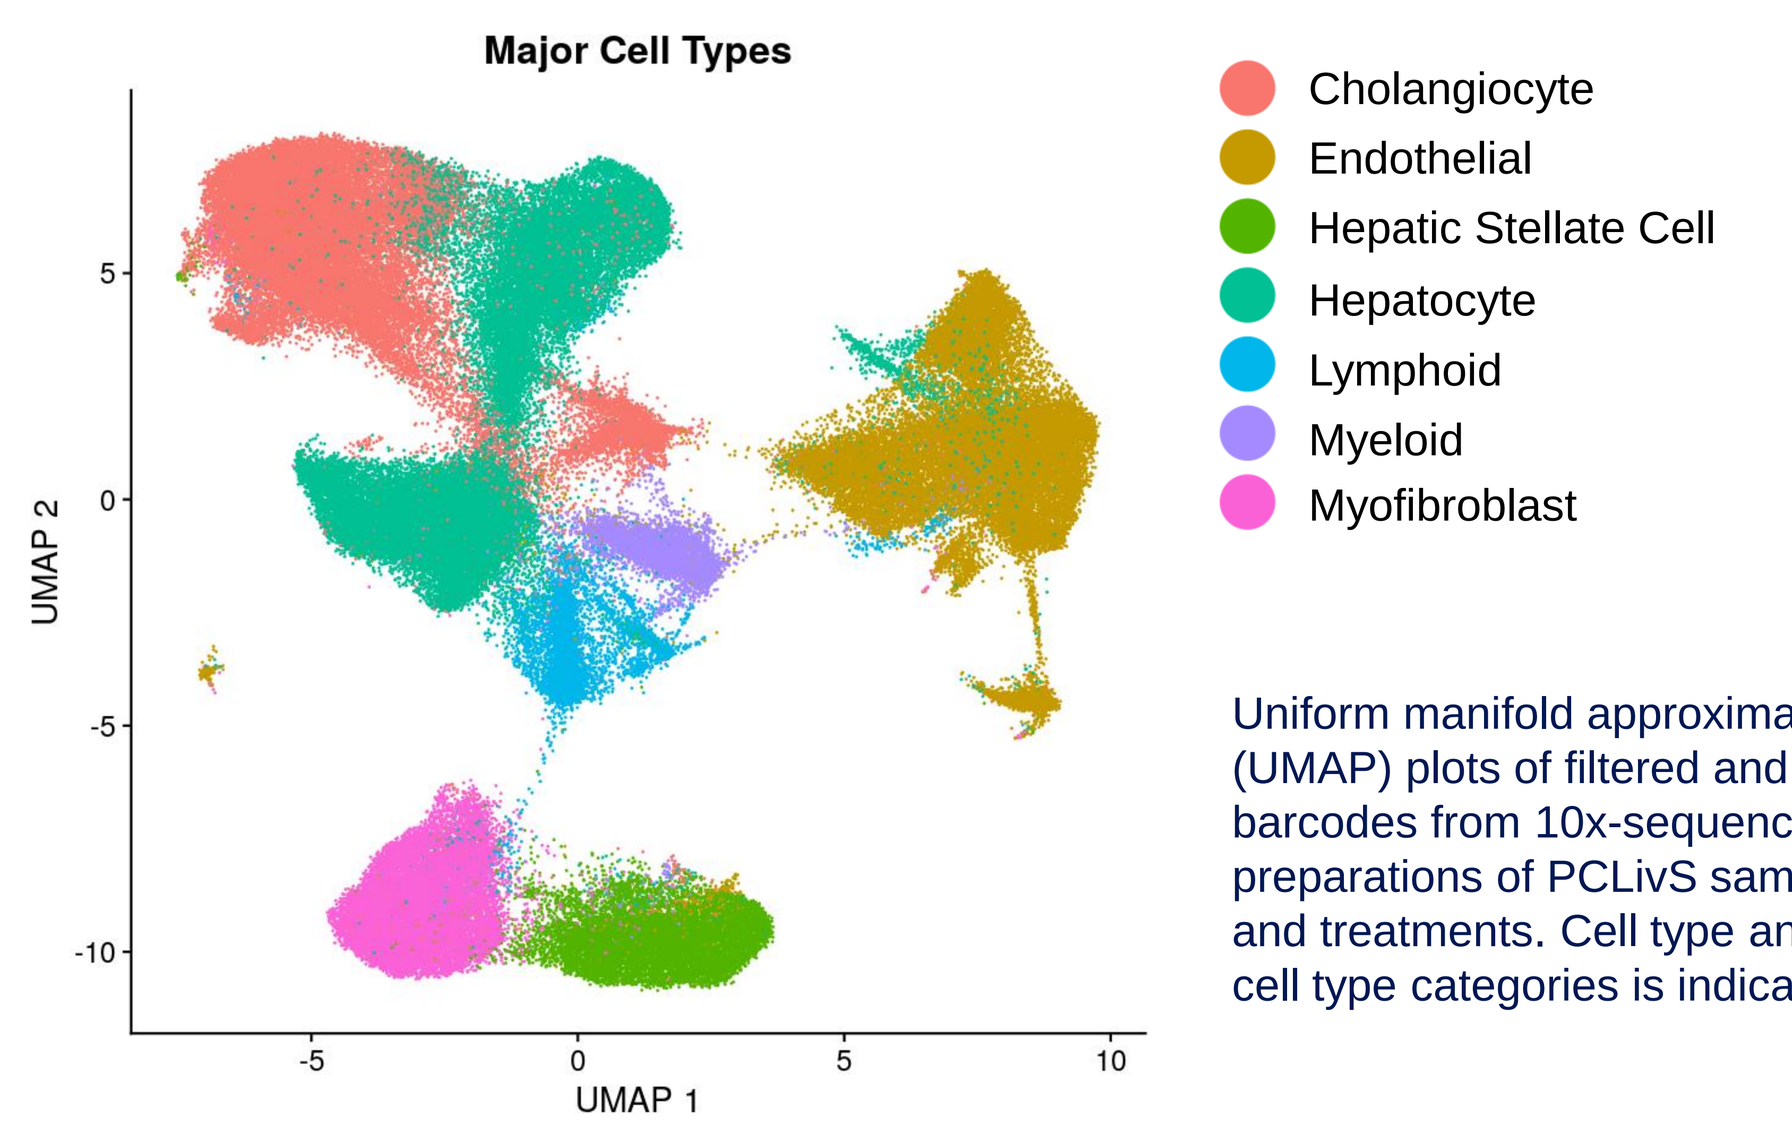
Major Cell Types
Cholangiocyte
Endothelial
Hepatic Stellate Cell
Hepatocyte
Lymphoid
Myeloid
Myofibroblast
UMAP 1
UMAP 2

Uniform manifold approximation and projection (UMAP) plots of filtered and normalized unique barcodes from 10x-sequenced single nuclei preparations of PCLivS samples from all donors and treatments. Cell type annotation for major cell type categories is indicated by color legend.

Evaluation of Differentially Expressed Genes in Individual Cell Populations

Figure 5. snRNA-seq Analysis Identified *PDGFRA*$^+$ Myofibroblast Population

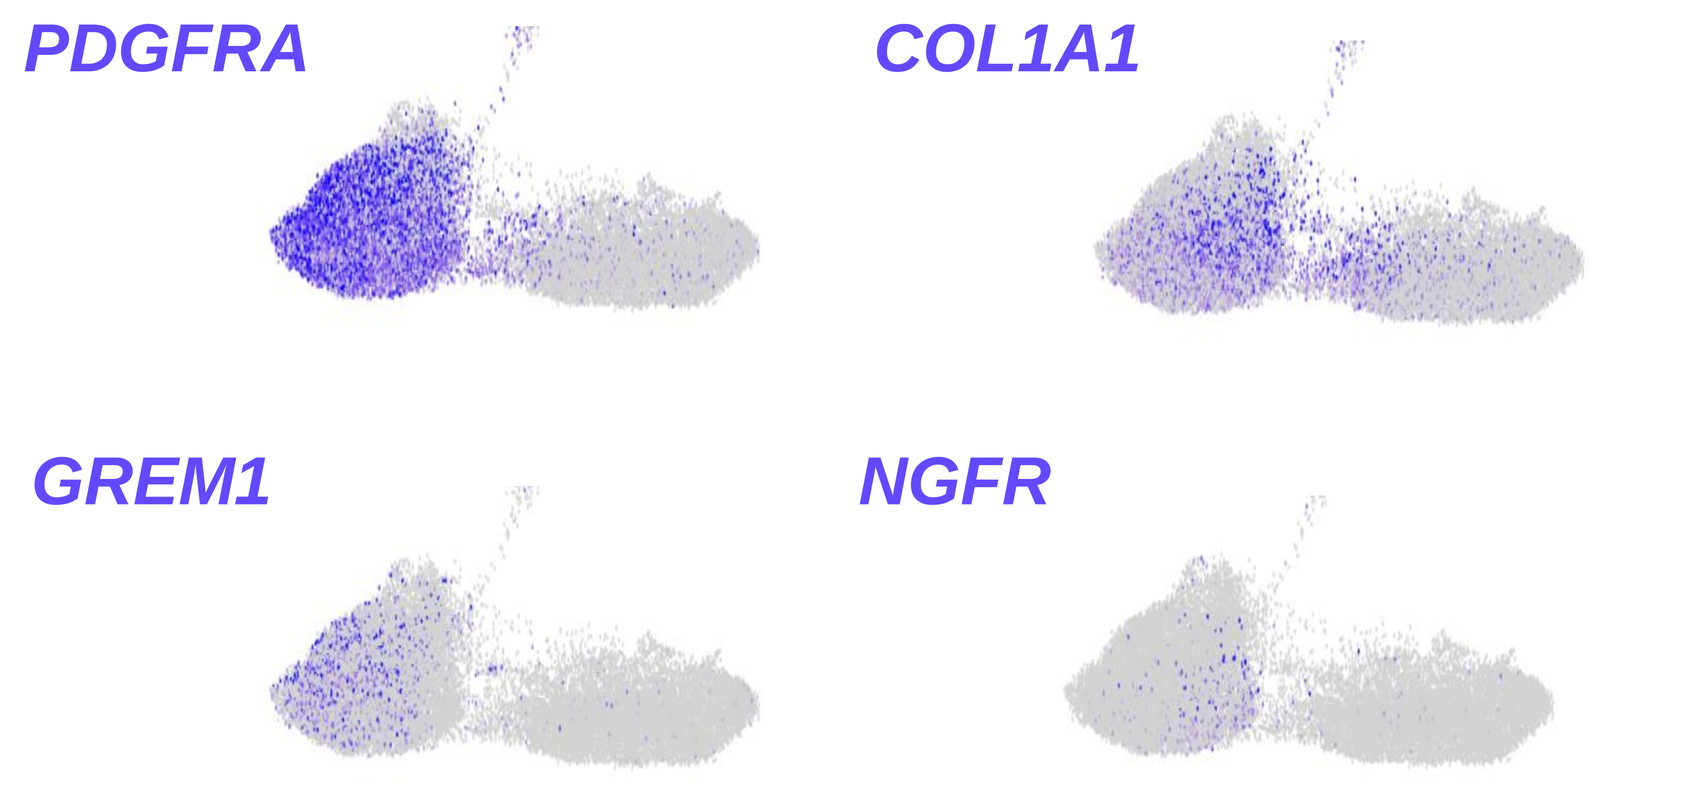
PDGFRA
COL1A1
GREM1
NGFR

UMAPs indicating *PDGFRA*, *COL1A1*, *GREM1*, and *NGFR* expression

- Myofibroblasts had high expression of *COL1A1* and were composed of activated portal fibroblasts (*GREM1*$^+$) and HSCs (*NGFR*$^+$)
- Differential gene expression analysis of bexotegrast-treated or ALK5i-treated versus vehicle-treated PCLivS showed downregulation of genes related to extracellular matrix and collagen fibril organization
- Bexotegrast significantly reduced *COL1A1* expression of myofibroblasts by >0.5 log2FC and at ~80% of the inhibition achieved with the ALK5i

Figure 6. Bexotegrast Treatment Significantly Decreased Profibrogenic Pathways in Myofibroblasts

A

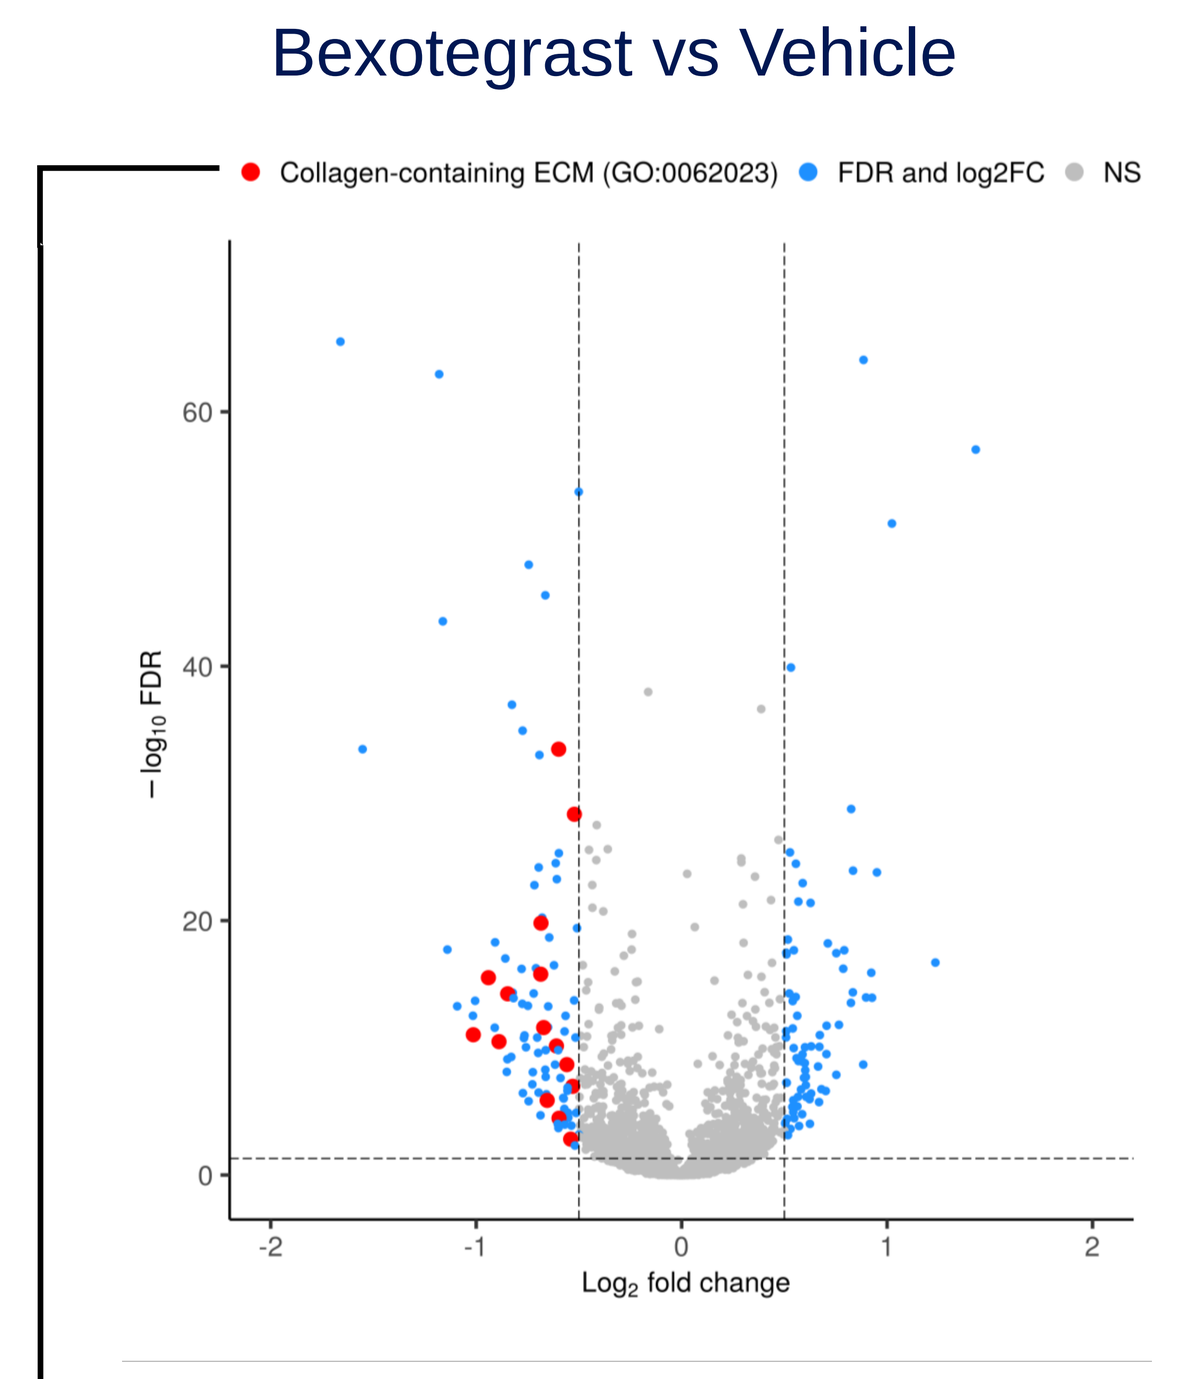
Bexotegrast vs Vehicle
ALK5i vs Vehicle
Collagen-containing ECM (GO:0062023)
FDR and log2FC
NS
Log2 fold change
−log10 FDR

COL16A1, *BGN*, *FBLN1*, *PCOLCE*, *LTBP2*, *AEPB1*, *SULF1*, *LOXL1*, *COL1A1*, *CCDC80*, *COL5A1*, *COL7A1*, *MGP*, *COL8A1*, *CTSC*

B

GO: Biological Process	Adj. P-Value
extracellular matrix organization (GO:0030198)	6.27E-05
extracellular structure organization (GO:0043062)	2.18E-03
external encapsulating structure organization (GO:0045229)	2.18E-03
collagen fibril organization (GO:0030199)	5.98E-03

C

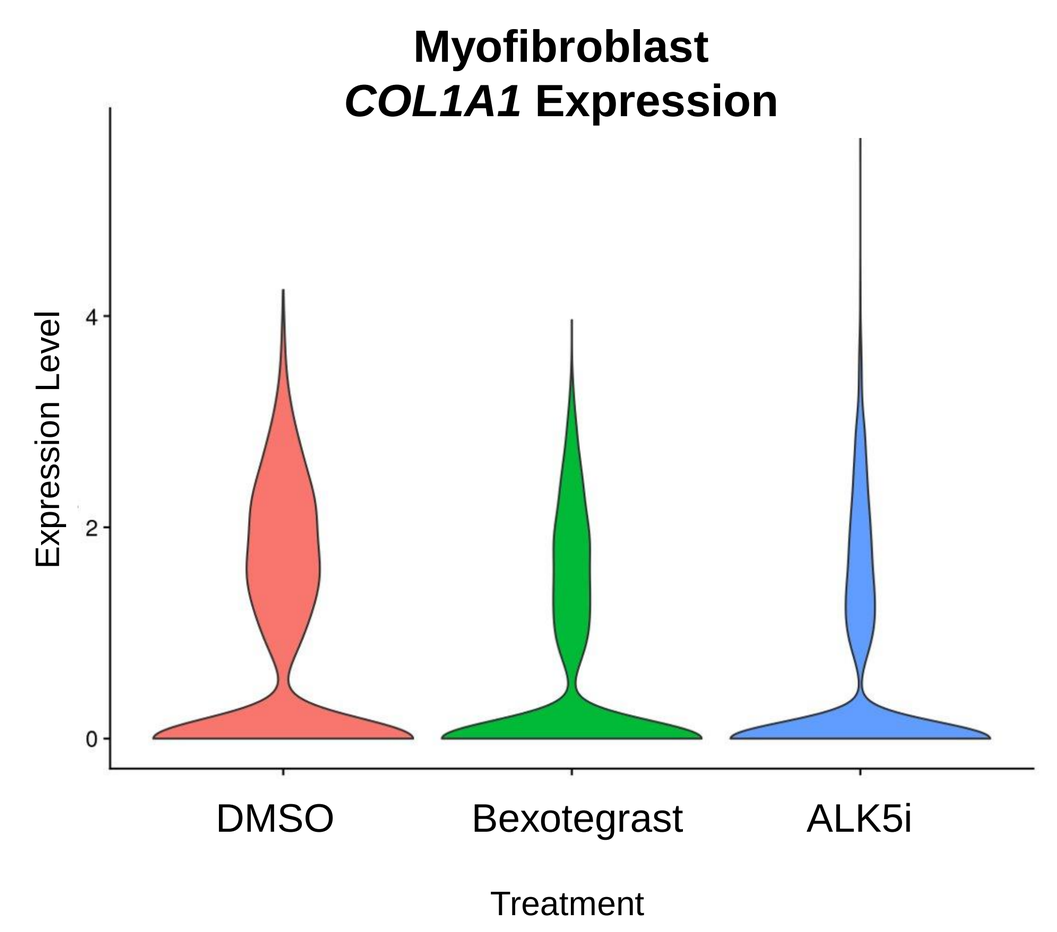
Myofibroblast
COL1A1 Expression
Expression Level
DMSO
Bexotegrast
ALK5i
Treatment

(A) Volcano plot of differentially expressed genes in bexotegrast-treated or ALK5i-treated vs vehicle-treated comparison in myofibroblasts. Genes indicated in blue are |log2FC| > 0.5 and FDR < 0.05. Enriched gene list for the significantly downregulated collagen-containing extracellular matrix term is shown and highlighted in the volcano plot (red). (B) Select GO:biological process terms from pathway enrichment analysis of downregulated genes (|log2FC| > 0.5, FDR < 0.05). (C) Violin plot for *COL1A1* expression by treatment in myofibroblasts.

Figure 7. snRNA-seq Analysis Identified Scar-associated Endothelial Cells

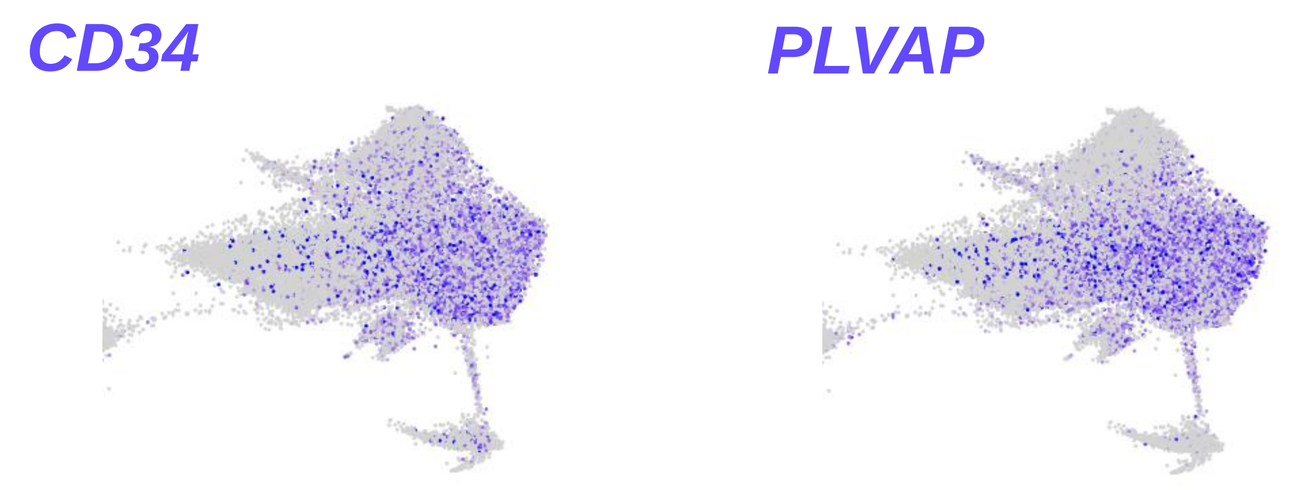
CD34
PLVAP

UMAPs indicating *CD34* and *PLVAP* expression

- Scar-associated endothelial cells were identified as a *CD34*$^+$*PLVAP*$^+$ subcluster

Figure 8. Bexotegrast Treatment Significantly Reduced Profibrogenic Gene Expression in Scar-associated Endothelial Cells

A

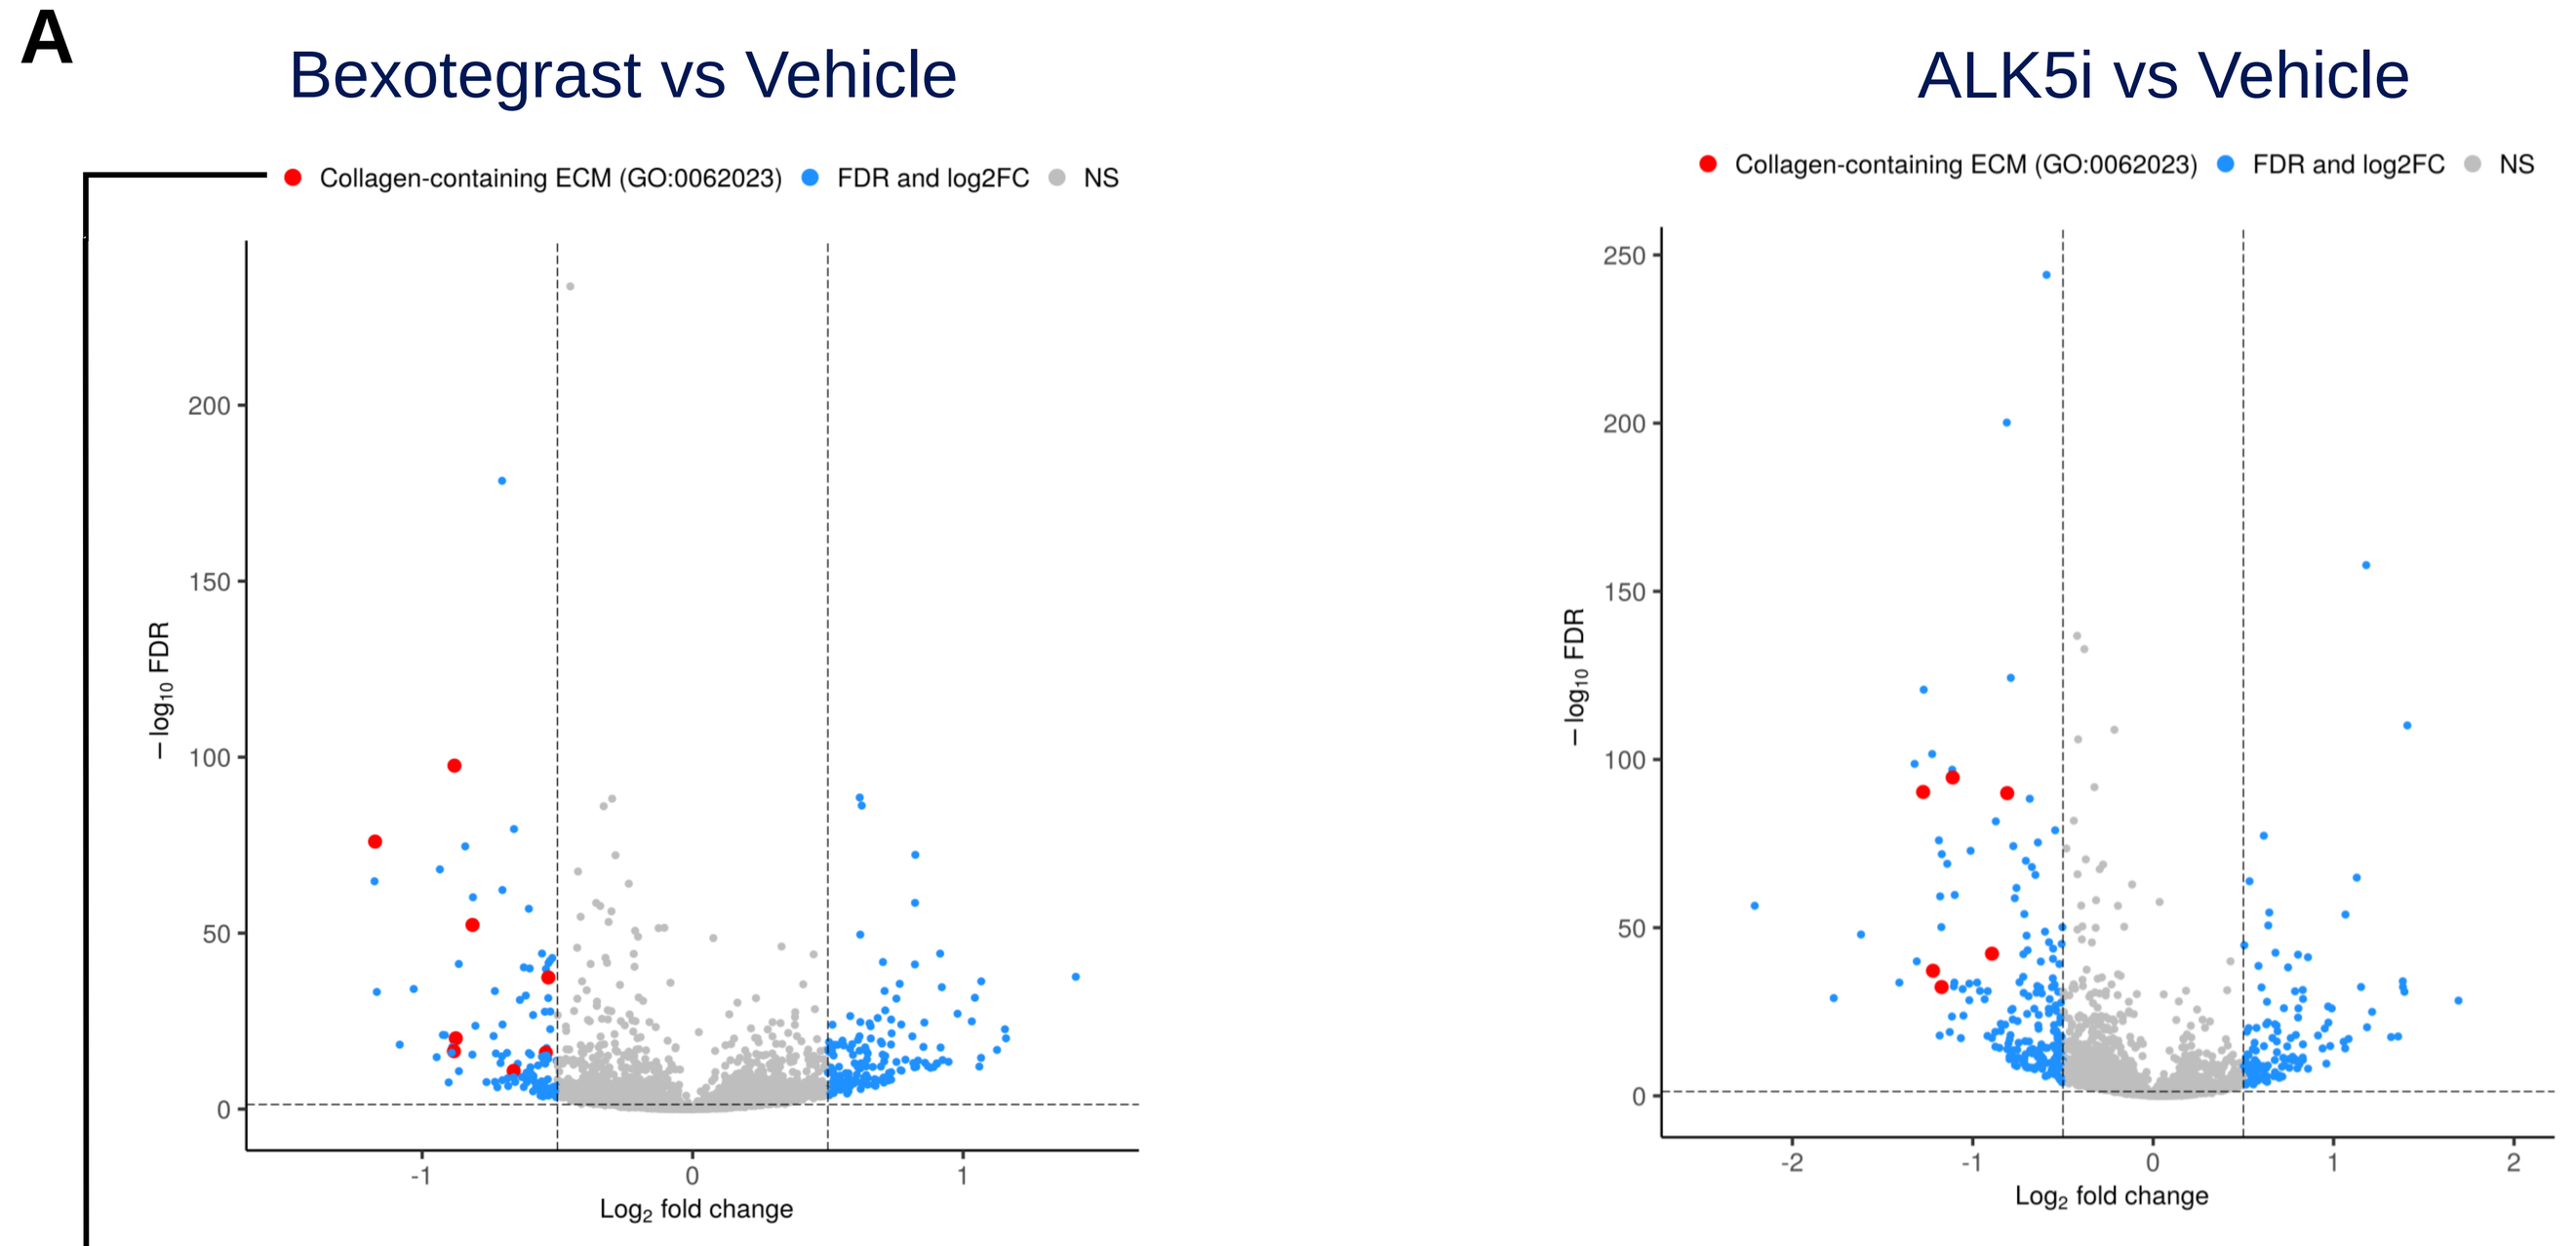
Bexotegrast vs Vehicle
ALK5i vs Vehicle
Collagen-containing ECM (GO:0062023)
FDR and log2FC
NS
Log2 fold change
−log10 FDR

COL27A1, *ADAMTS3*, *SRPX2*, *VWF*, *PDGFB*, *HTRA1*, *PLAT*, *NID2*

B

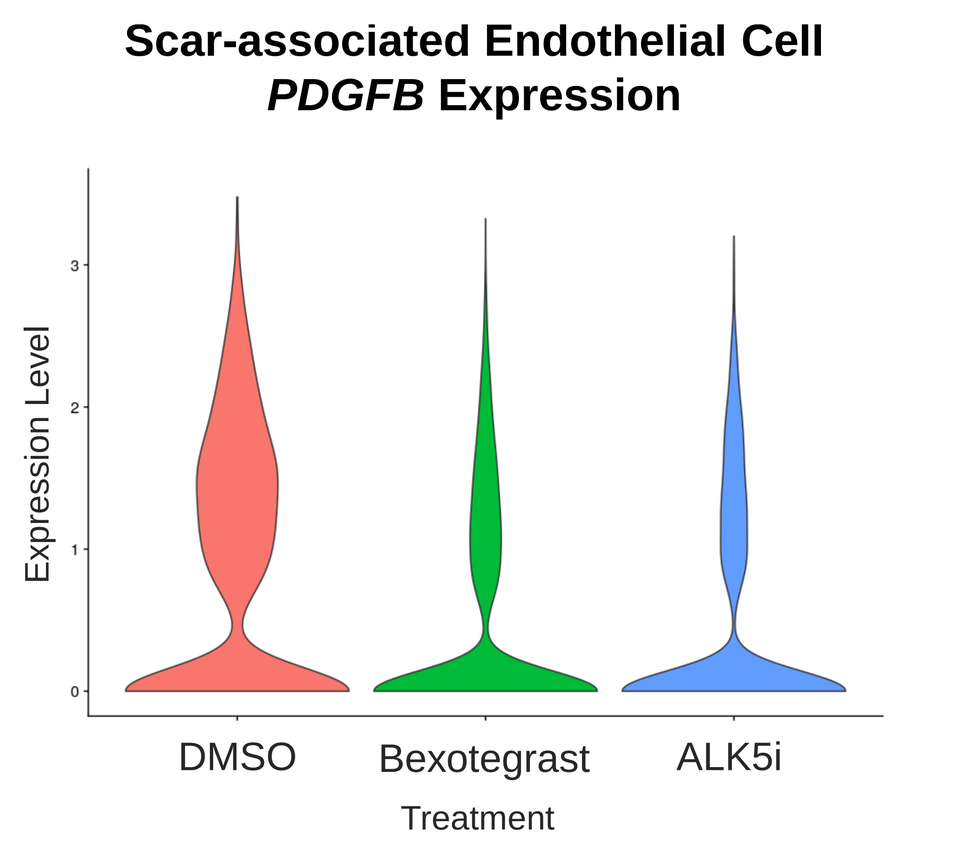
Scar-associated Endothelial Cell
PDGFB Expression
Expression Level
DMSO
Bexotegrast
ALK5i
Treatment

(A) Volcano plot of differentially expressed genes in bexotegrast-treated or ALK5i-treated vs vehicle-treated comparison in scar-associated endothelial cells. Genes indicated in blue are |log2FC| > 0.5 and FDR < 0.05. Enriched gene list for the significantly downregulated collagen-containing extracellular matrix term is shown and highlighted in the volcano plot (red). (B) Violin plot for *PDGFB* expression by treatment in scar-associated endothelial cells.

- Differential gene expression analysis of bexotegrast-treated or ALK5i-treated versus vehicle-treated PCLivS showed downregulation of genes related to extracellular matrix
- Bexotegrast treatment significantly reduced expression of *PDGFB*, suggesting a disruption of profibrogenic endothelial to myofibroblast interactions

CONCLUSIONS

- **Bexotegrast treatment resulted in clear reductions in profibrogenic gene expression across multiple pathologic cell populations in PCLivS from liver explants with biliary fibrosis**
- **The anti-fibrotic effect from bexotegrast was similar to ALK5i demonstrating the importance the $\alpha_v\beta_6/\alpha_v\beta_1$ integrin-TGF-β activation pathway in fibrotic biliary disease**
- **These data support ongoing clinical studies evaluating the anti-fibrotic effects of bexotegrast in PSC**

References: 1. Ramachandran P, et al. Nature 2019; 575(7783), 2. Andrews TS, et al. Hepatol Commun 2022; 6(4). 3. Chen EY, et al. BMC Bioinformatics 2013; 128(14).

Contact information: JSchaub@pliantrx.com

Miscellaneous: Copyright © 2023 Pliant Therapeutics, Inc. | All Rights Reserved

Disclosures: SH, CH, RA, VR, JL, MA, ST, MD, and JS are employees and shareholders of Pliant Therapeutics