Selective targeting of integrins αVß8 and αVß1 within the dynamic ecosystem of pancreatic cancer to improve overall anti-tumor response

Diego Chacon Fajardo, PhD[1,2], Dannielle H Upton, PhD[1,2], Chad L Moore, PhD[1], Sean Porazinski, PhD[1,2], Benjamin McLean[1,2], Eve Borbilas[1], Australian Pancreatic Cancer Genome Initiative[3], Vishal Kothari, PhD[4], Darren Finkelstein, PhD[4], Tim Machajewski, PhD[4], Fernando Rock, PhD[4], Scott M Turner, PhD[4] and **Marina Pajic, PhD**[1,2]

[1] Garvan Institute of Medical Research, Sydney, NSW, Australia [2] University of New South Wales, Sydney, NSW, Australia [3] http://www.pancreaticcancer.net.au [4] Pliant Therapeutics, South San Francisco, CA

Background

- Pancreatic ductal adenocarcinoma (PDA) has a 5-year survival of less than 10% and remains the 3rd leading cause of cancer-related death in Western societies. New treatment options are urgently needed.
- We previously characterized molecular subsets of PDA, including fibrotic elements of the disease, associated with pre-clinical and clinical response to select tailored treatment strategies[1-4].
- TGF-β promotes stromal cell reprogramming, immunosuppression, and fibrinogenesis in cancers, including PDA[5,6]. Integrins αVβ8 and αVβ1 are important activators of TGF-β signaling. Selective integrin blockade has recently emerged as a promising therapeutic approach to address TGFβ-mediated immunotherapy resistance, and improve anti-tumor response across cancer models[7-9].
- PLN-101095, a selective small molecule inhibitor of αVβ8 and αVβ1, is in early Phase I trials. Here, we assessed the preclinical efficacy and potential of PLN-101095 for the treatment of PDA.

Results

PLN-101095 mediated targeting of αVβ8/ αVβ1 improves chemoresponse in a FOLFIRINOX resistant patient-derived PDA setting

Development of a PDA model of FOLFIRINOX resistance

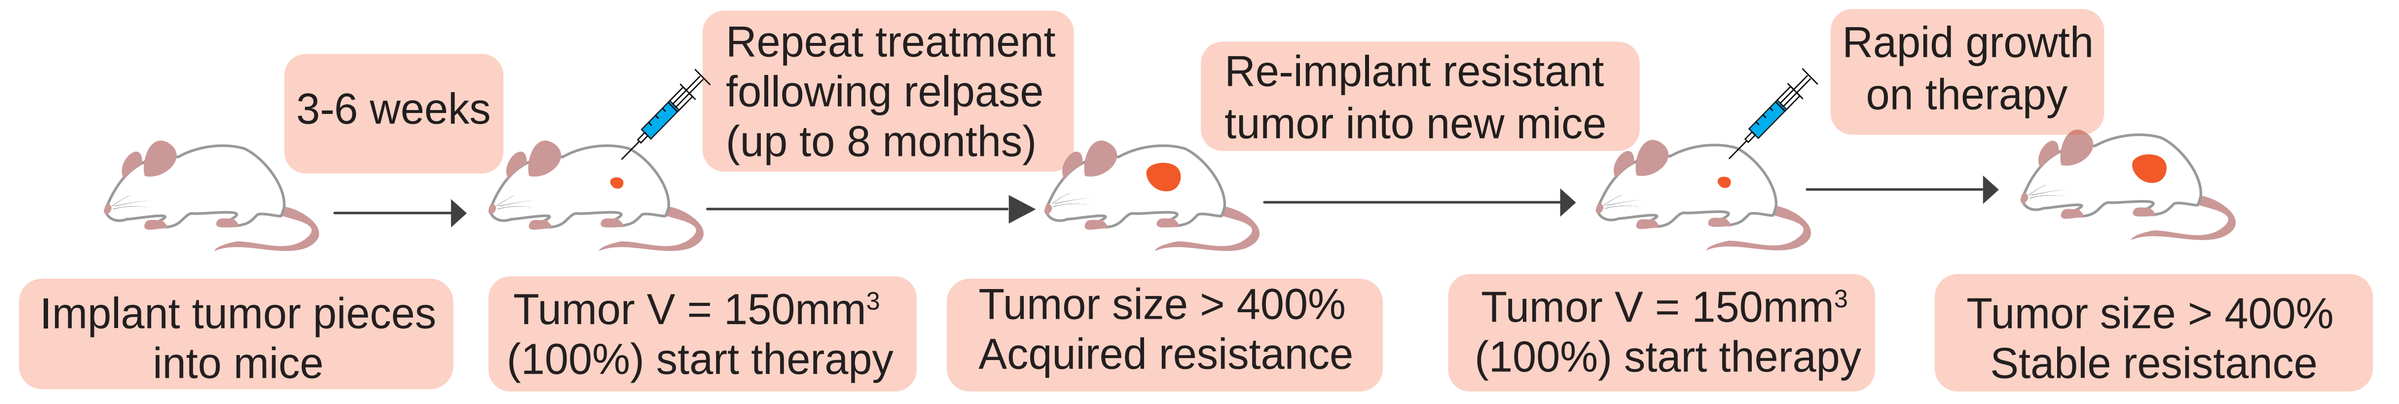
3-6 weeks
Repeat treatment following relpase (up to 8 months)
Re-implant resistant tumor into new mice
Rapid growth on therapy
Implant tumor pieces into mice
Tumor V = 150mm³ (100%) start therapy
Tumor size > 400% Acquired resistance
Tumor V = 150mm³ (100%) start therapy
Tumor size > 400% Stable resistance

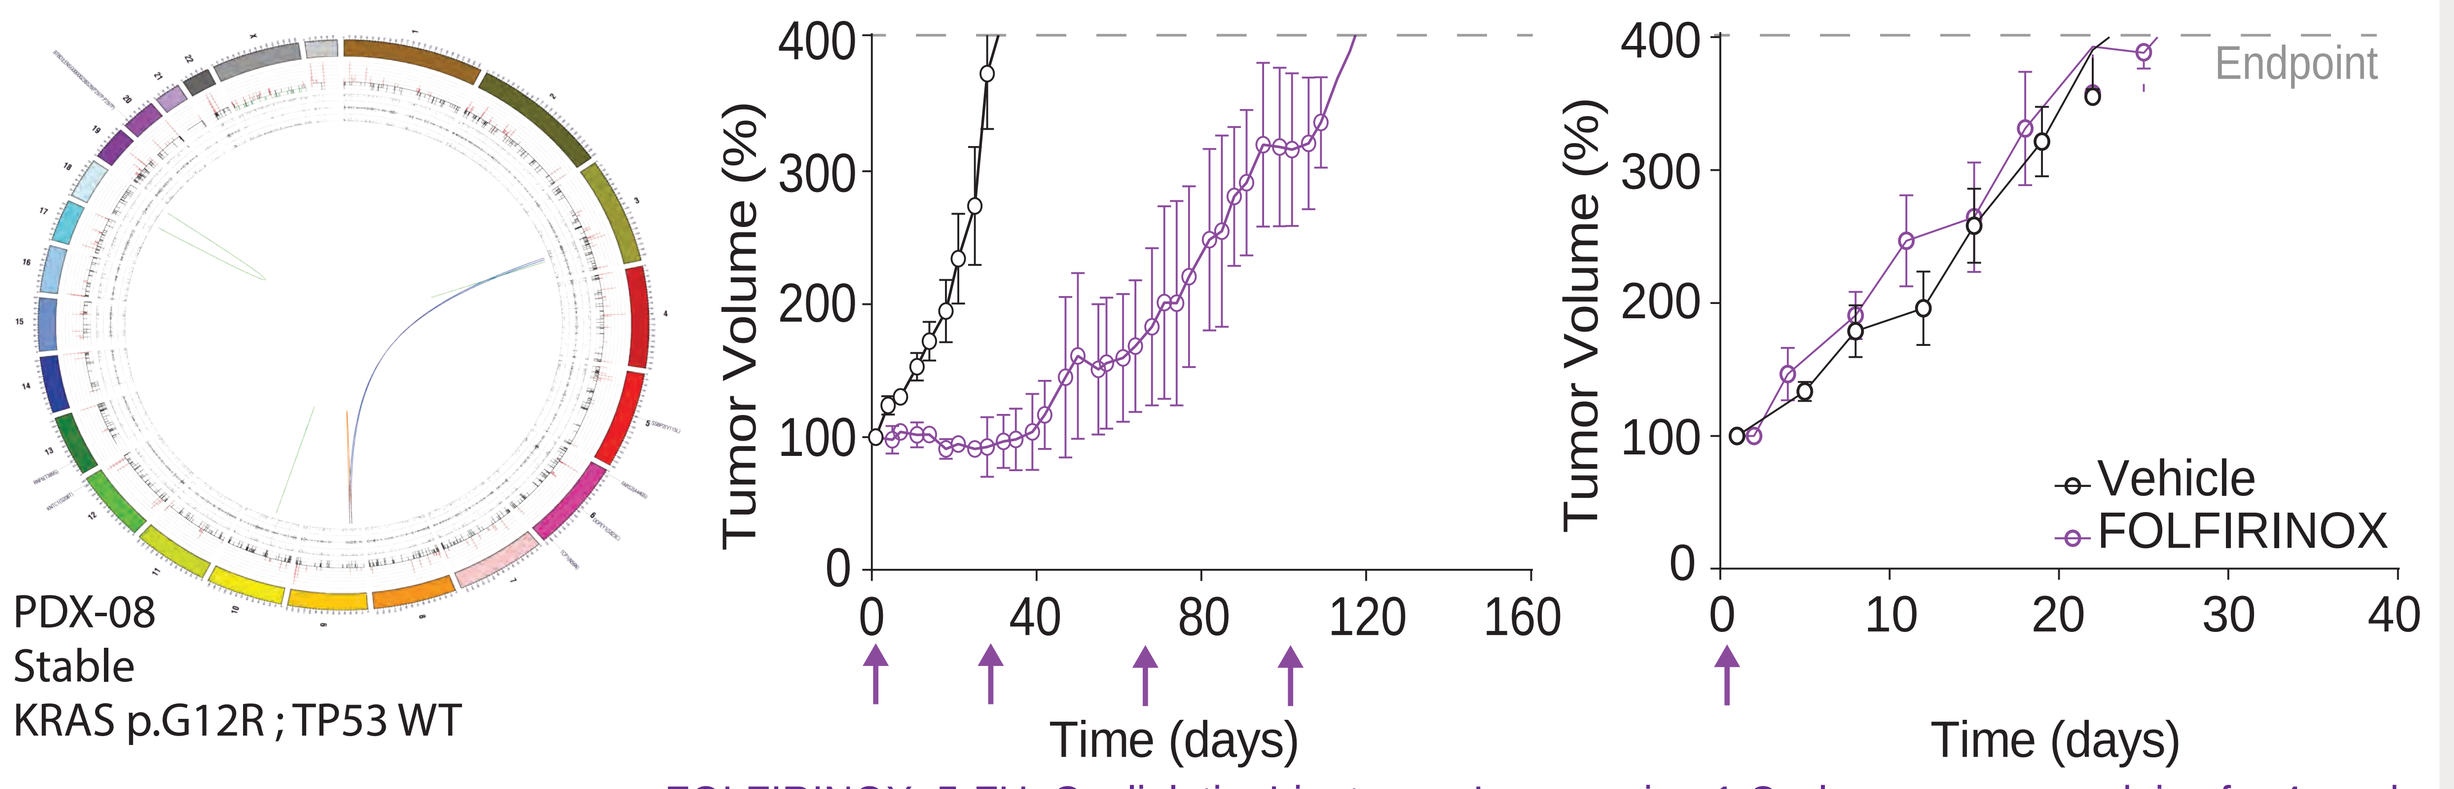
PDX-08
Stable
KRAS p.G12R ; TP53 WT
Tumor Volume (%)
Time (days)
Endpoint
Vehicle
FOLFIRINOX

FOLFIRINOX: 5-FU, Oxaliplatin, Irinotecan, Leucovorin - 1 Cycle: once per week i.v. for 4 weeks

- PLN-101095 + FOLFIRINOX combination delayed disease progression and significantly reduced in vivo tumor growth in a PDA model of FOLFIRINOX resistance (*p<0.05; **p<0.01; ***p<0.001).

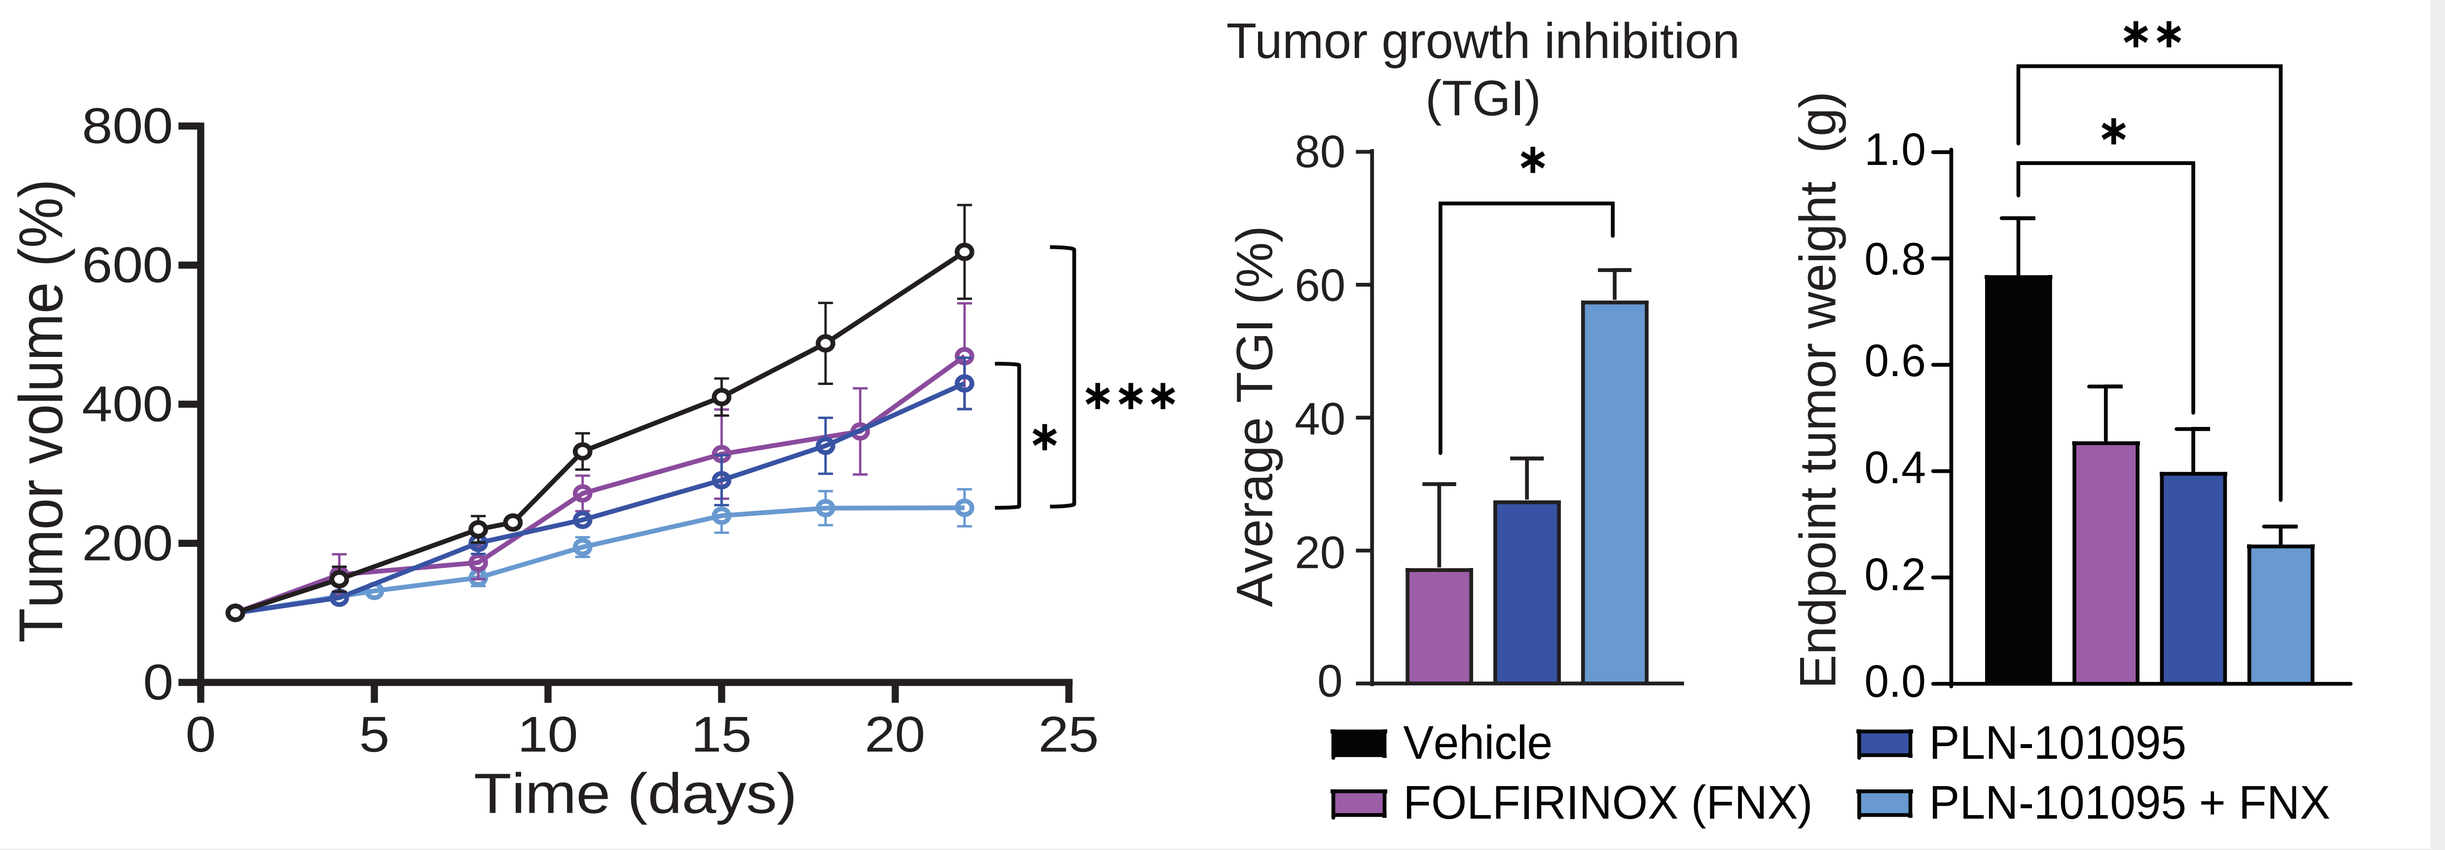
Tumor volume (%)
Time (days)
Tumor growth inhibition (TGI)
Average TGI (%)
Endpoint tumor weight (g)
Vehicle
FOLFIRINOX (FNX)
PLN-101095
PLN-101095 + FNX

PLN-101095 decreases primary tumor growth, improves chemosensitivity in metastatic PDA

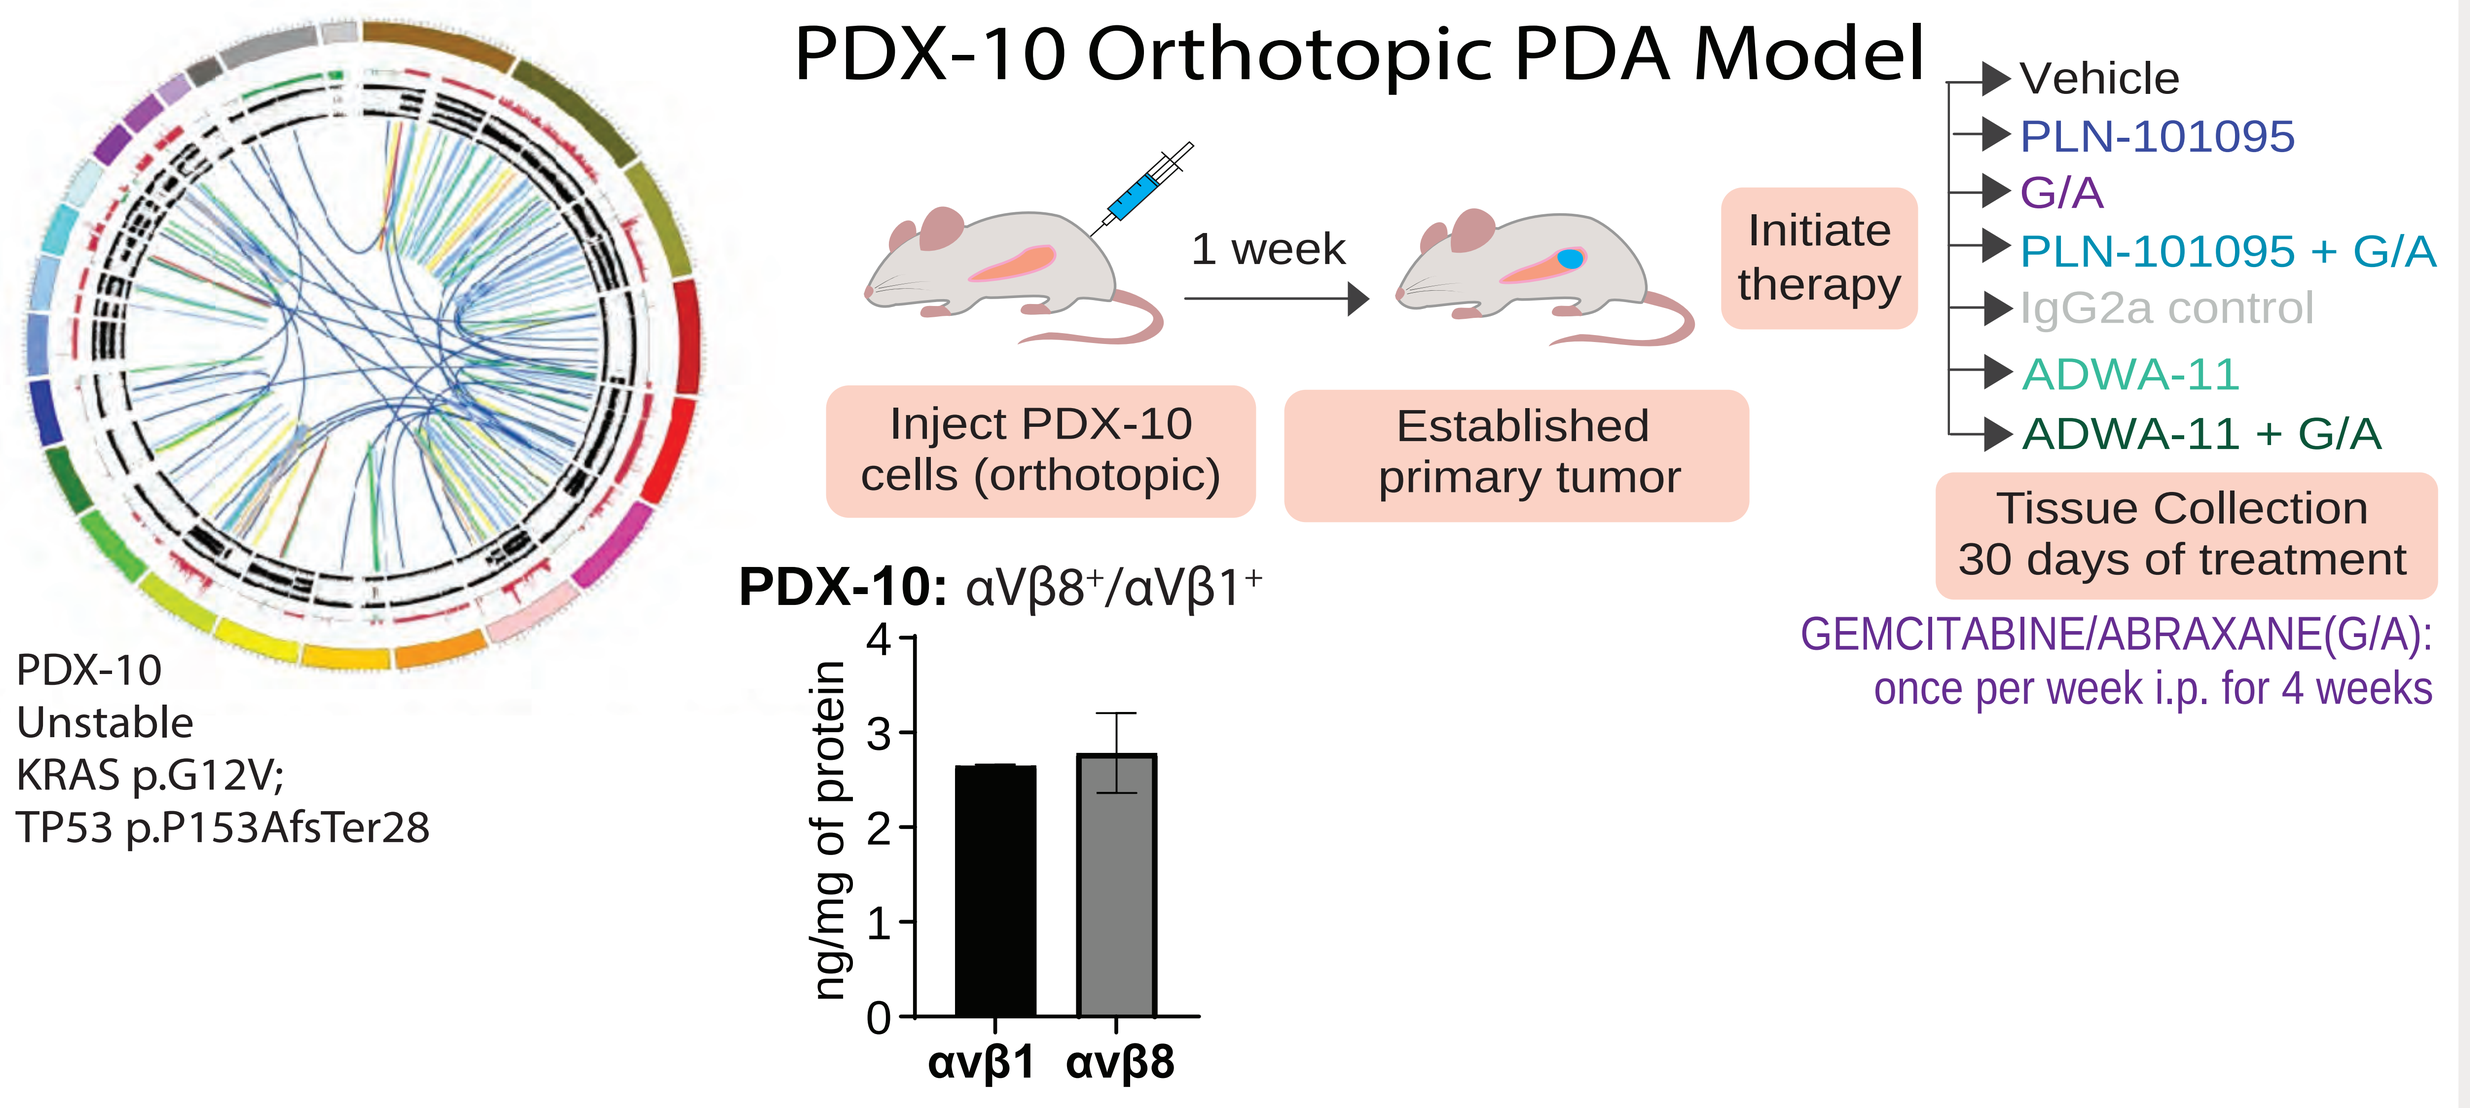
PDX-10 Orthotopic PDA Model
PDX-10
Unstable
KRAS p.G12V;
TP53 p.P153AfsTer28
1 week
Inject PDX-10 cells (orthotopic)
Established primary tumor
Initiate therapy
Vehicle
PLN-101095
G/A
PLN-101095 + G/A
IgG2a control
ADWA-11
ADWA-11 + G/A
Tissue Collection 30 days of treatment
GEMCITABINE/ABRAXANE(G/A): once per week i.p. for 4 weeks
PDX-10: αVβ8⁺/αVβ1⁺
ng/mg of protein
αvβ1
αvβ8

- Targeting αVβ8/ αVβ1 signaling with PLN-101095 significantly decreased primary tumor growth and improved efficacy of standard chemotherapy gemcitabine/Abraxane in a PDX model of metastatic PDA, reducing metastatic spread to the lungs.
- In contrast, ADWA-11, an αvβ8 blocking monoclonal antibody, did not elicit a similar response.

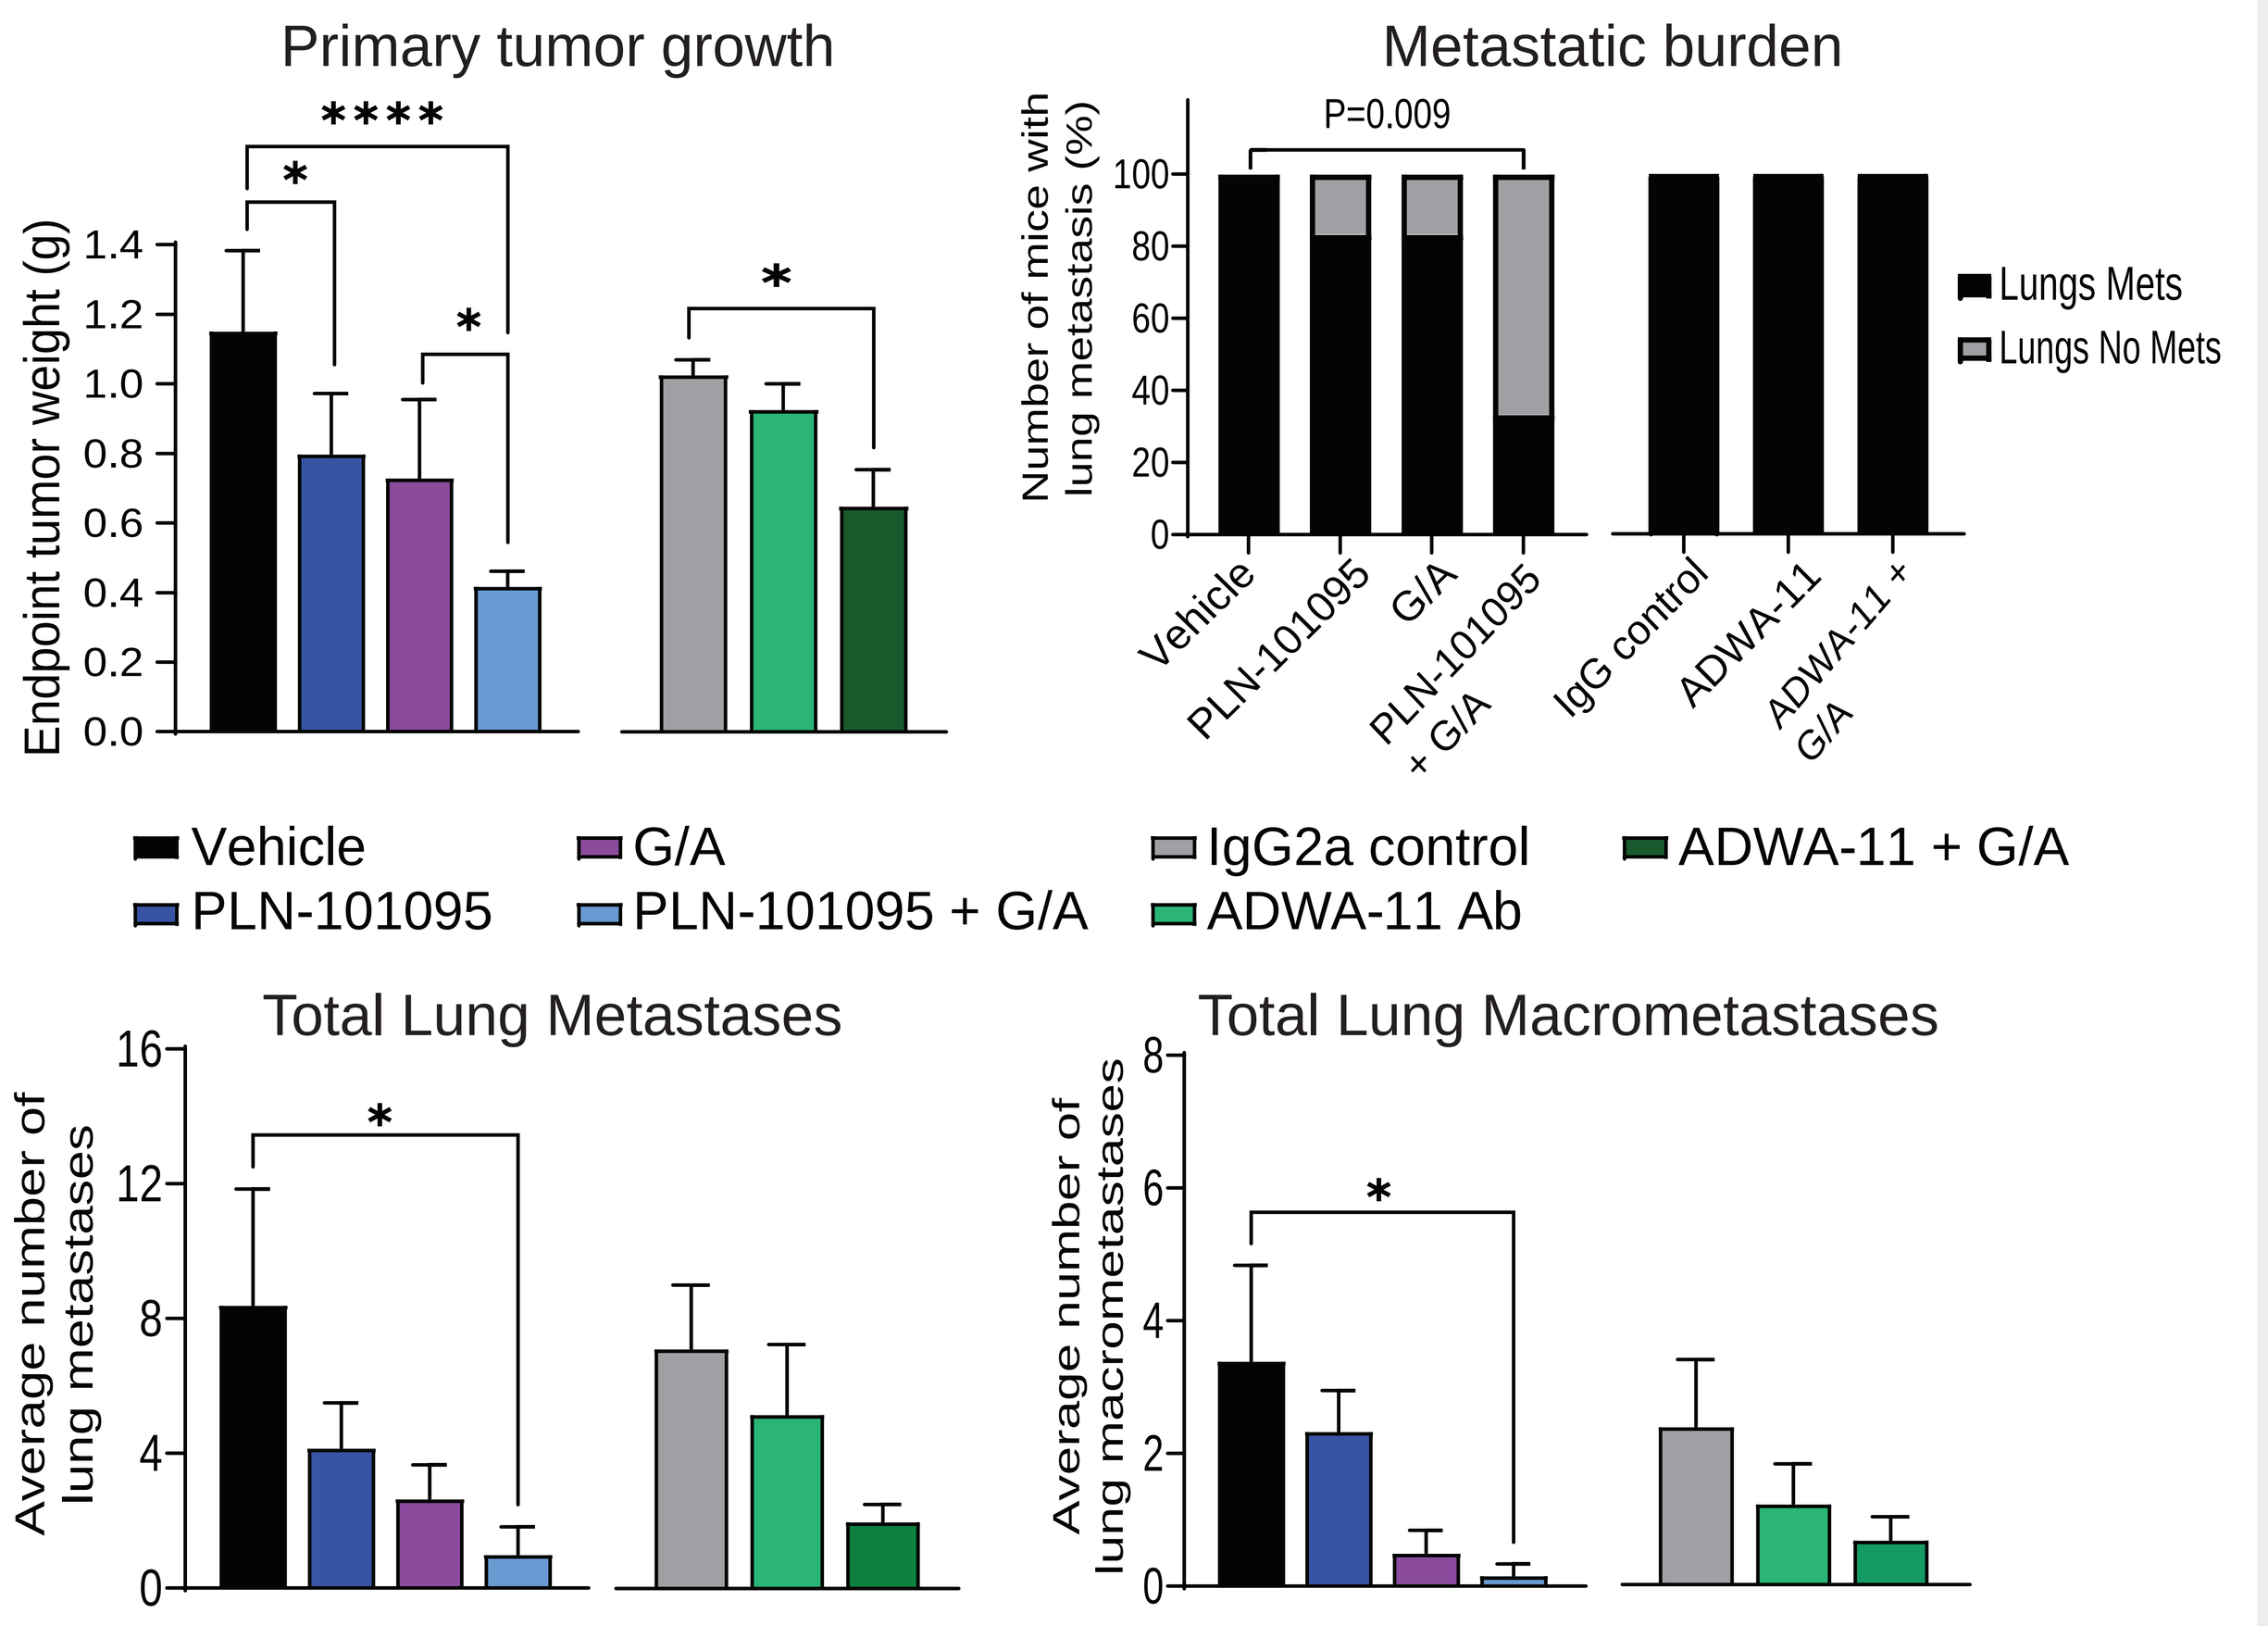
Primary tumor growth
Endpoint tumor weight (g)
Metastatic burden
Number of mice with lung metastasis (%)
P=0.009
Lungs Mets
Lungs No Mets
Vehicle
PLN-101095
G/A
PLN-101095 + G/A
IgG2a control
ADWA-11 Ab
ADWA-11 + G/A
Total Lung Metastases
Average number of lung metastases
Total Lung Macrometastases
Average number of lung macrometastases

PLN-101095 treatment reduces primary tumor growth and improves anti-PD1 response in a range of 'immune-cold' models of advanced PDA

Pan02 syngeneic PDA Model

- PLN-101095 improves response to immune checkpoint blockade (ICB) and improves intra-tumoral CD8+ lymphocyte infiltration.

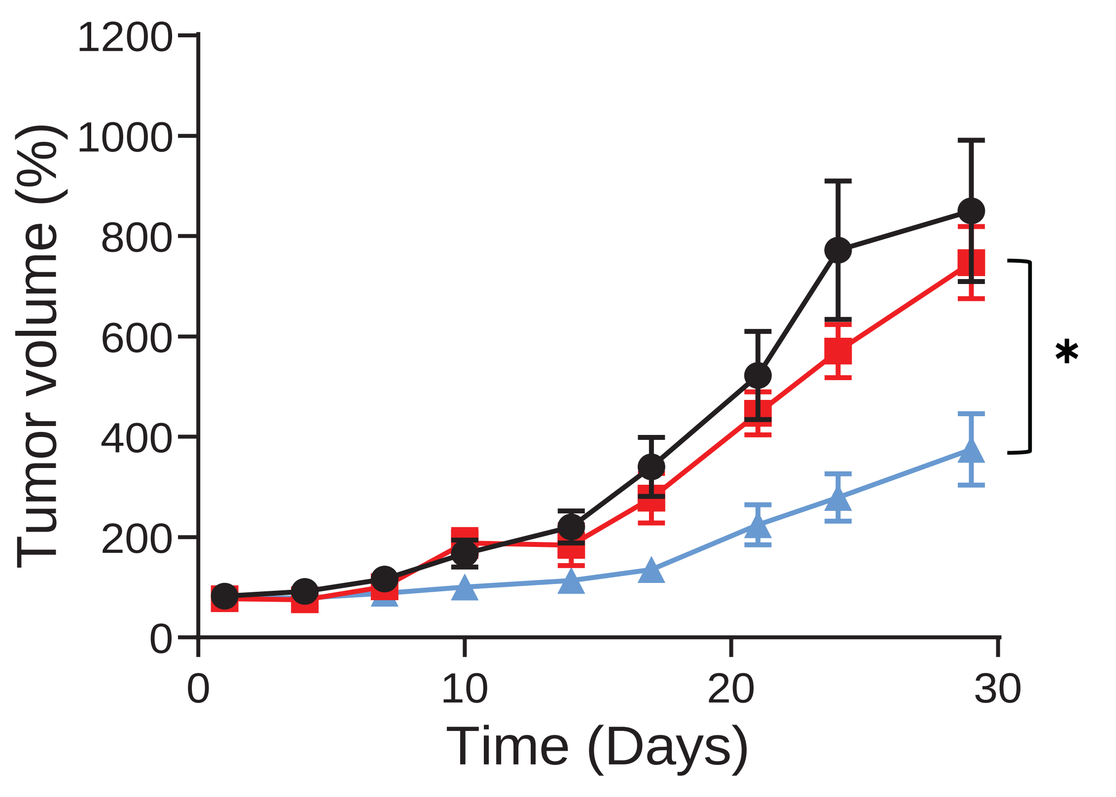
Tumor volume (%)
Time (Days)

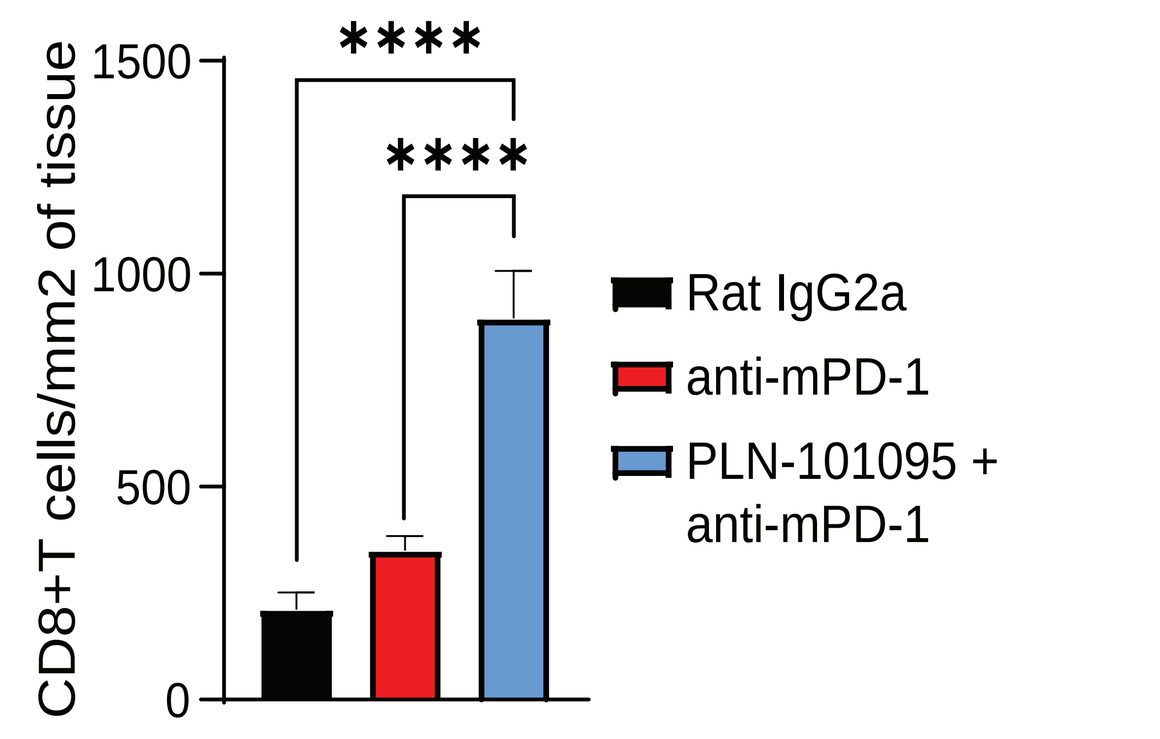
CD8+T cells/mm2 of tissue
Rat IgG2a
anti-mPD-1
PLN-101095 + anti-mPD-1

- PLN-101095 combined with ICB induces expression of MHC and IFN response genes in PDA tumors.

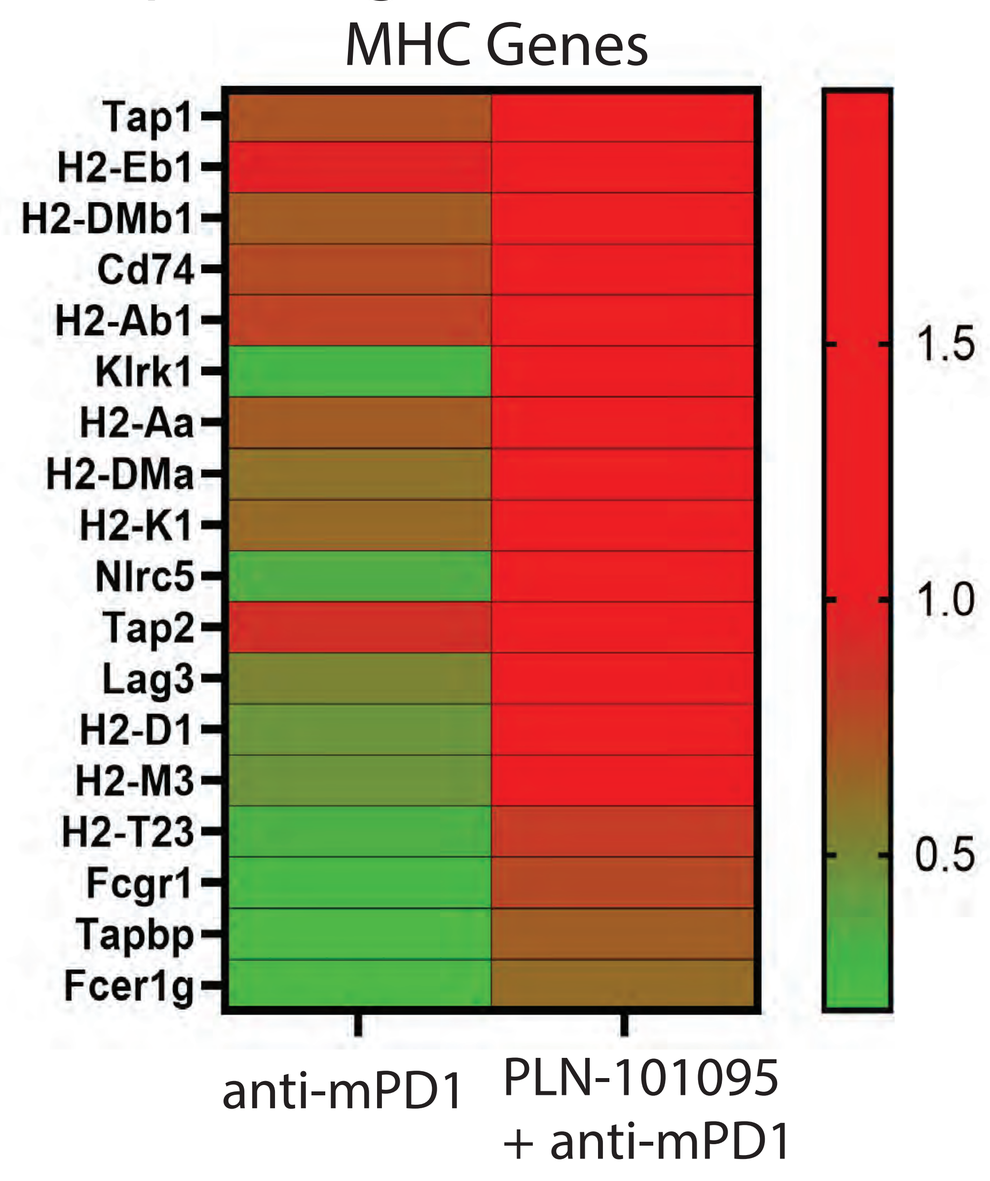
MHC Genes
Tap1
H2-Eb1
H2-DMb1
Cd74
H2-Ab1
Klrk1
H2-Aa
H2-DMa
H2-K1
Nlrc5
Tap2
Lag3
H2-D1
H2-M3
H2-T23
Fcgr1
Tapbp
Fcer1g
anti-mPD1
PLN-101095 + anti-mPD1

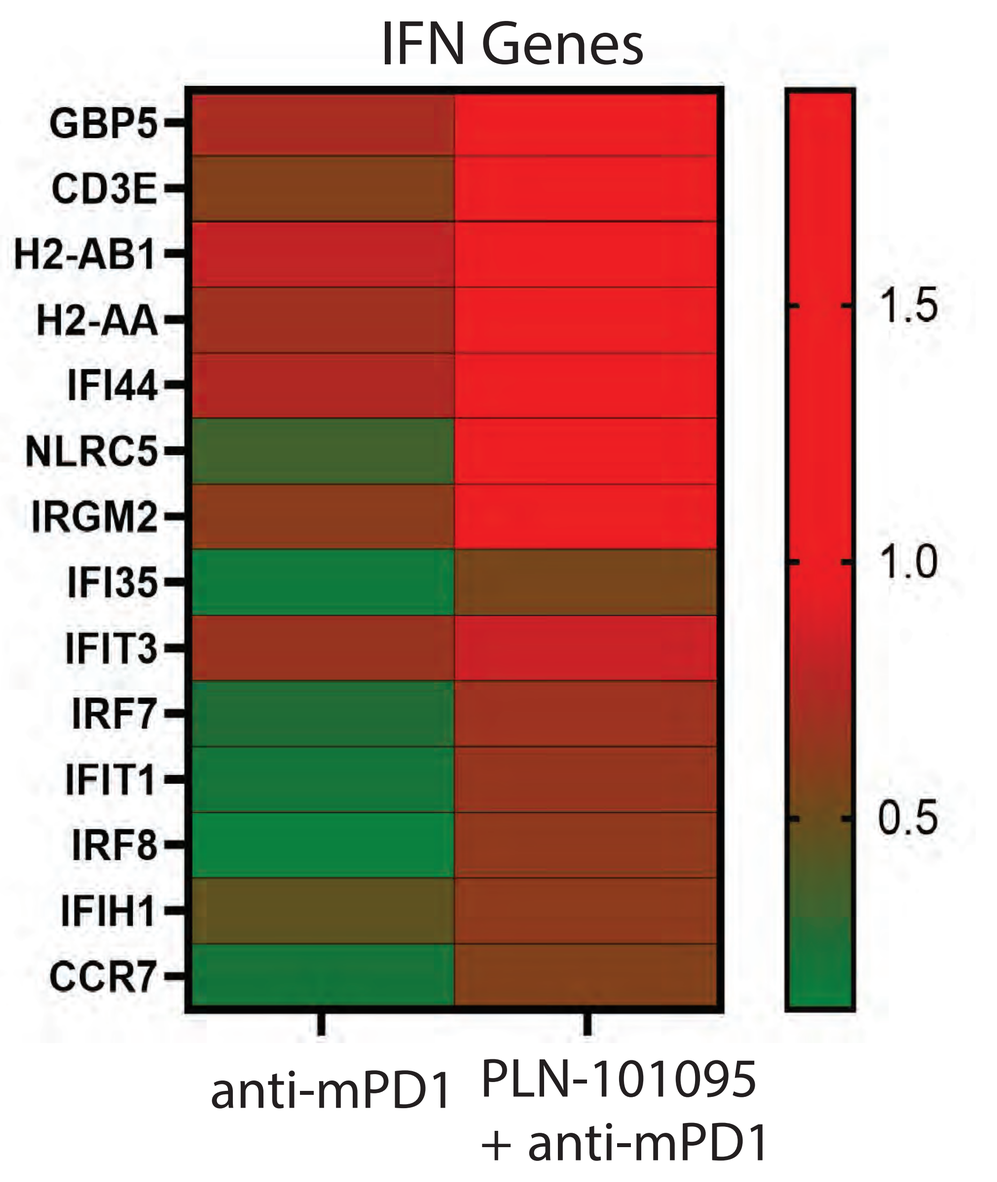
IFN Genes
GBP5
CD3E
H2-AB1
H2-AA
IFI44
NLRC5
IRGM2
IFI35
IFIT3
IRF7
IFIT1
IRF8
IFIH1
CCR7
anti-mPD1
PLN-101095 + anti-mPD1

LSL-Kras$^{G12D/+}$; LSL-Trp53$^{R172H/+}$; Pdx1-Cre (KPC) PDA Model

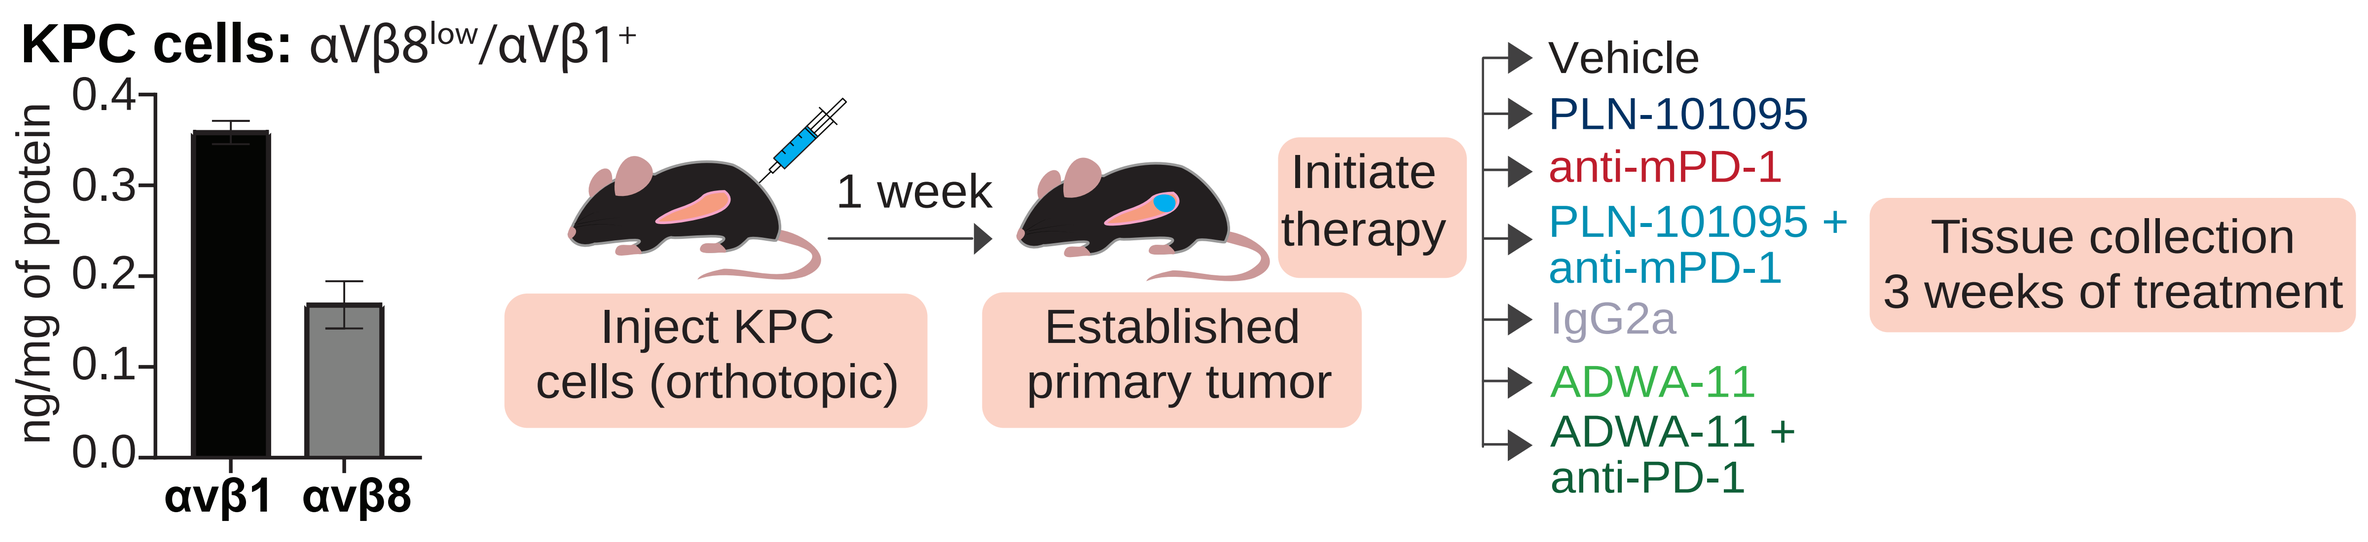
KPC cells: αVβ8^low/αVβ1⁺
ng/mg of protein
αvβ1
αvβ8
1 week
Inject KPC cells (orthotopic)
Established primary tumor
Initiate therapy
Vehicle
PLN-101095
anti-mPD-1
PLN-101095 + anti-mPD-1
IgG2a
ADWA-11
ADWA-11 + anti-PD-1
Tissue collection 3 weeks of treatment

- PLN-101095 monotherapy significantly delayed PDA progression in vivo, and significantly improved ICB response (22% CR observed).

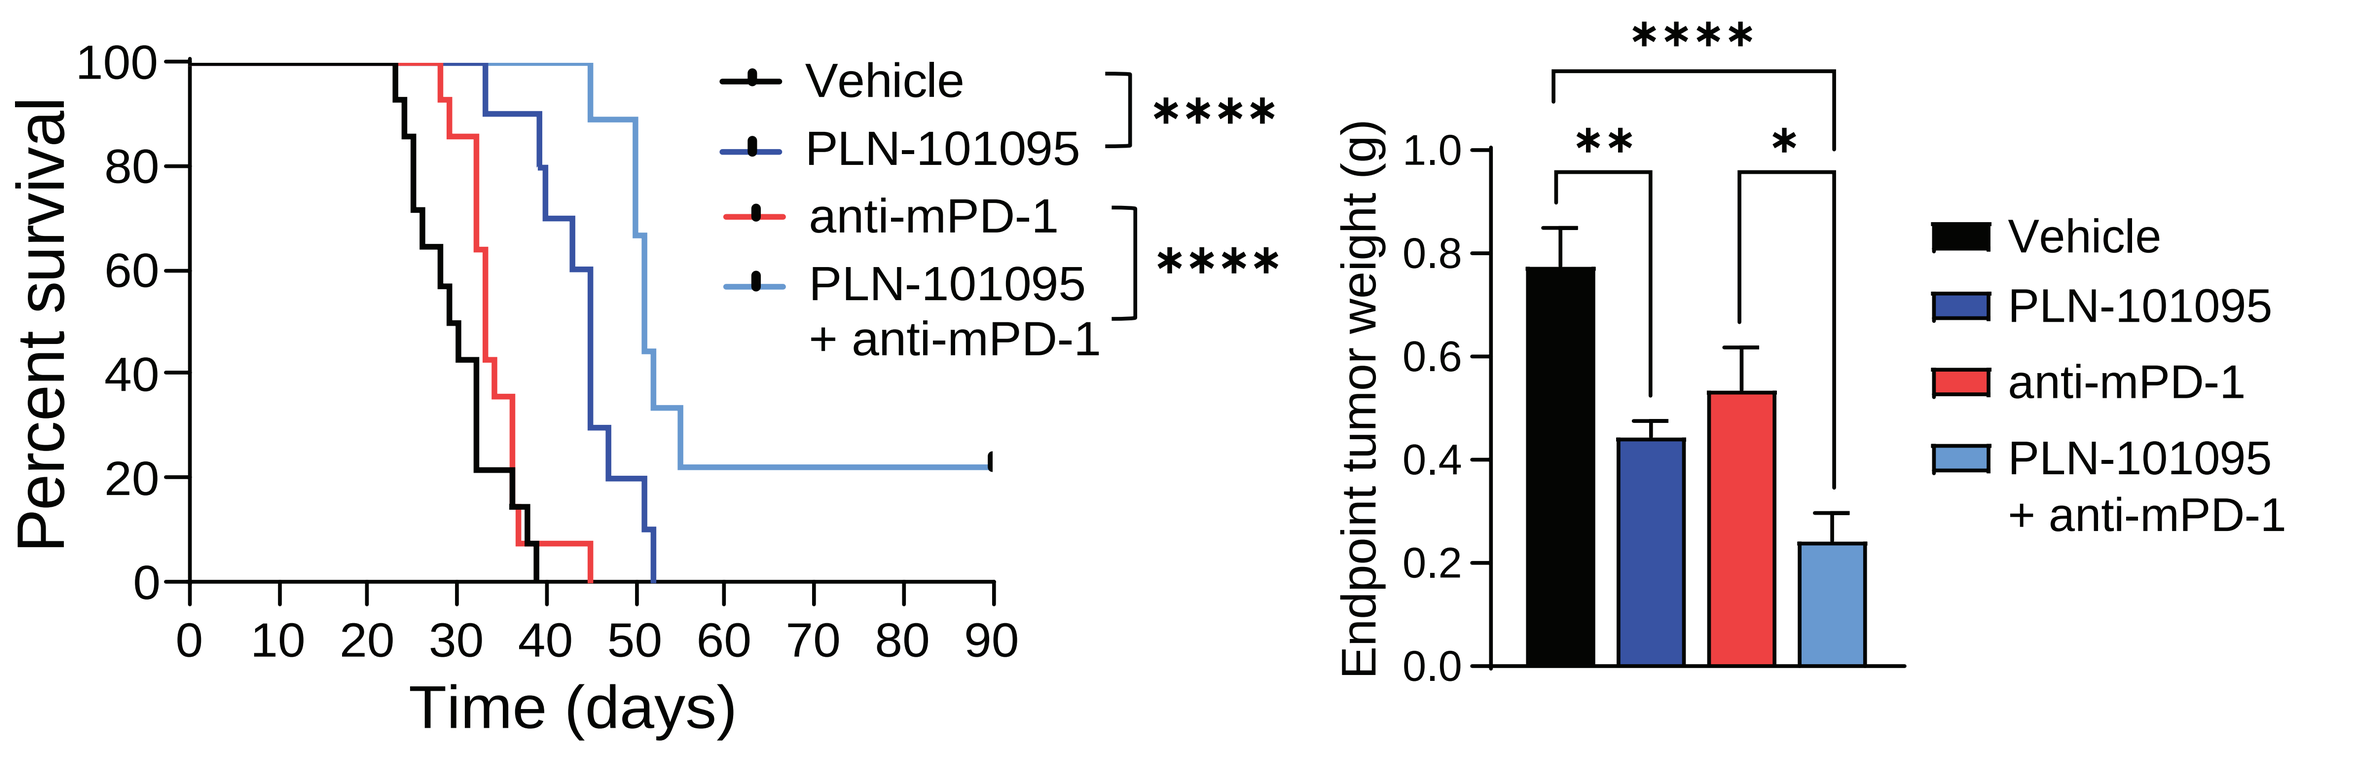
Percent survival
Time (days)
Vehicle
PLN-101095
anti-mPD-1
PLN-101095 + anti-mPD-1
Endpoint tumor weight (g)

- In comparison, blocking αvβ8 with ADWA-11 Ab elicited a modest response when combined with ICB in the KPC orthotopic PDA model.

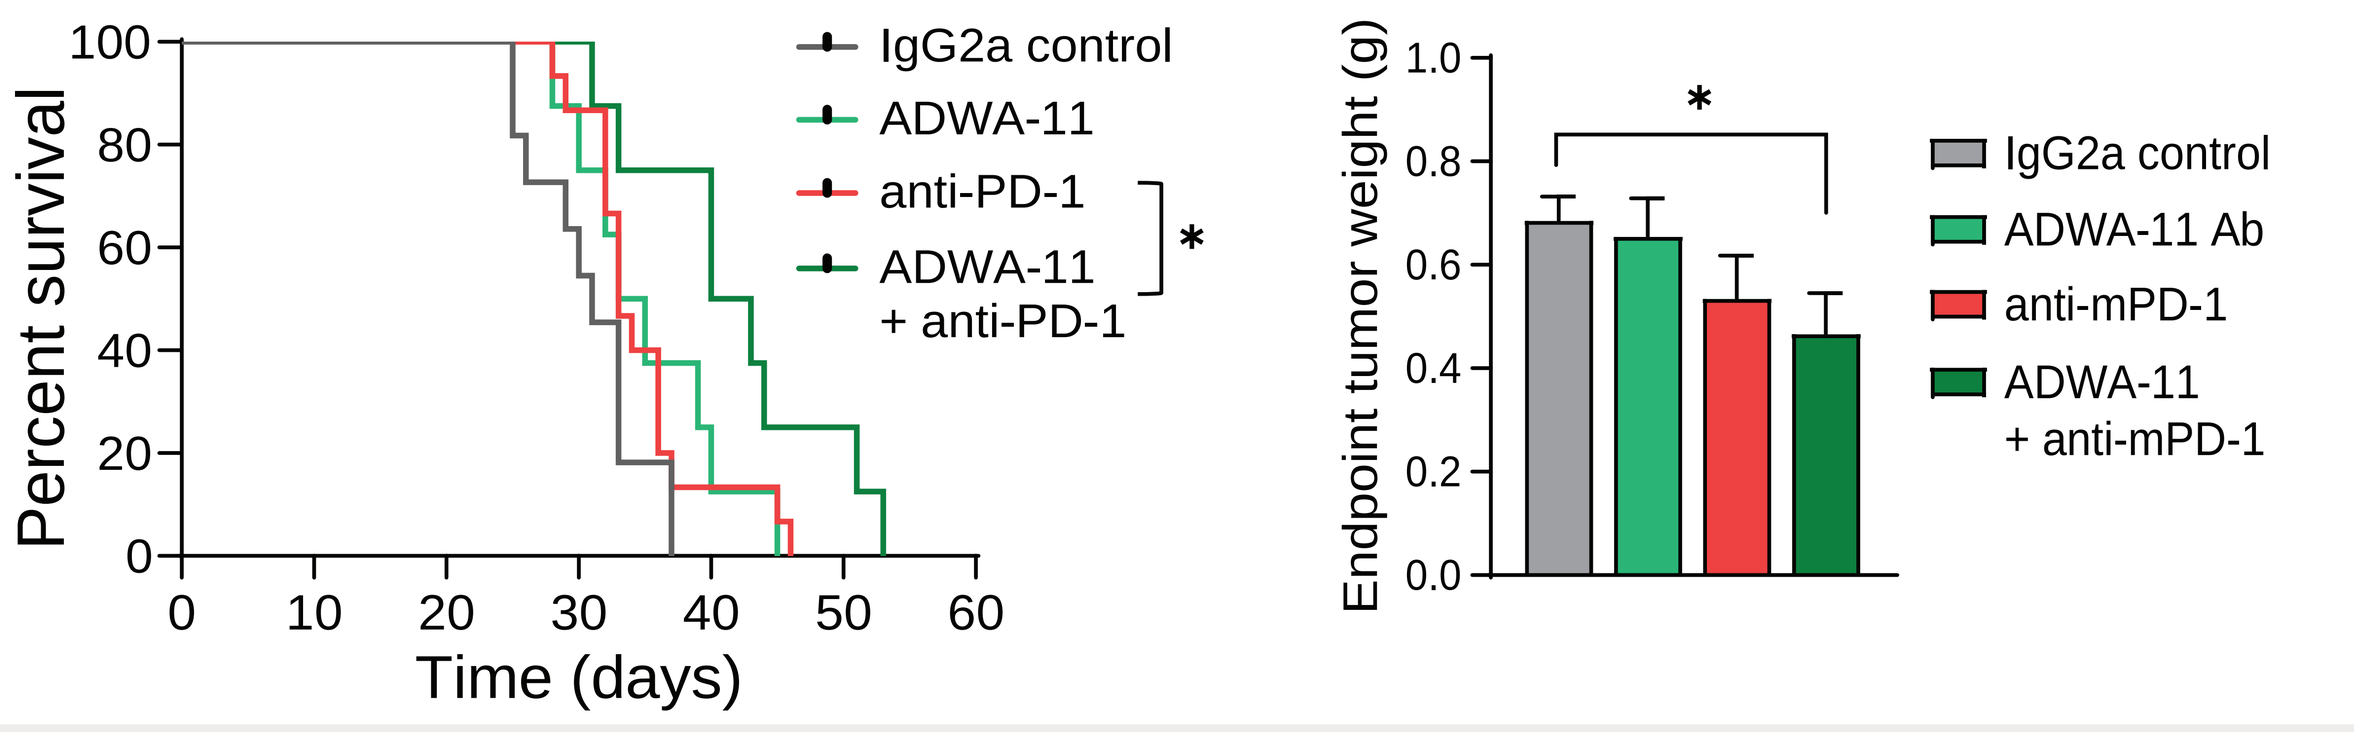
Percent survival
Time (days)
IgG2a control
ADWA-11
anti-PD-1
ADWA-11 + anti-PD-1
Endpoint tumor weight (g)
IgG2a control
ADWA-11 Ab
anti-mPD-1
ADWA-11 + anti-mPD-1

Charting the complex tumor landscape in the KPC model of PDA post-PLN-101095 therapy

- Dual targeting of αVß1/αVß8 induced a shift towards a more epithelial-like phenotype across diverse cancer cell subsets in murine PDA (KPC) tumors ('Thierry signature' Tan TZ et al EMBO Mol Med 2014), previously associated with improved prognosis in PDA.

Combined UMAP of scRNA-seq data from PLN-101095 or vehicle treated KPC mice (n=32521 cells)

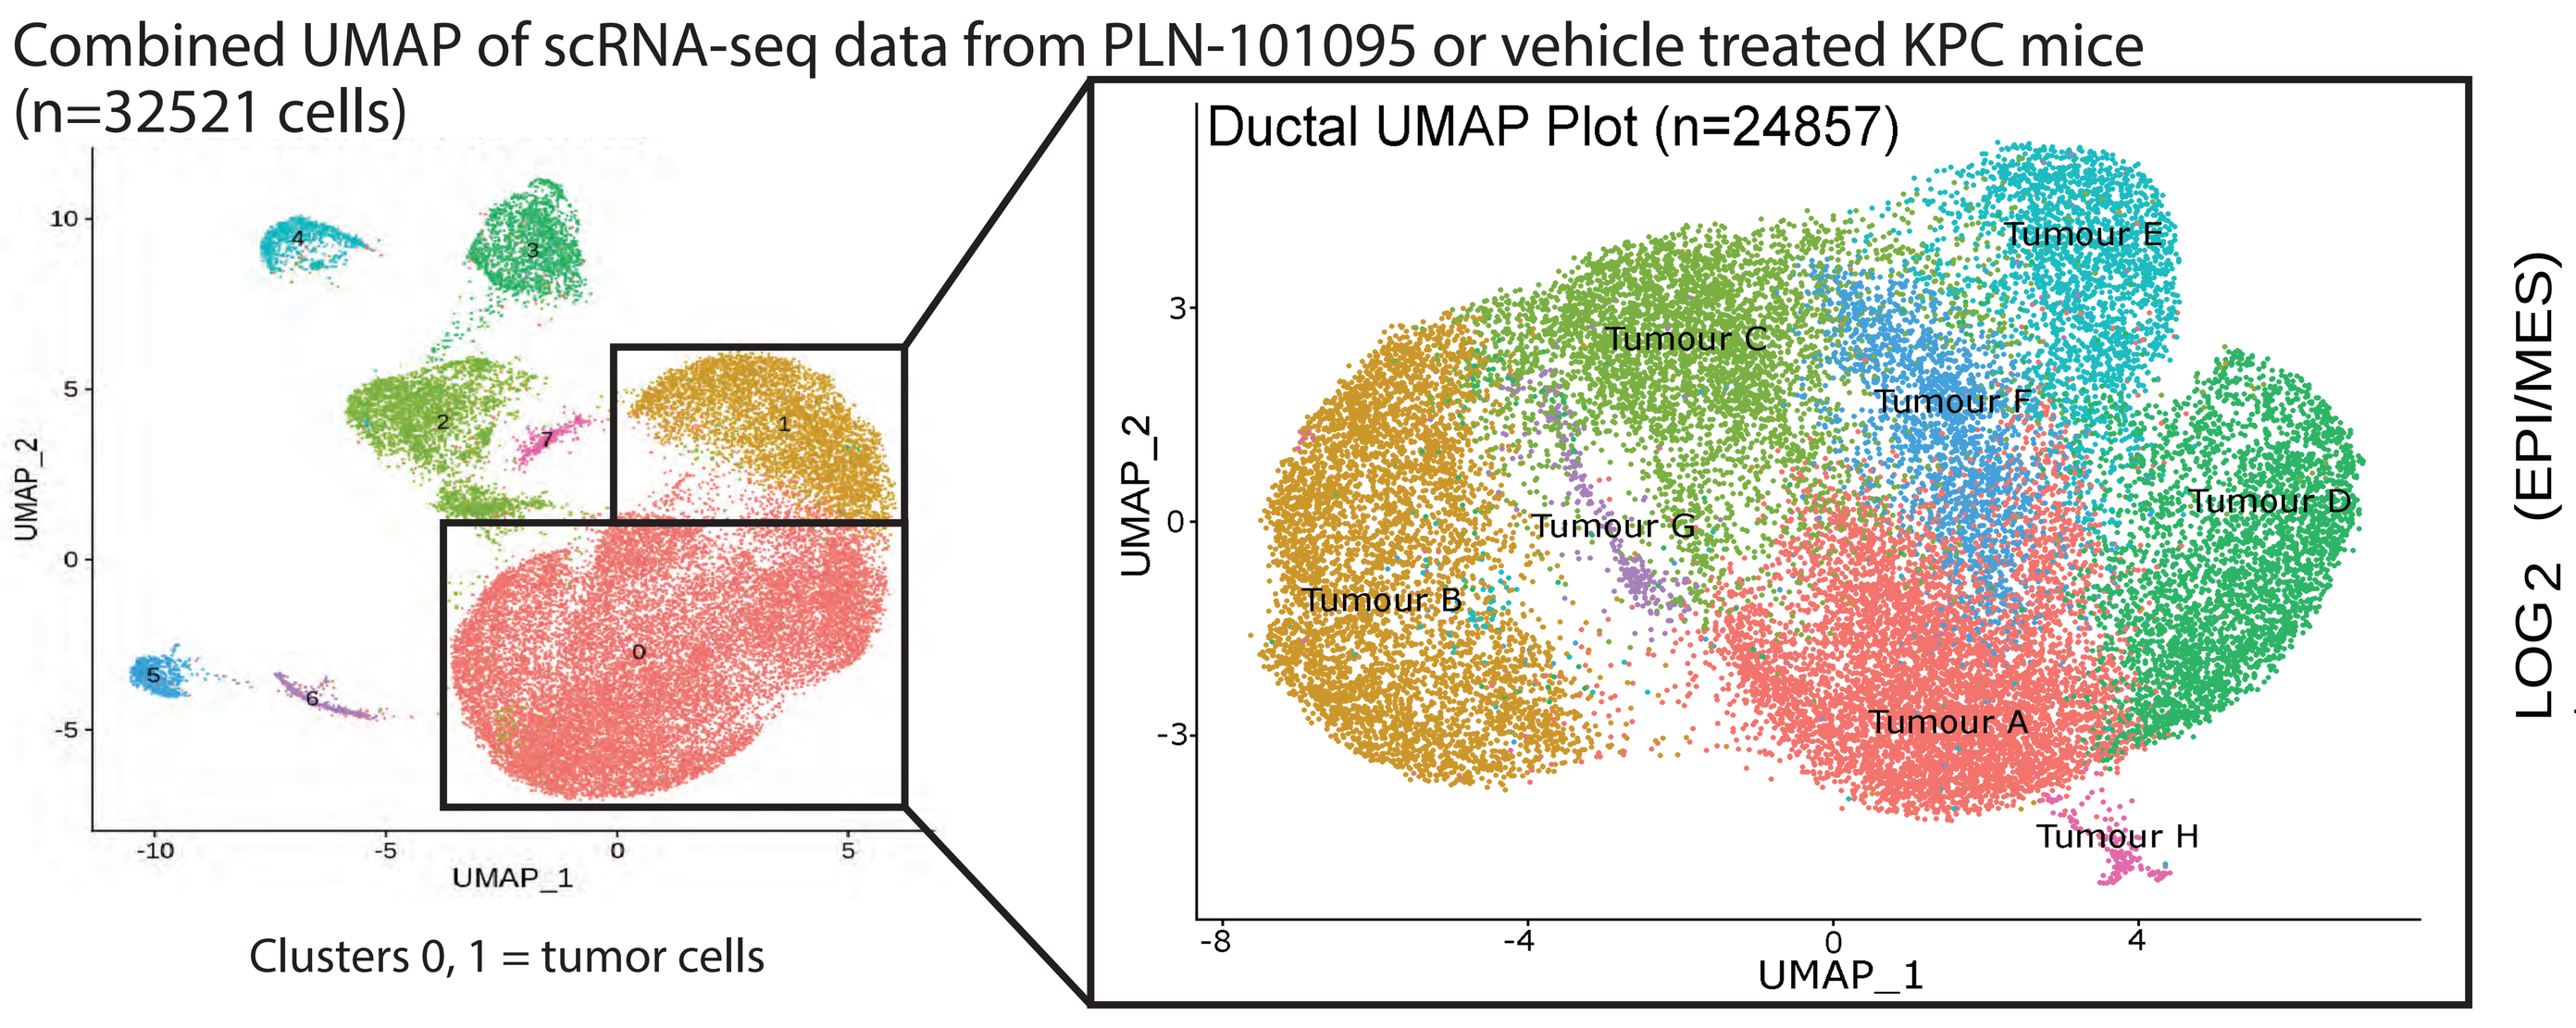
UMAP_1
UMAP_2
Ductal UMAP Plot (n=24857)
Tumour A
Tumour B
Tumour C
Tumour D
Tumour E
Tumour F
Tumour G
Tumour H

Clusters 0, 1 = tumor cells

EMT Score across diverse tumor cell subsets

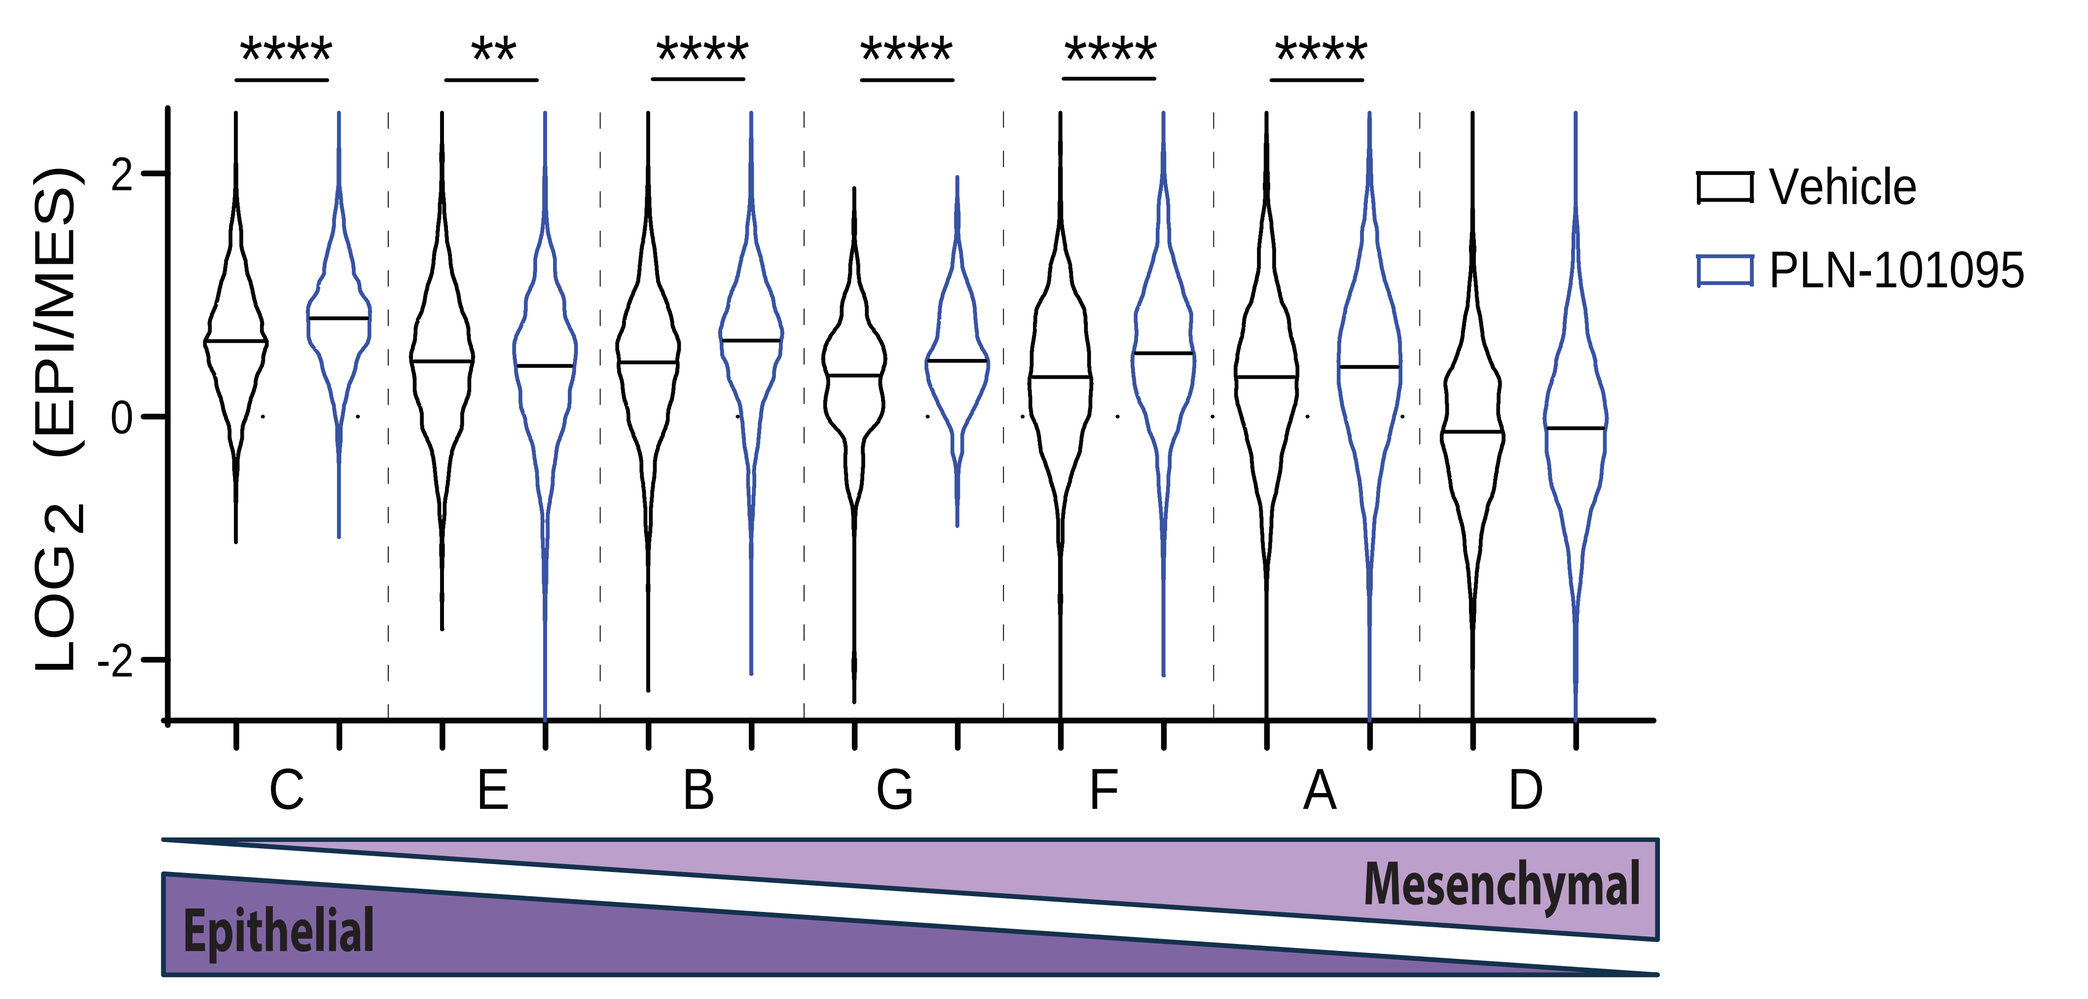
LOG 2 (EPI/MES)
Vehicle
PLN-101095
Epithelial
Mesenchymal

- PLN-101095 therapy induced MHCI class antigens and their transactivators (Nlrc5, H2-k1, H2-q6), genes associated with low tumor grade (S100a14), in parallel leading to decreased expression of pro-metastatic factors associated with poor prognosis (S100a4, Ankrd37, Slc16a3).

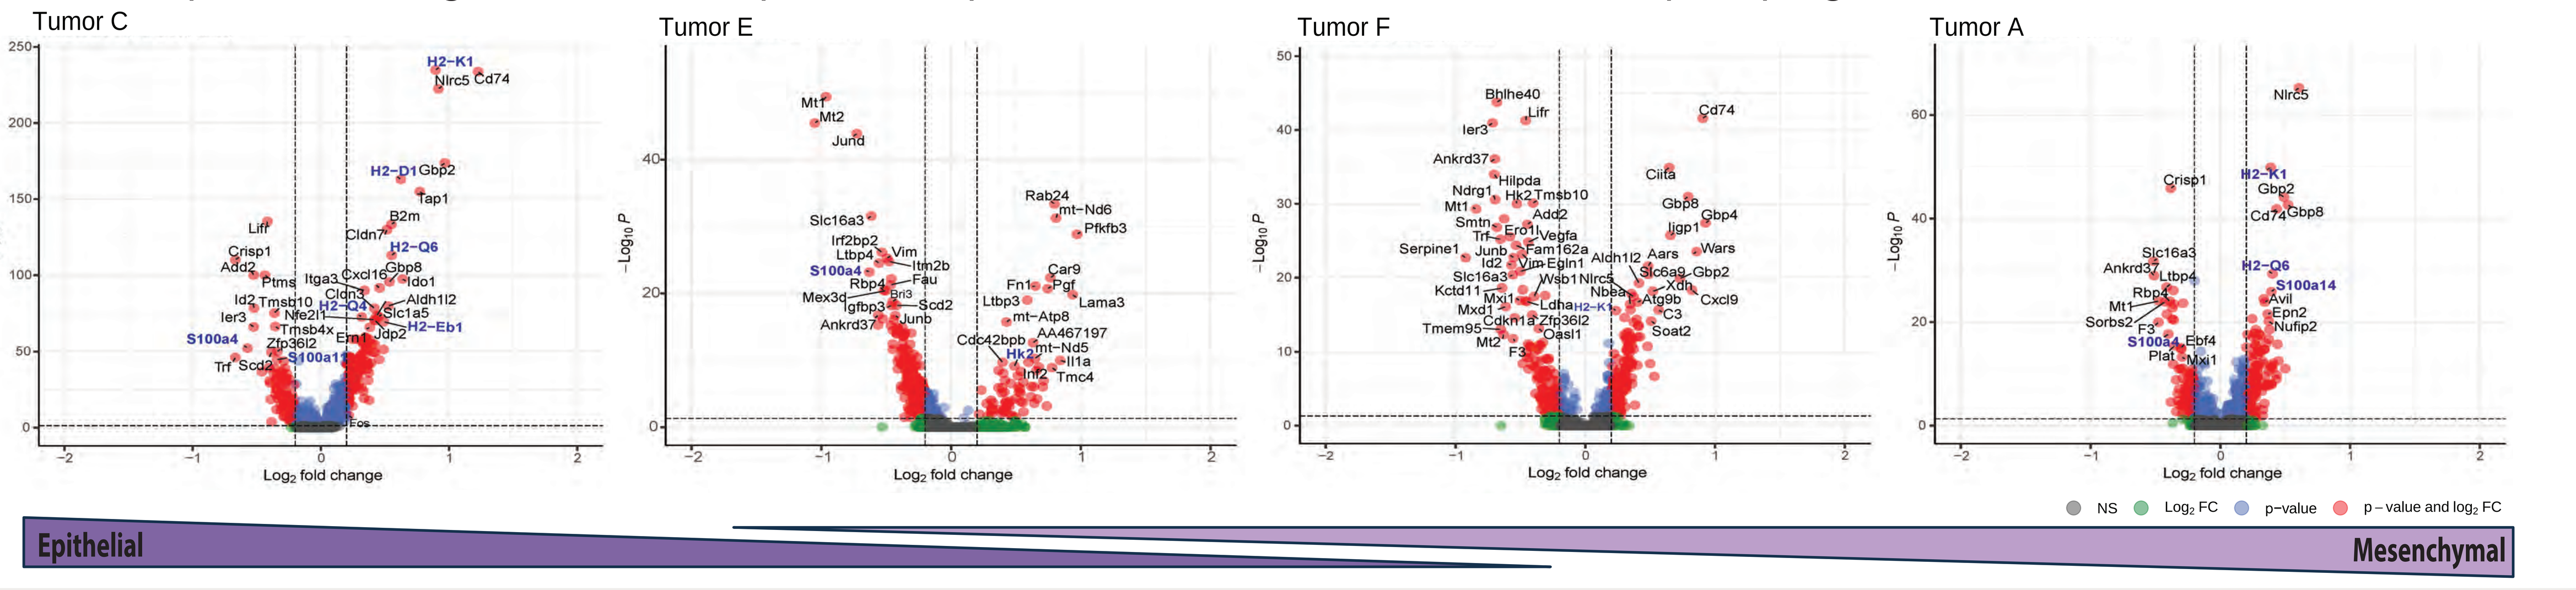
Tumor C
Tumor E
Tumor F
Tumor A
Log₂ fold change
NS
Log₂ FC
p-value
p-value and log₂ FC
Epithelial
Mesenchymal

Selective targeting of αVβ8 and αVβ1 (PLN-101095) or αVβ1 signaling (PLN-76104) potently inhibit primary tumor growth, spread and improve chemotherapy response in models of PDA

- Promising therapeutic efficacy of PLN-101095 and PLN-76104 in a αVß1⁺ PDX model of metastatic pancreatic cancer: robust inhibition of tumor growth and metastasis (liver and lungs), suggesting blocking αVß1 integrin alone may elicit a significant therapeutic response in PDA.

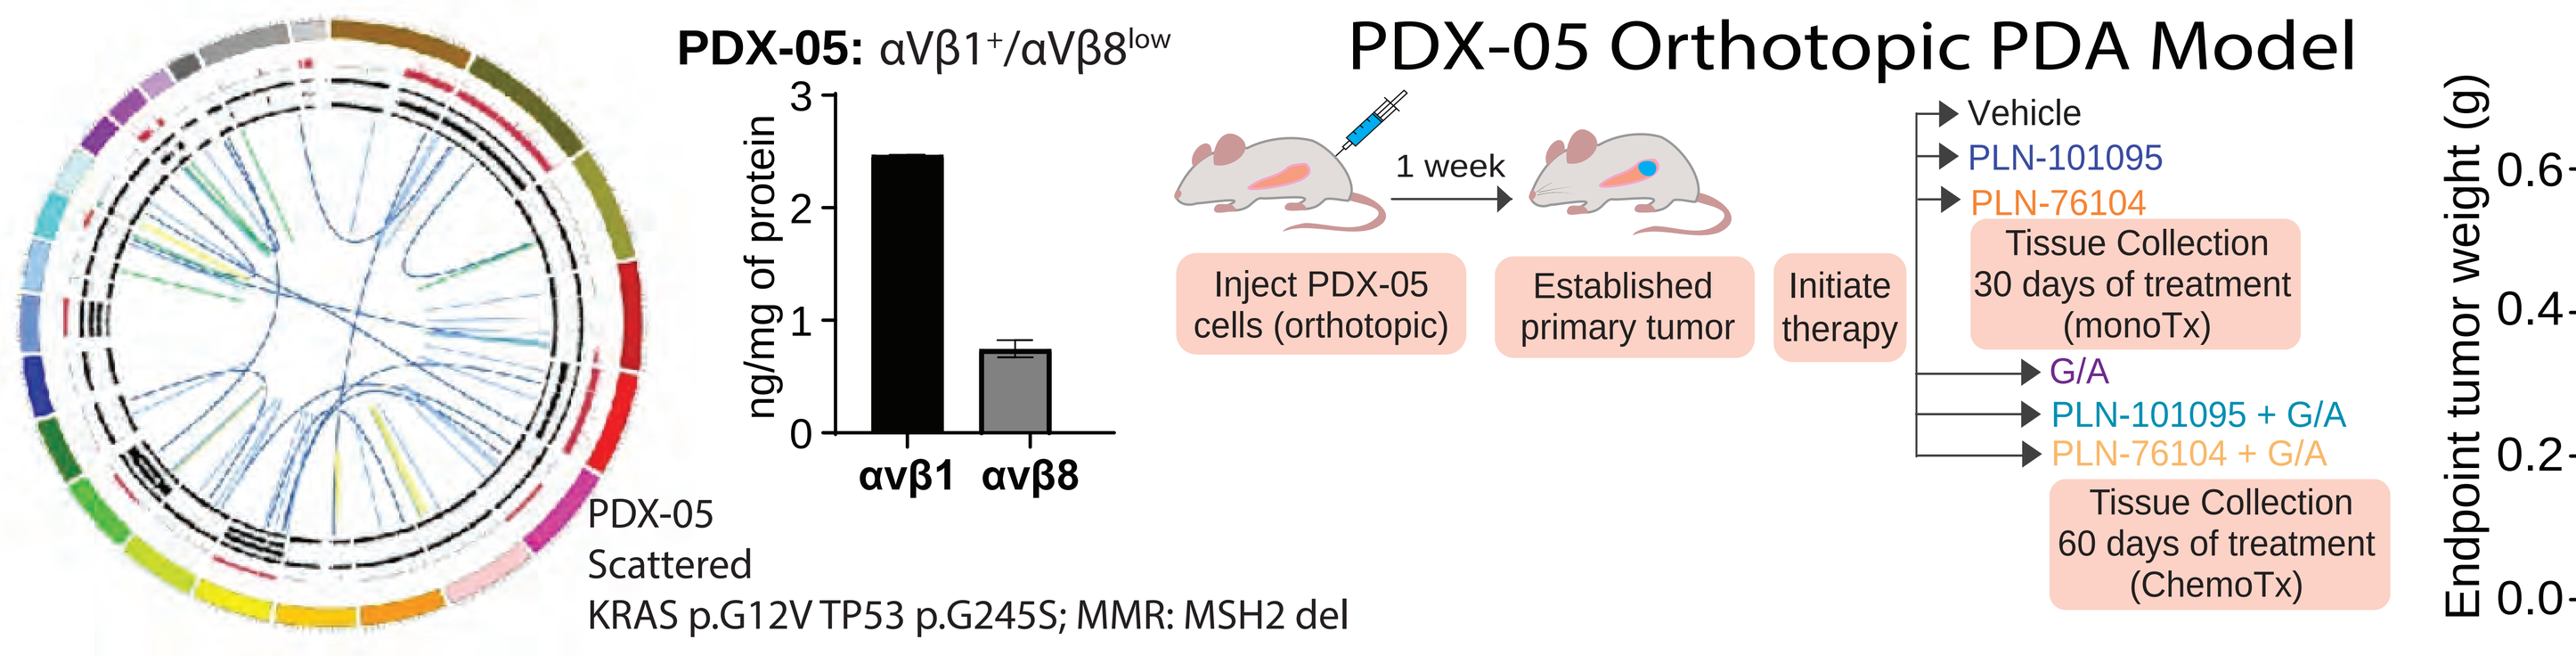
PDX-05: αVβ1⁺/αVβ8^low
PDX-05 Orthotopic PDA Model
ng/mg of protein
αvβ1
αvβ8
PDX-05
Scattered
KRAS p.G12V TP53 p.G245S; MMR: MSH2 del
1 week
Inject PDX-05 cells (orthotopic)
Established primary tumor
Initiate therapy
Vehicle
PLN-101095
PLN-76104
Tissue Collection 30 days of treatment (monoTx)
G/A
PLN-101095 + G/A
PLN-76104 + G/A
Tissue Collection 60 days of treatment (ChemoTx)

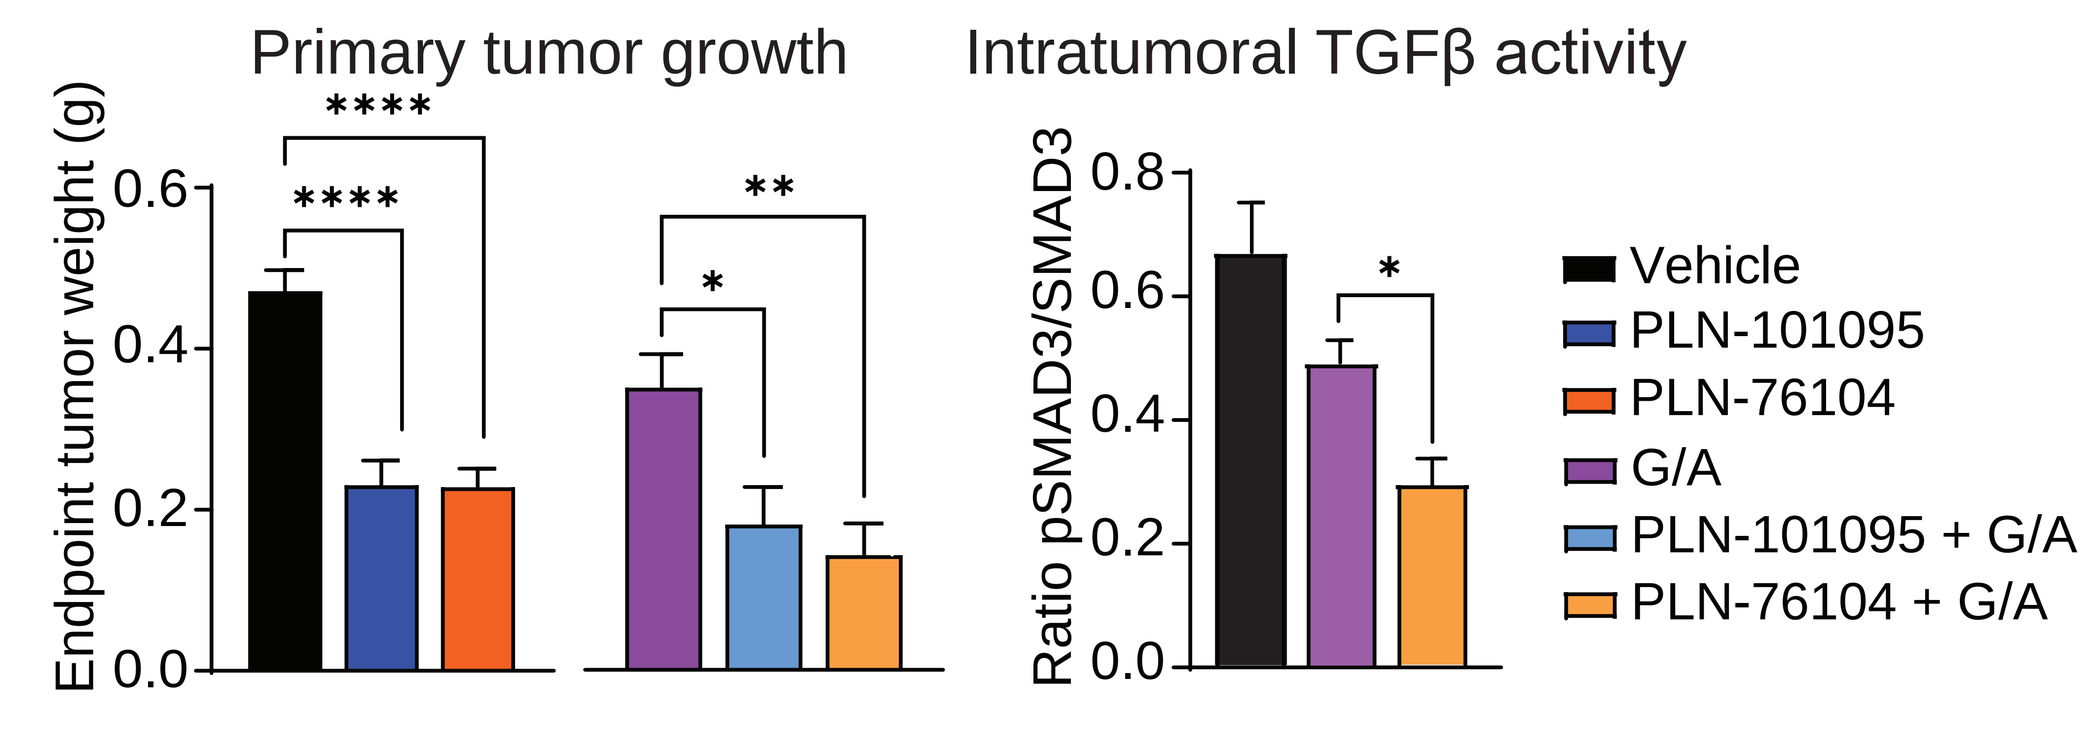
Primary tumor growth
Endpoint tumor weight (g)
Intratumoral TGFβ activity
Ratio pSMAD3/SMAD3
Vehicle
PLN-101095
PLN-76104
G/A
PLN-101095 + G/A
PLN-76104 + G/A

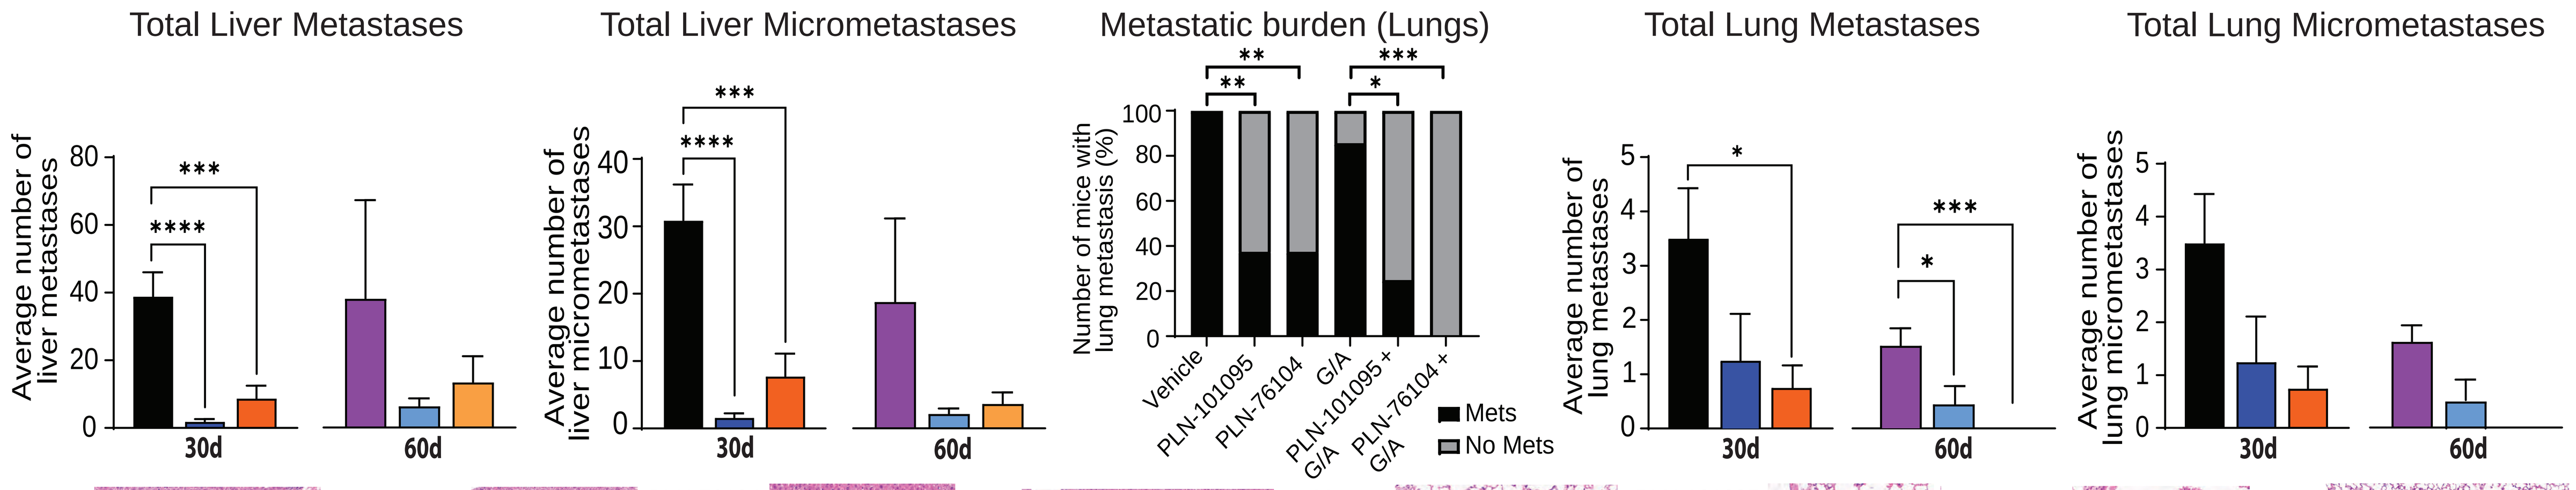
Total Liver Metastases
Average number of liver metastases
Total Liver Micrometastases
Average number of liver micrometastases
Metastatic burden (Lungs)
Number of mice with lung metastasis (%)
Mets
No Mets
Total Lung Metastases
Average number of lung metastases
Total Lung Micrometastases
Average number of lung micrometastases
30d
60d

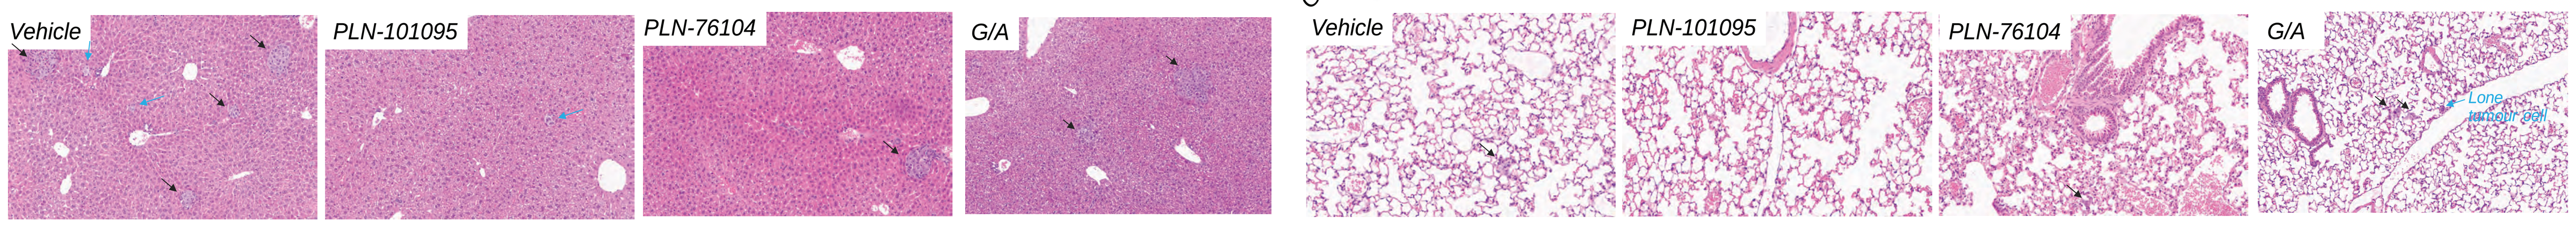
Vehicle
PLN-101095
PLN-76104
G/A

Conclusions

Dual targeting of αVβ8/αVβ1 signaling by PLN-101095 significantly enhanced response to immunotherapy or contemporary chemotherapies in models of advanced PDA. Promising signal of in vivo activity was also observed for PLN-76104, selective inhibitor of αVβ1 integrin. Mechanistic studies revealed PLN-101095-induced positive reprogramming of malignant PDA cells towards an epithelial expression state and gene expression changes associated with favorable immune response and improved prognosis. These data provide scientific rationale for the design of future PLN-101095 and SoC chemotherapy as well as immunotherapy combinations in pancreatic cancer, with Phase I first-in-human oncology studies with PLN-101095 plus ICB aleady underway.

References

1. Waddell N et al Nature (2015) 518(7540):495; **2.** Chou A et al Gut (2018) 67(12):2142; **3.** Vennin C et al Science Translational Medicine (2017) pii: eaai8504; **4.** Murphy KJ et al Science Advances (2021) 7(40):eabh0363; **5.** Biffi G et al Cancer Discovery (2019) 9(2):282; **6.** Krishnamurty AT et al Nature (2022) 611(7934):148; **7.** Takasaka N et al JCI Insight (2018) 3(20): e122591; **8.** Dodagatta-Marri E et al Cell Reports (2021) 36(1):109309; **9.** Qiang L et al Gastroenterology (2023) 165(4): 874-890

Acknowledgements

This work is co-funded by the Snow Medical and NHMRC fellowships (M.P.) and Pliant Therapeutics (U.S.).

Contact Details

Marina Pajic E: m.pajic@garvan.org.au