POST-HOC ANALYSIS OF BIOMARKERS OF INTERSTITIAL LUNG DISEASE PROGRESSION IN PARTICIPANTS WITH IDIOPATHIC PULMONARY FIBROSIS RECEIVING BEXOTEGRAST OVER 12-WEEKS IN INTEGRIS-IPF

Martin Decaris, Richard Ahn, Chris N. Barnes, Éric Lefebvre, Gregory Cosgrove, Scott Turner

Pliant Therapeutics, Inc., South San Francisco, CA, USA

RATIONALE & METHODS

- Biomarkers of early therapeutic response are needed to inform clinical decision making and improve efficiency of clinical trials in interstitial lung disease (ILD)
- Plasma levels of 17 proteins were recently shown to predict progression of ILD at one year as defined by death, lung transplant or ≥ 10% relative decline in forced vital capacity (FVC)[1] (Table 1)

Table 1: Plasma biomarkers predicting ILD progression at one year

Protein ID	Name	Protein ID	Name
AGER	advanced glycosylation end-product specific receptor	KRT19	keratin 19
ANGPTL4	angiopoietin like 4	MMP10	matrix metallopeptidase 10
CXCL17	C-X-C motif chemokine ligand 17	PLAUR	plasminogen activator, urokinase receptor
DPP10	dipeptidyl peptidase like 10	PRSS8	serine protease 8
FASLG	Fas ligand	SCGB3A2	secretoglobin family 3A member 2
FCAR	Fc alpha receptor	SPON1	spondin 1
HGF	hepatocyte growth factor	TGFA	transforming growth factor alpha
IL17C	interleukin 17C	TNFRSF11B	TNF receptor superfamily member 11b
ITGB6	integrin subunit beta 6		

- Here we assessed longitudinal changes in these biomarkers in participants with idiopathic pulmonary fibrosis (IPF) receiving bexotegrast (PLN-74809), to investigate their potential for detection of therapeutic response. Bexotegrast is an oral, once daily, dual-selective inhibitor of TGF-β-activating integrins $\alpha_V\beta_6$ and $\alpha_V\beta_1$ shown to reduce FVC decline in participants with IPF over 12 weeks[2].
- Plasma samples from INTEGRIS-IPF (NCT04396756) collected at baseline, Week 4 and Week 12 were analyzed using the Olink Explore 384 Inflammation panel. Mixed effects model for repeated measures (MMRM) and pairwise comparisons were used to identify differences in protein expression across time between bexotegrast-treated and placebo groups.

MECHANISM OF ACTION OF BEXOTEGRAST IN IPF

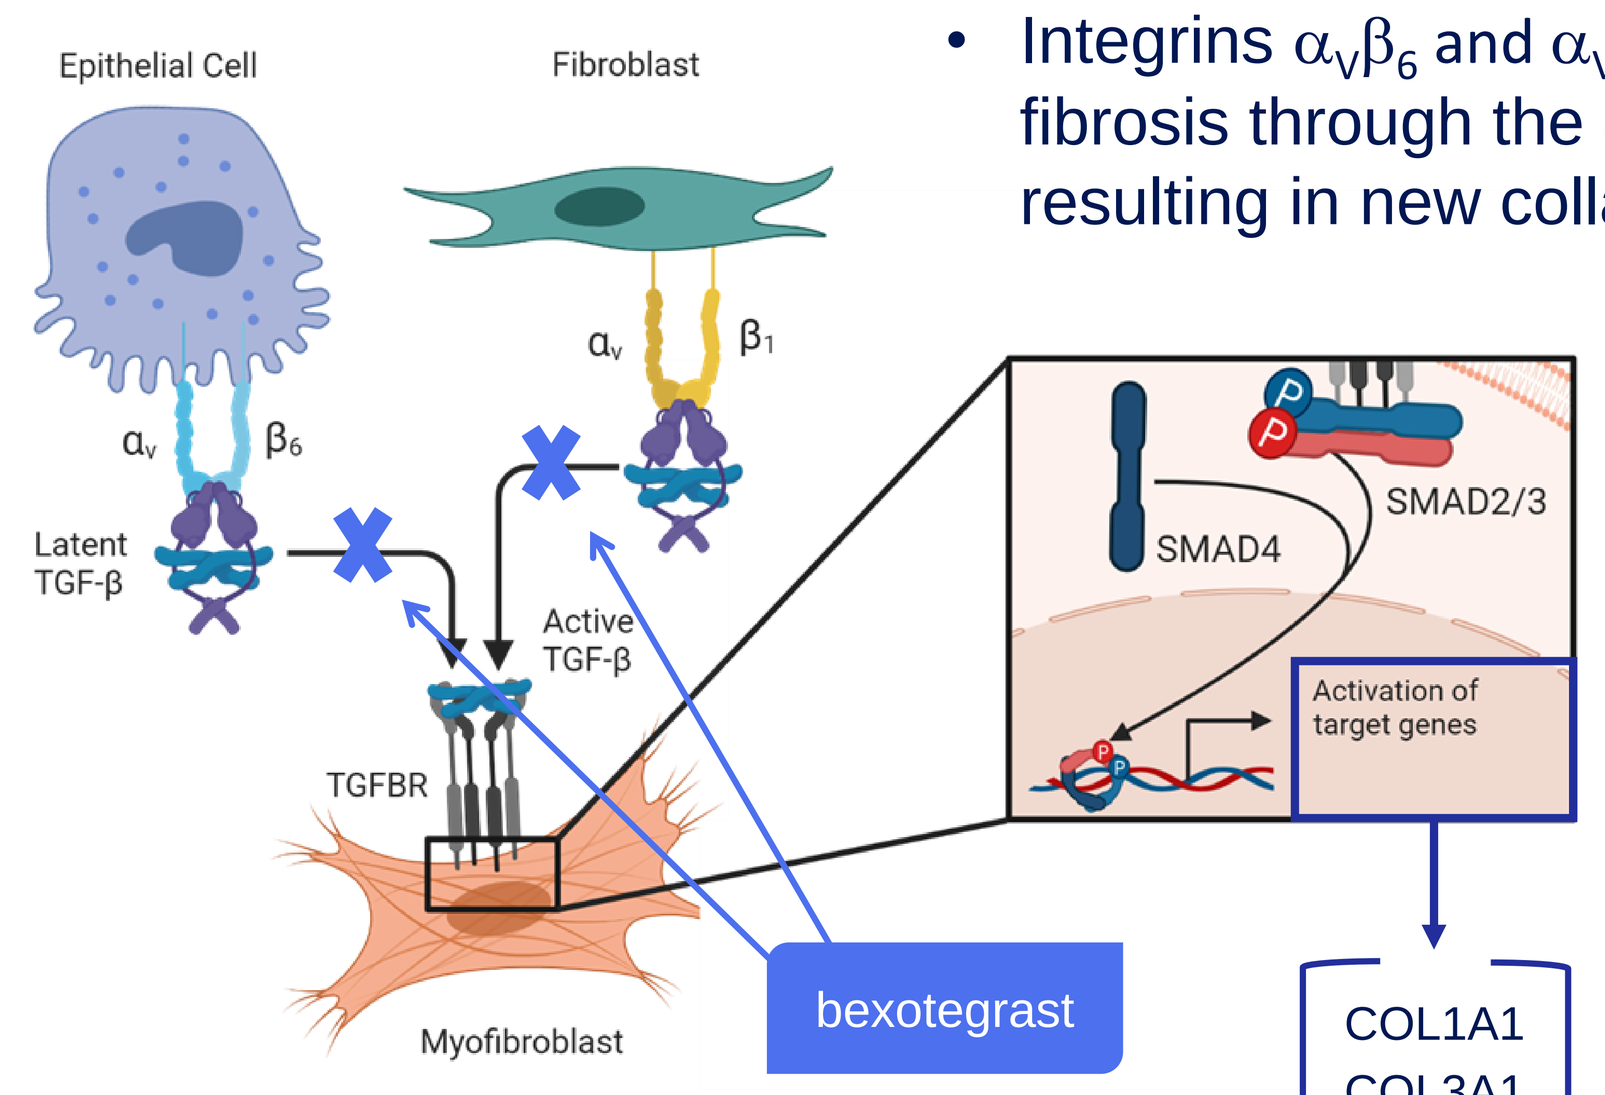

Epithelial Cell
Fibroblast
α_v
β_6
β_1
Latent TGF-β
Active TGF-β
TGFBR
Myofibroblast
bexotegrast
SMAD4
SMAD2/3
Activation of target genes

- Integrins $\alpha_V\beta_6$ and $\alpha_V\beta_1$ promote pulmonary fibrosis through the activation of latent TGF-β, resulting in new collagen synthesis.
- TGF-β signaling also drives ITGB6 gene expression, creating a positive feedback loop.

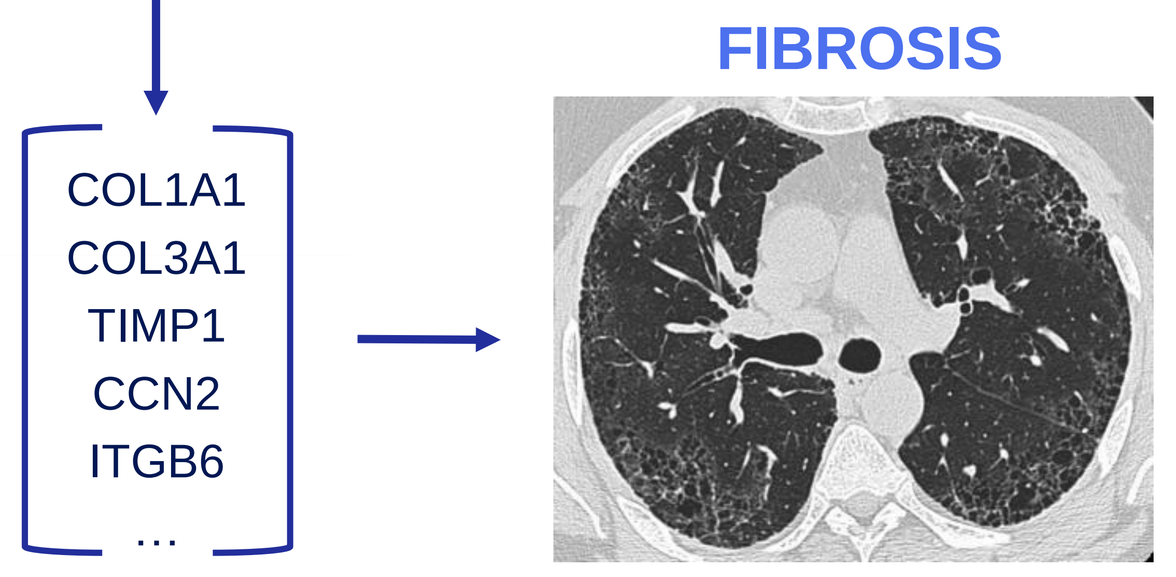

COL1A1
COL3A1
TIMP1
CCN2
ITGB6
…
FIBROSIS

- Bexotegrast, a dual inhibitor of integrins $\alpha_V\beta_6$ and $\alpha_V\beta_1$, is the subject of late-stage evaluation in the BEACON-IPF study.

INTEGRIS-IPF STUDY

- INTEGRIS-IPF, a Phase 2a clinical trial evaluating bexotegrast in participants with IPF (Fig 1A), previously showed a significant reduction in FVC decline (Fig 1B) in participants treated with bexotegrast vs placebo (320 mg QD vs placebo data shown here)[2,3].

A

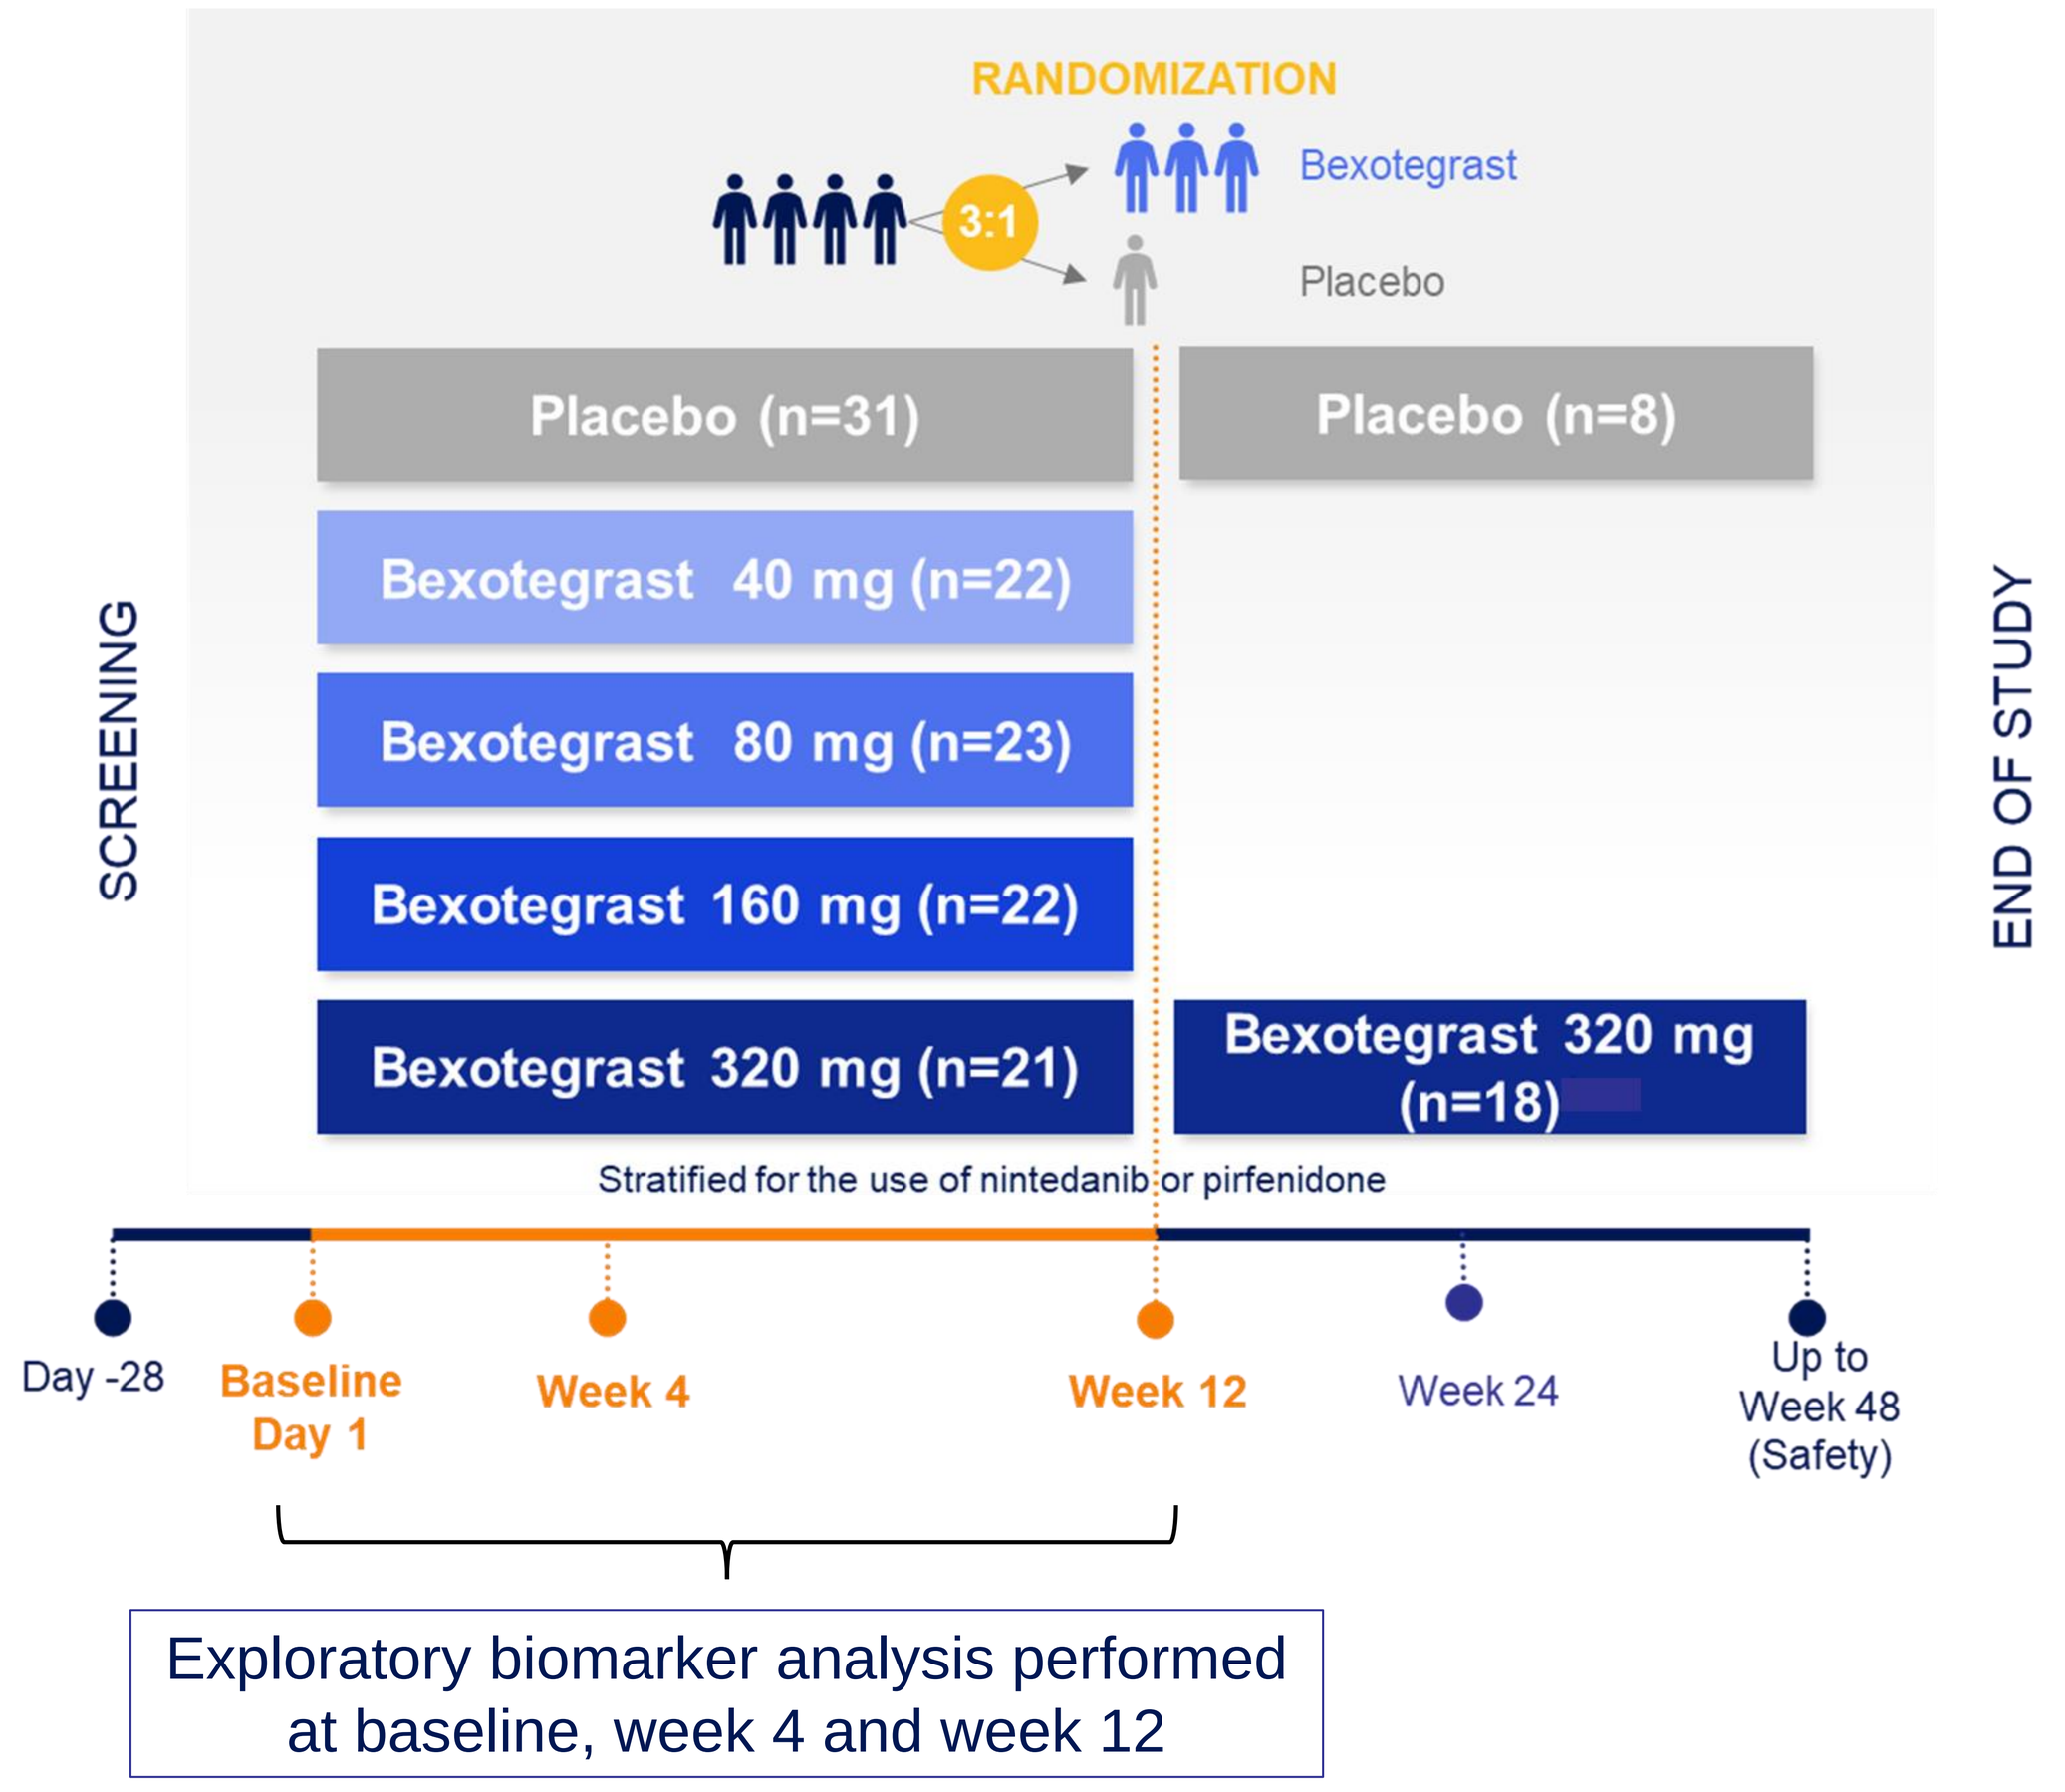

RANDOMIZATION
3:1
Bexotegrast
Placebo
Placebo (n=31)
Placebo (n=8)
Bexotegrast 40 mg (n=22)
Bexotegrast 80 mg (n=23)
Bexotegrast 160 mg (n=22)
Bexotegrast 320 mg (n=21)
Bexotegrast 320 mg (n=18)
SCREENING
END OF STUDY
Stratified for the use of nintedanib or pirfenidone
Day -28
Baseline Day 1
Week 4
Week 12
Week 24
Up to Week 48 (Safety)
Exploratory biomarker analysis performed at baseline, week 4 and week 12

B

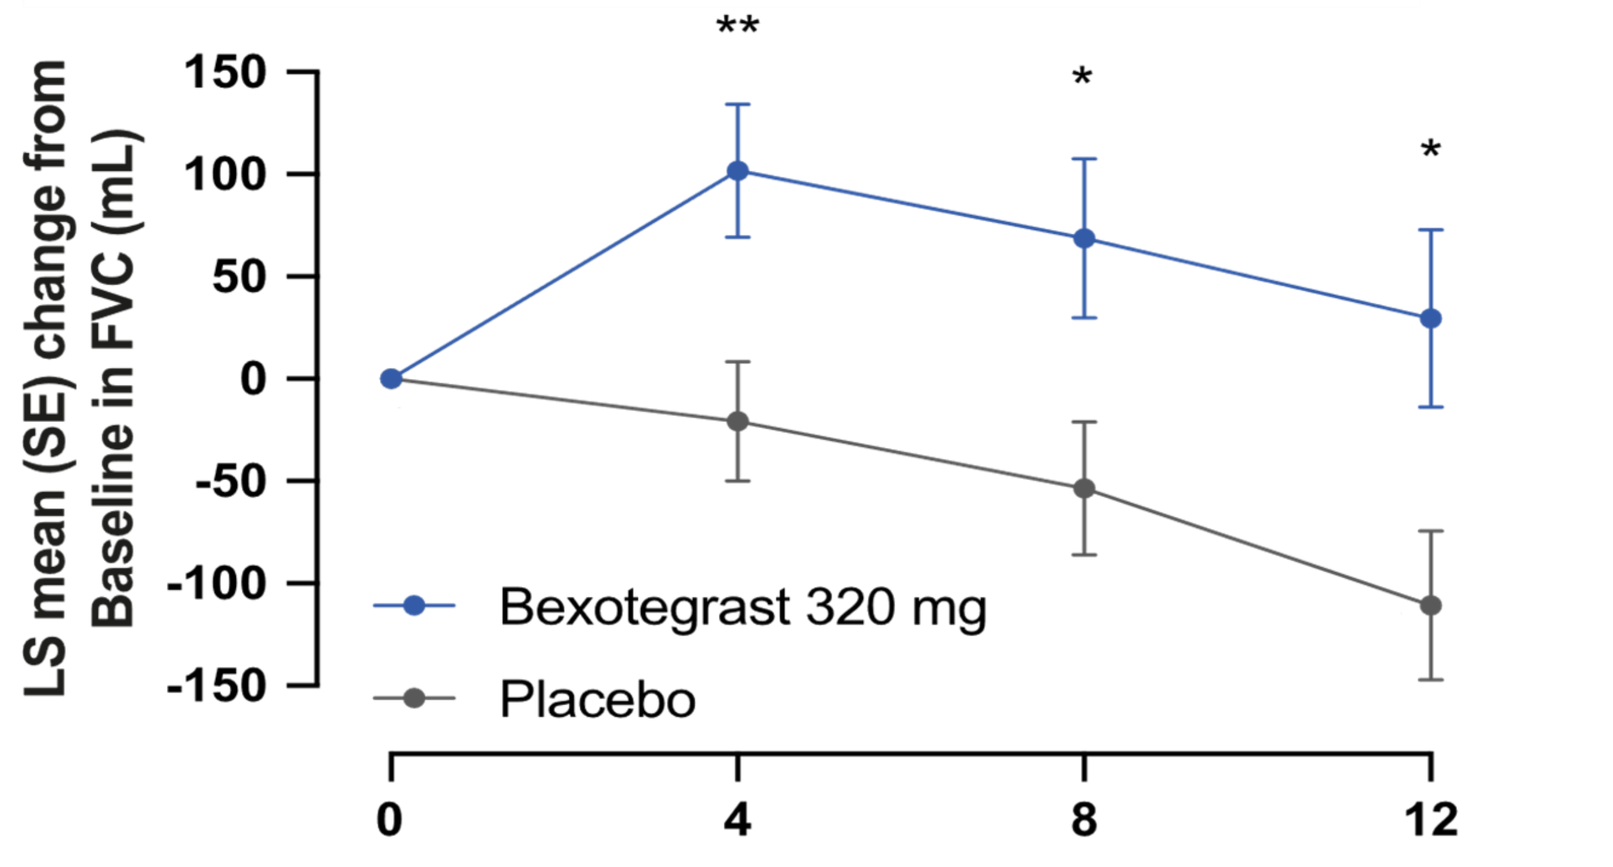

LS mean (SE) change from Baseline in FVC (mL)
150
100
50
0
-50
-100
-150
**
*
*
Bexotegrast 320 mg
Placebo
0
4
8
12

Figure 1: A) Study design for INTEGRIS-IPF. **B)** Change from Baseline in FVC through Week 12 in participants receiving 320 mg bexotegrast compared with placebo. FVC change from Baseline analyzed in efficacy mITT population by MMRM. *p<0.05 vs. placebo; **p<0.01 vs. placebo; FVC, forced vital capacity; LS, least squares; mITT, modified intent-to-treat; SE, standard error

- Prospective exploratory analysis of ILD progression plasma biomarker ITGB6 in INTEGRIS-IPF also showed a significant reduction in participants receiving bexotegrast vs placebo (Fig 2).

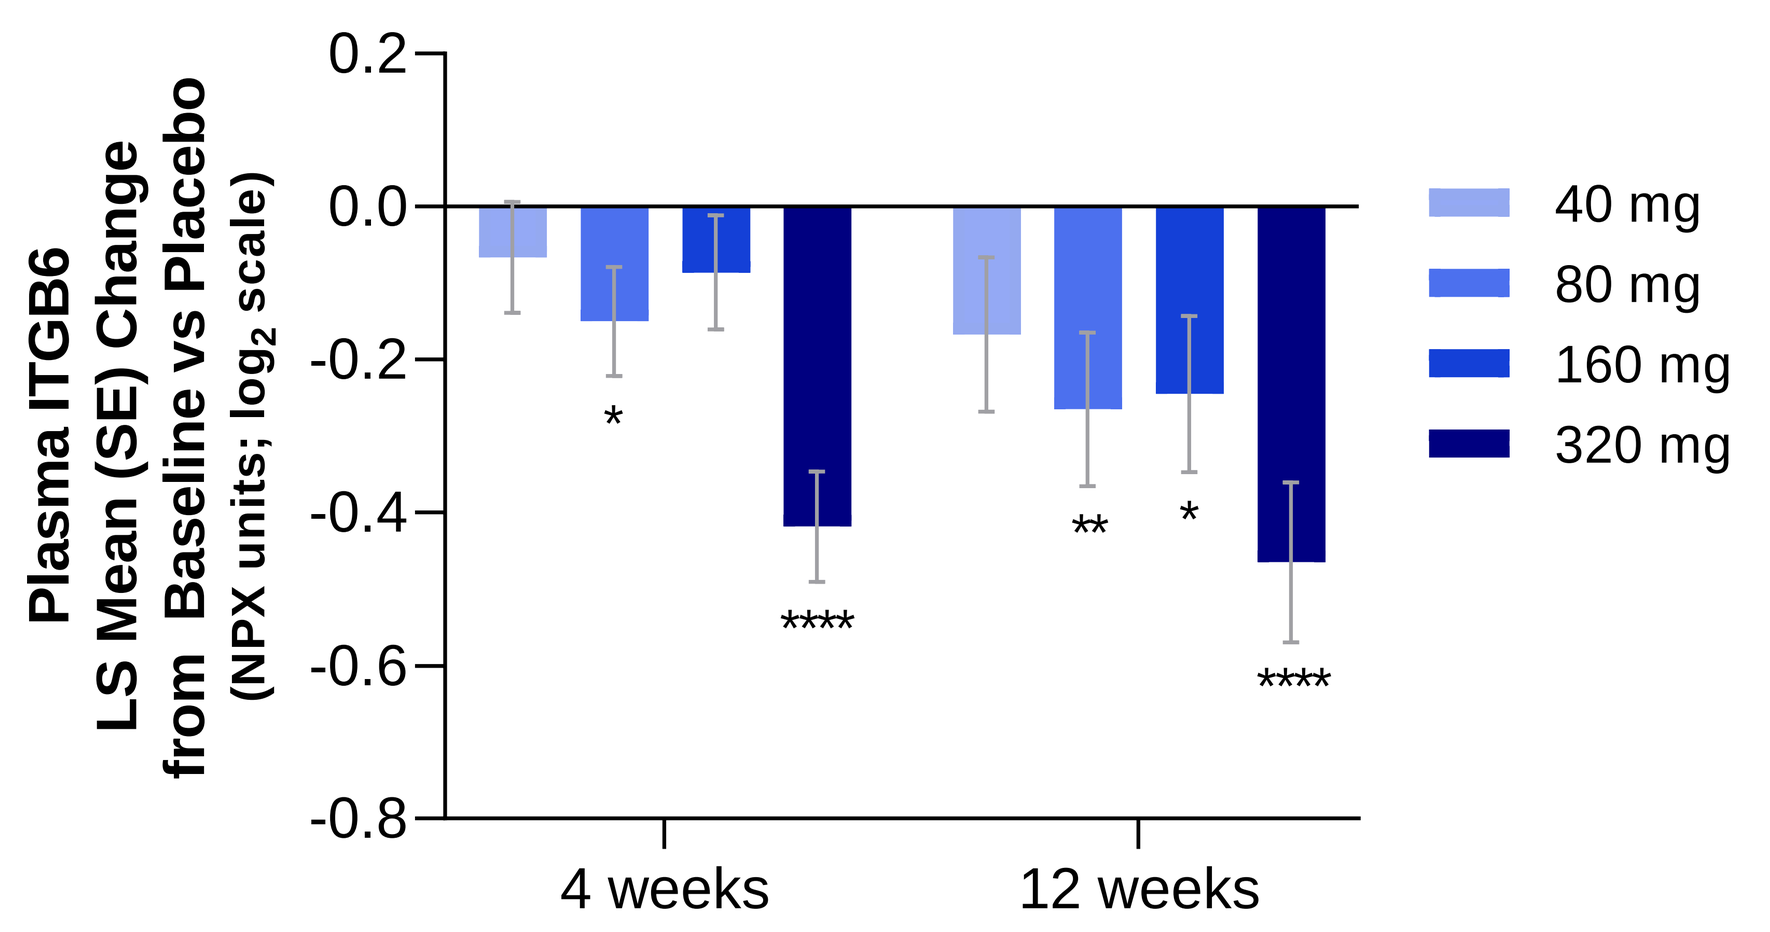

Plasma ITGB6 LS Mean (SE) Change from Baseline vs Placebo (NPX units; log2 scale)
0.2
0.0
-0.2
-0.4
-0.6
-0.8
*

**
*

4 weeks
12 weeks
40 mg
80 mg
160 mg
320 mg

Figure 2: Change from Baseline to Week 12 for exploratory endpoint plasma ITGB6 analyzed in the pharmacodynamic analysis population by MMRM. *p<0.05 vs. placebo; **p<0.01 vs. placebo; ****p<0.0001 vs. placebo; ITGB6, integrin beta-6; LS, least squares; mITT, modified intent-to-treat; SE, standard error

- Post-hoc analyses of the additional plasma proteins measured by the Olink Explore 384 inflammation panel used to prospectively assess ITGB6 levels, including 16 additional biomarkers of ILD progression, are presented here.

RESULTS

- A reduction in change from baseline for plasma levels of AGER, HGF, ITGB6, MMP10, PLAUR, SCGB3A2, and SPON1, seven biomarkers previously shown to predict outcome in ILD[1], was observed in participants receiving bexotegrast (160 and 320 mg QD) vs placebo (Figure 3). Statistical significance (320 mg QD vs placebo; $p < 0.05$) for each of these proteins was confirmed by MMRM analysis (data not presented).

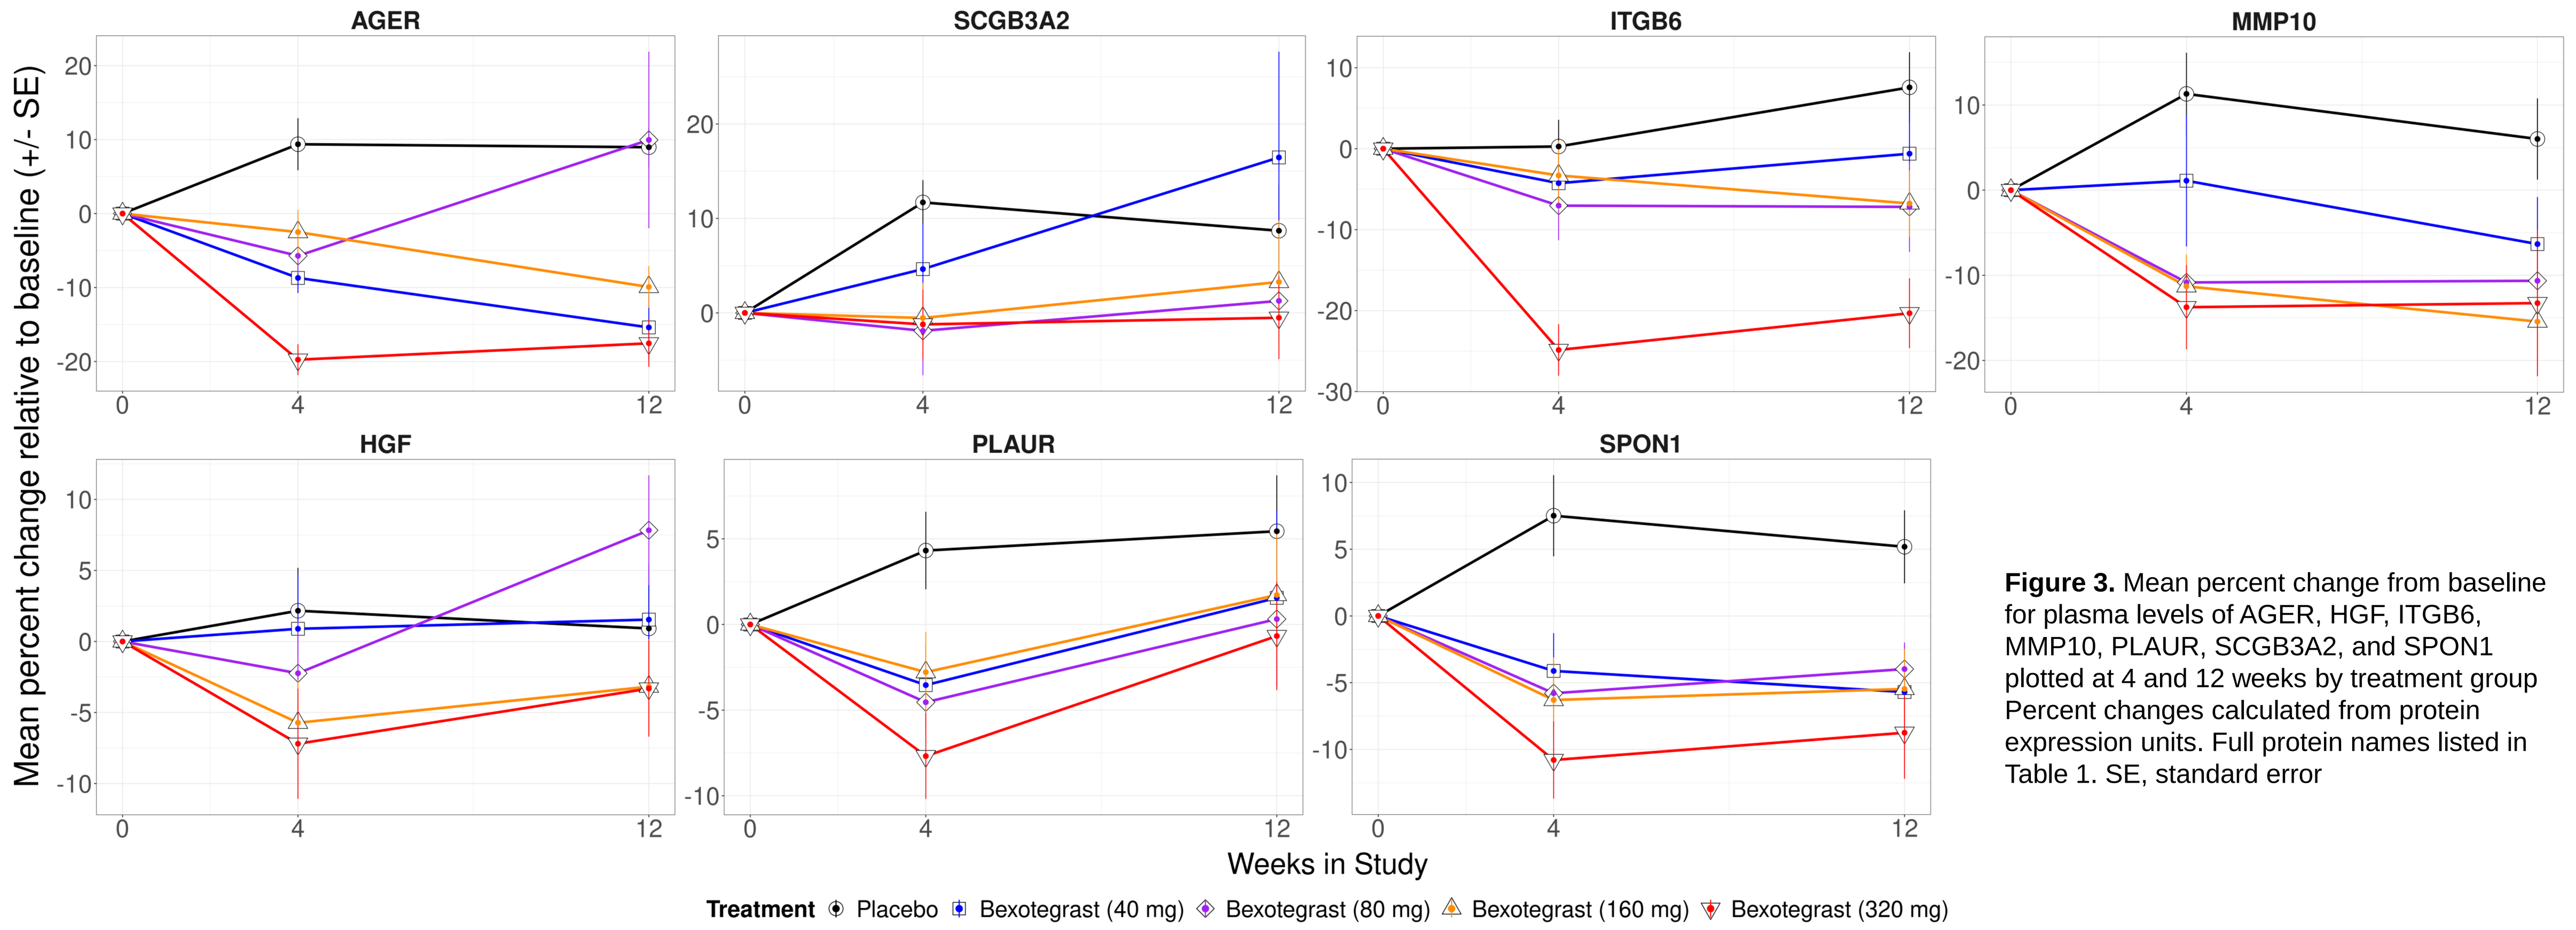

AGER
SCGB3A2
ITGB6
MMP10
HGF
PLAUR
SPON1
Mean percent change relative to baseline (+/- SE)
Weeks in Study
0
4
12
Treatment
Placebo
Bexotegrast (40 mg)
Bexotegrast (80 mg)
Bexotegrast (160 mg)
Bexotegrast (320 mg)

Figure 3. Mean percent change from baseline for plasma levels of AGER, HGF, ITGB6, MMP10, PLAUR, SCGB3A2, and SPON1 plotted at 4 and 12 weeks by treatment group Percent changes calculated from protein expression units. Full protein names listed in Table 1. SE, standard error

- Cross-sectional pairwise comparison of all proteins was performed between cohorts receiving bexotegrast (320 mg QD) and placebo at baseline, 4 weeks and 12 weeks
- A downward shift in the majority of plasma protein levels was observed at 4 and 12 weeks relative to baseline in participants receiving bexotegrast

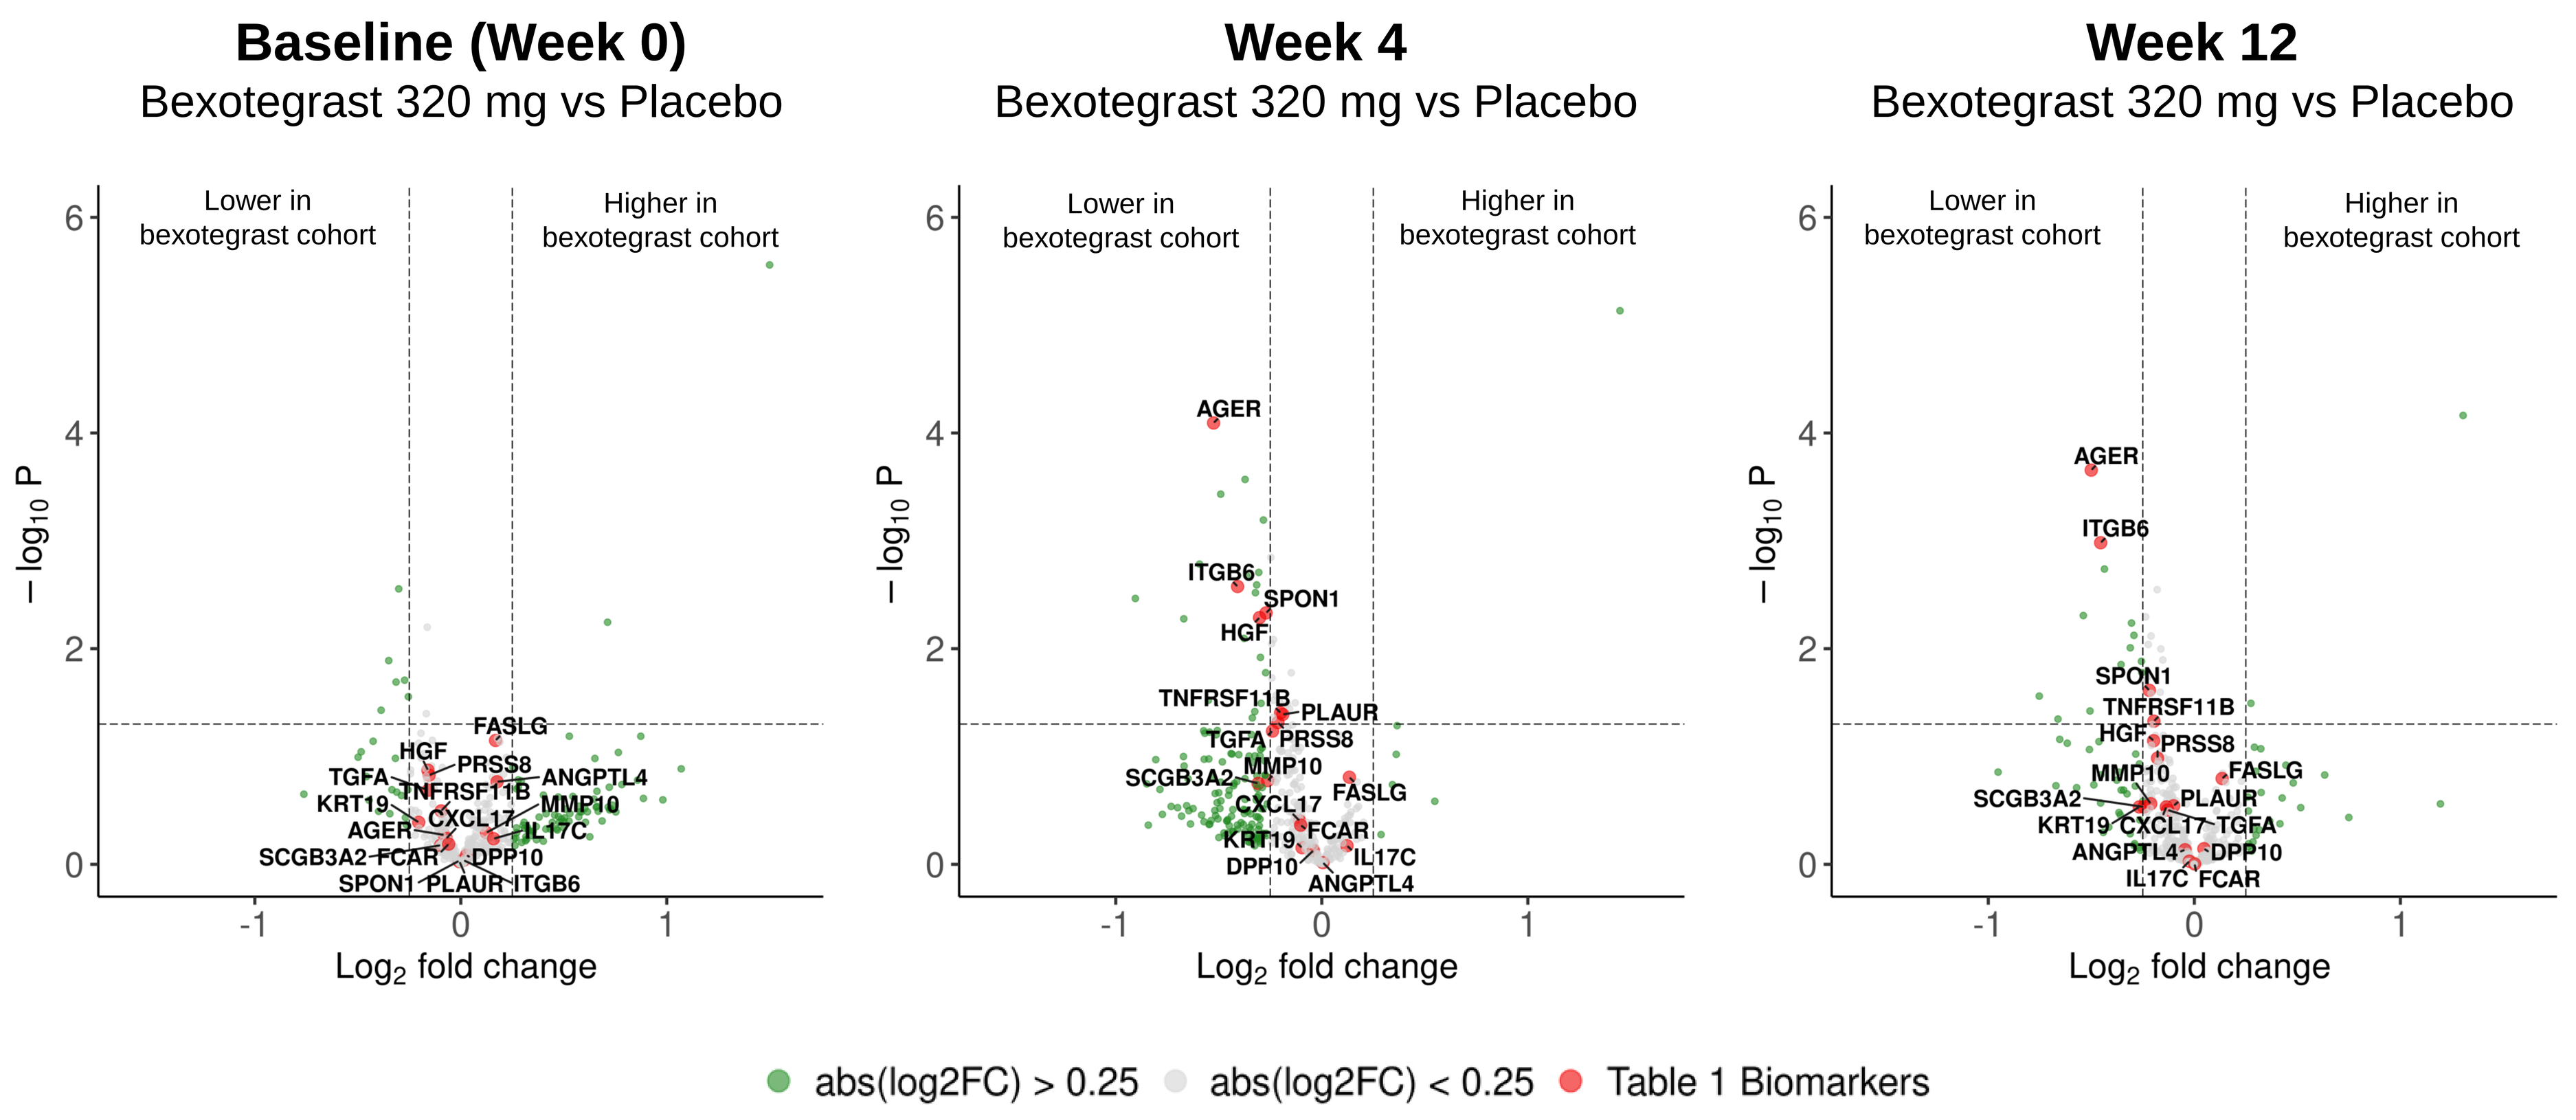

Baseline (Week 0)
Bexotegrast 320 mg vs Placebo
Week 4
Bexotegrast 320 mg vs Placebo
Week 12
Bexotegrast 320 mg vs Placebo
Lower in bexotegrast cohort
Higher in bexotegrast cohort
– log10 P
Log2 fold change
AGER
ITGB6
SPON1
HGF
TNFRSF11B
PLAUR
TGFA
PRSS8
SCGB3A2
MMP10
FASLG
CXCL17
KRT19
FCAR
DPP10
IL17C
ANGPTL4
abs(log2FC) > 0.25
abs(log2FC) < 0.25
Table 1 Biomarkers

Figure 4. Cross-sectional pairwise comparison of plasma protein levels between participants receiving 320 mg QD bexotegrast or placebo was performed at (A) baseline, (B) 4 weeks and (C) 12 weeks. 365 proteins passing QC were included in the analysis.17 protein biomarkers previously shown to predict 1 year outcome in ILD (see Table 1) highlighted in red (details regarding additional proteins to be reported separately). Proteins with log2 fold change > 0.25 indicated in green.

CONCLUSIONS

- Seven previously identified plasma biomarkers of ILD progression (AGER, HGF, ITGB6, MMP10, PLAUR, SCGB3A2, and SPON1) were also found to be significantly modulated in participants with IPF receiving bexotegrast over 12 weeks.
- Future analyses of plasma biomarkers are planned in the enrolling Phase 2b BEACON-IPF study (NCT06097260) to better understand their potential utility for early detection of therapeutic response.

References: 1. Bowman, W.S. et al. Lancet Respir Med 2022; 2. Lancaster, L. et al. ATS International Conference 2023; 3. Wuyts, W. et al. Eur. Respir. J. 2023 62: OA1423

Contact information: MDecaris@pliantrx.com

Miscellaneous: Copyright © 2024 Pliant Therapeutics, Inc. | All Rights Reserved

Disclosures: MD, RA, CB, EL, GC and ST are employees and/or shareholders of Pliant Therapeutics