EVALUATION OF QUANTITATIVE IMAGING IN A PHASE 2A STUDY FOR THE TREATMENT OF IDIOPATHIC PULMONARY FIBROSIS WITH BEXOTEGRAST (INTEGRIS-IPF)

Jonathan G. Goldin,[1] Grace J. Kim,[2] Chris N. Barnes,[3] Martin Decaris,[3] Scott Turner,[3] Hardean E. Achneck,[3] Éric Lefebvre,[3] Gregory P. Cosgrove,[3] Lisa H. Lancaster[4]

[1]David Geffen School of Medicine at UCLA, Santa Monica, CA, USA; [2]Radiological Science, UCLA, Los Angeles, CA, USA; [3]Pliant Therapeutics Inc., South San Francisco, CA, USA; [4]Division of Allergy, Pulmonary, and Critical Care Medicine, Vanderbilt University Medical Center, Nashville, TN, USA

Poster Number: PD05

BACKGROUND

- Idiopathic pulmonary fibrosis (IPF) is a rare progressive disease characterized by progressive lung scarring[1]
- $\alpha_v\beta_6$ and $\alpha_v\beta_1$ are integrins upregulated in patients with IPF that promote fibrosis through the activation of transforming growth factor β, a central mediator of fibrosis[2-4]
- High-resolution computed tomography (HRCT) can provide detailed characterization of the anatomy of the lung and has become an important technique in the diagnosis and monitoring of diseases such as IPF[5,6]
 - IPF is characterized by usual interstitial pneumonia (UIP) or probable UIP–like radiographic patterns, which are predominately subpleural, basal reticular abnormalities that include honeycombing with or without peripheral traction bronchiectasis or bronciolectasis[5]
 - HRCT imaging can be used to monitor IPF progression by quantifying fibrosis and ground glass
 - Fibrosis can be measured via the quantitative lung fibrosis (QLF) score, which measures the extent of reticular patterns
 - Ground glass opacities, hazy opacities that do not obscure bronchial structures, can be associated with alveolar inflammation and measured by quantitative ground glass (QGG)
 - Higher QLF and QGG scores indicate increased radiological lung fibrotic disease extent; there is a significant inverse association between radiological scores and pulmonary function measurements such as forced vital capacity[7,8]
 - New or worsening ground glass opacities may suggest clinically significant respiratory deteriorations such as an acute exacerbation of IPF
- Bexotegrast (PLN-74809) is an oral, once-daily, dual selective inhibitor of $\alpha_v\beta_6$ and $\alpha_v\beta_1$ integrins that is currently in development for the treatment of IPF

OBJECTIVE

- This analysis reports the effect of bexotegrast on quantitative imaging parameters compared with placebo in the INTEGRIS-IPF clinical program

METHODS

- INTEGRIS-IPF (NCT04396756) was a Phase 2a, global, randomized, double-blind, placebo-controlled trial (**Figure 1**)
- Participants with a diagnosis of IPF (based on ATS/ERS/JRS/ALAT 2018 guidelines[5]) for ≥5 years, percent predicted forced vital capacity of ≥45%, diffusing capacity for carbon monoxide (hemoglobin-adjusted) of ≥30% and life expectancy of ≥6 months were randomized 3:1 to receive once-daily bexotegrast or placebo in 3 cohorts (40 mg or placebo for 12 weeks; 80 mg, 160 mg or placebo for 12 weeks; 320 mg or placebo for up to 48 weeks); participants randomized to placebo were pooled
- Background therapy for IPF with pirfenidone or nintedanib was permitted, provided dosage was stable for ≥3 months prior to screening

Figure 1. INTEGRIS-IPF Trial Schematic

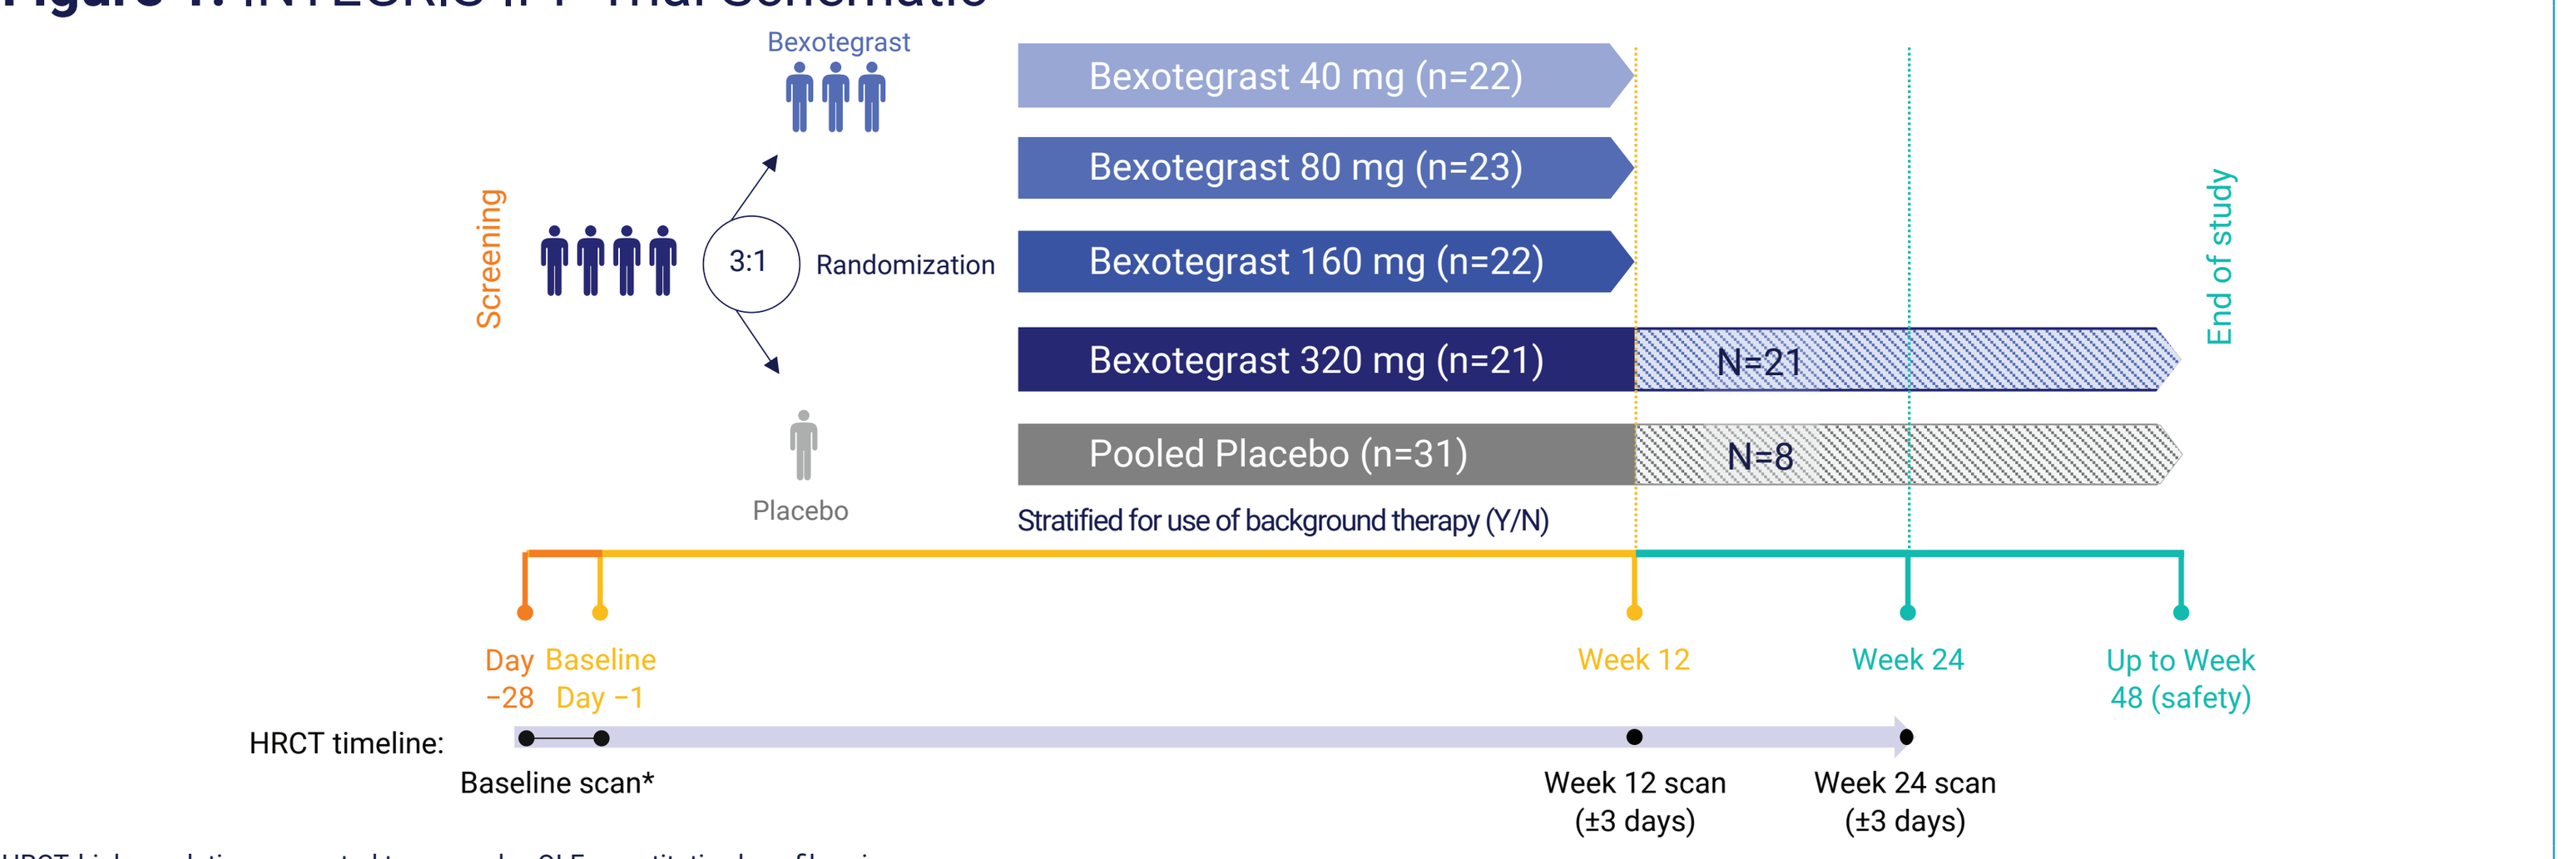
Bexotegrast
Screening
3:1
Randomization
Placebo
Bexotegrast 40 mg (n=22)
Bexotegrast 80 mg (n=23)
Bexotegrast 160 mg (n=22)
Bexotegrast 320 mg (n=21)
N=21
Pooled Placebo (n=31)
N=8
Stratified for use of background therapy (Y/N)
End of study
Day −28
Baseline Day −1
Week 12
Week 24
Up to Week 48 (safety)
HRCT timeline:
Baseline scan*
Week 12 scan (±3 days)
Week 24 scan (±3 days)

HRCT, high-resolution computed tomography; QLF, quantitative lung fibrosis.
*This figure represents the intent-to-treat population. An HRCT scan performed within 2 years of the screening date may be used for emphysema vs fibrotic changes. For baseline QLF, an HRCT scan performed within 1 month may be used if it meets the central reader specifications.

- Quantitative imaging was based on a supervised machine learning algorithm using QLF and QGG as radiomic features for each HRCT image
- Whole-lung QLF and QGG extent (in percentage) were assessed by volumetric HRCT scans at baseline, Week 12 and Week 24 on the per computed tomography (CT) protocol population, a subset of the intent-to-treat population with available HRCT imaging data that met quality standards per the imaging protocol. The per CT protocol population included participants who had:
 - Baseline scans conducted ≤1 month from randomization
 - Follow-up scans conducted within a 3-day window from the Week 12 or 24 visit date
 - No report of an exacerbation or progression of IPF adverse event <2 weeks prior to a scan
 - No issue with image capture or read as identified by third-party personnel
- Frequency counts and percentages were used for categorical variables; means, SDs and SEs were used for continuous variables

RESULTS

Summary of Participant Disposition

- Of the 119 participants in the INTEGRIS-IPF study, 100 were included in the per CT protocol population (**Figure 2**)
 - Eighty-eight participants (88.0%) in the per CT protocol population had scans at both baseline and Week 12
 - Participant data were most commonly excluded from the per CT protocol population due to issues with HRCT quality (volume artifacts and wrong CT parameters)

Figure 2. Participant Disposition Summary

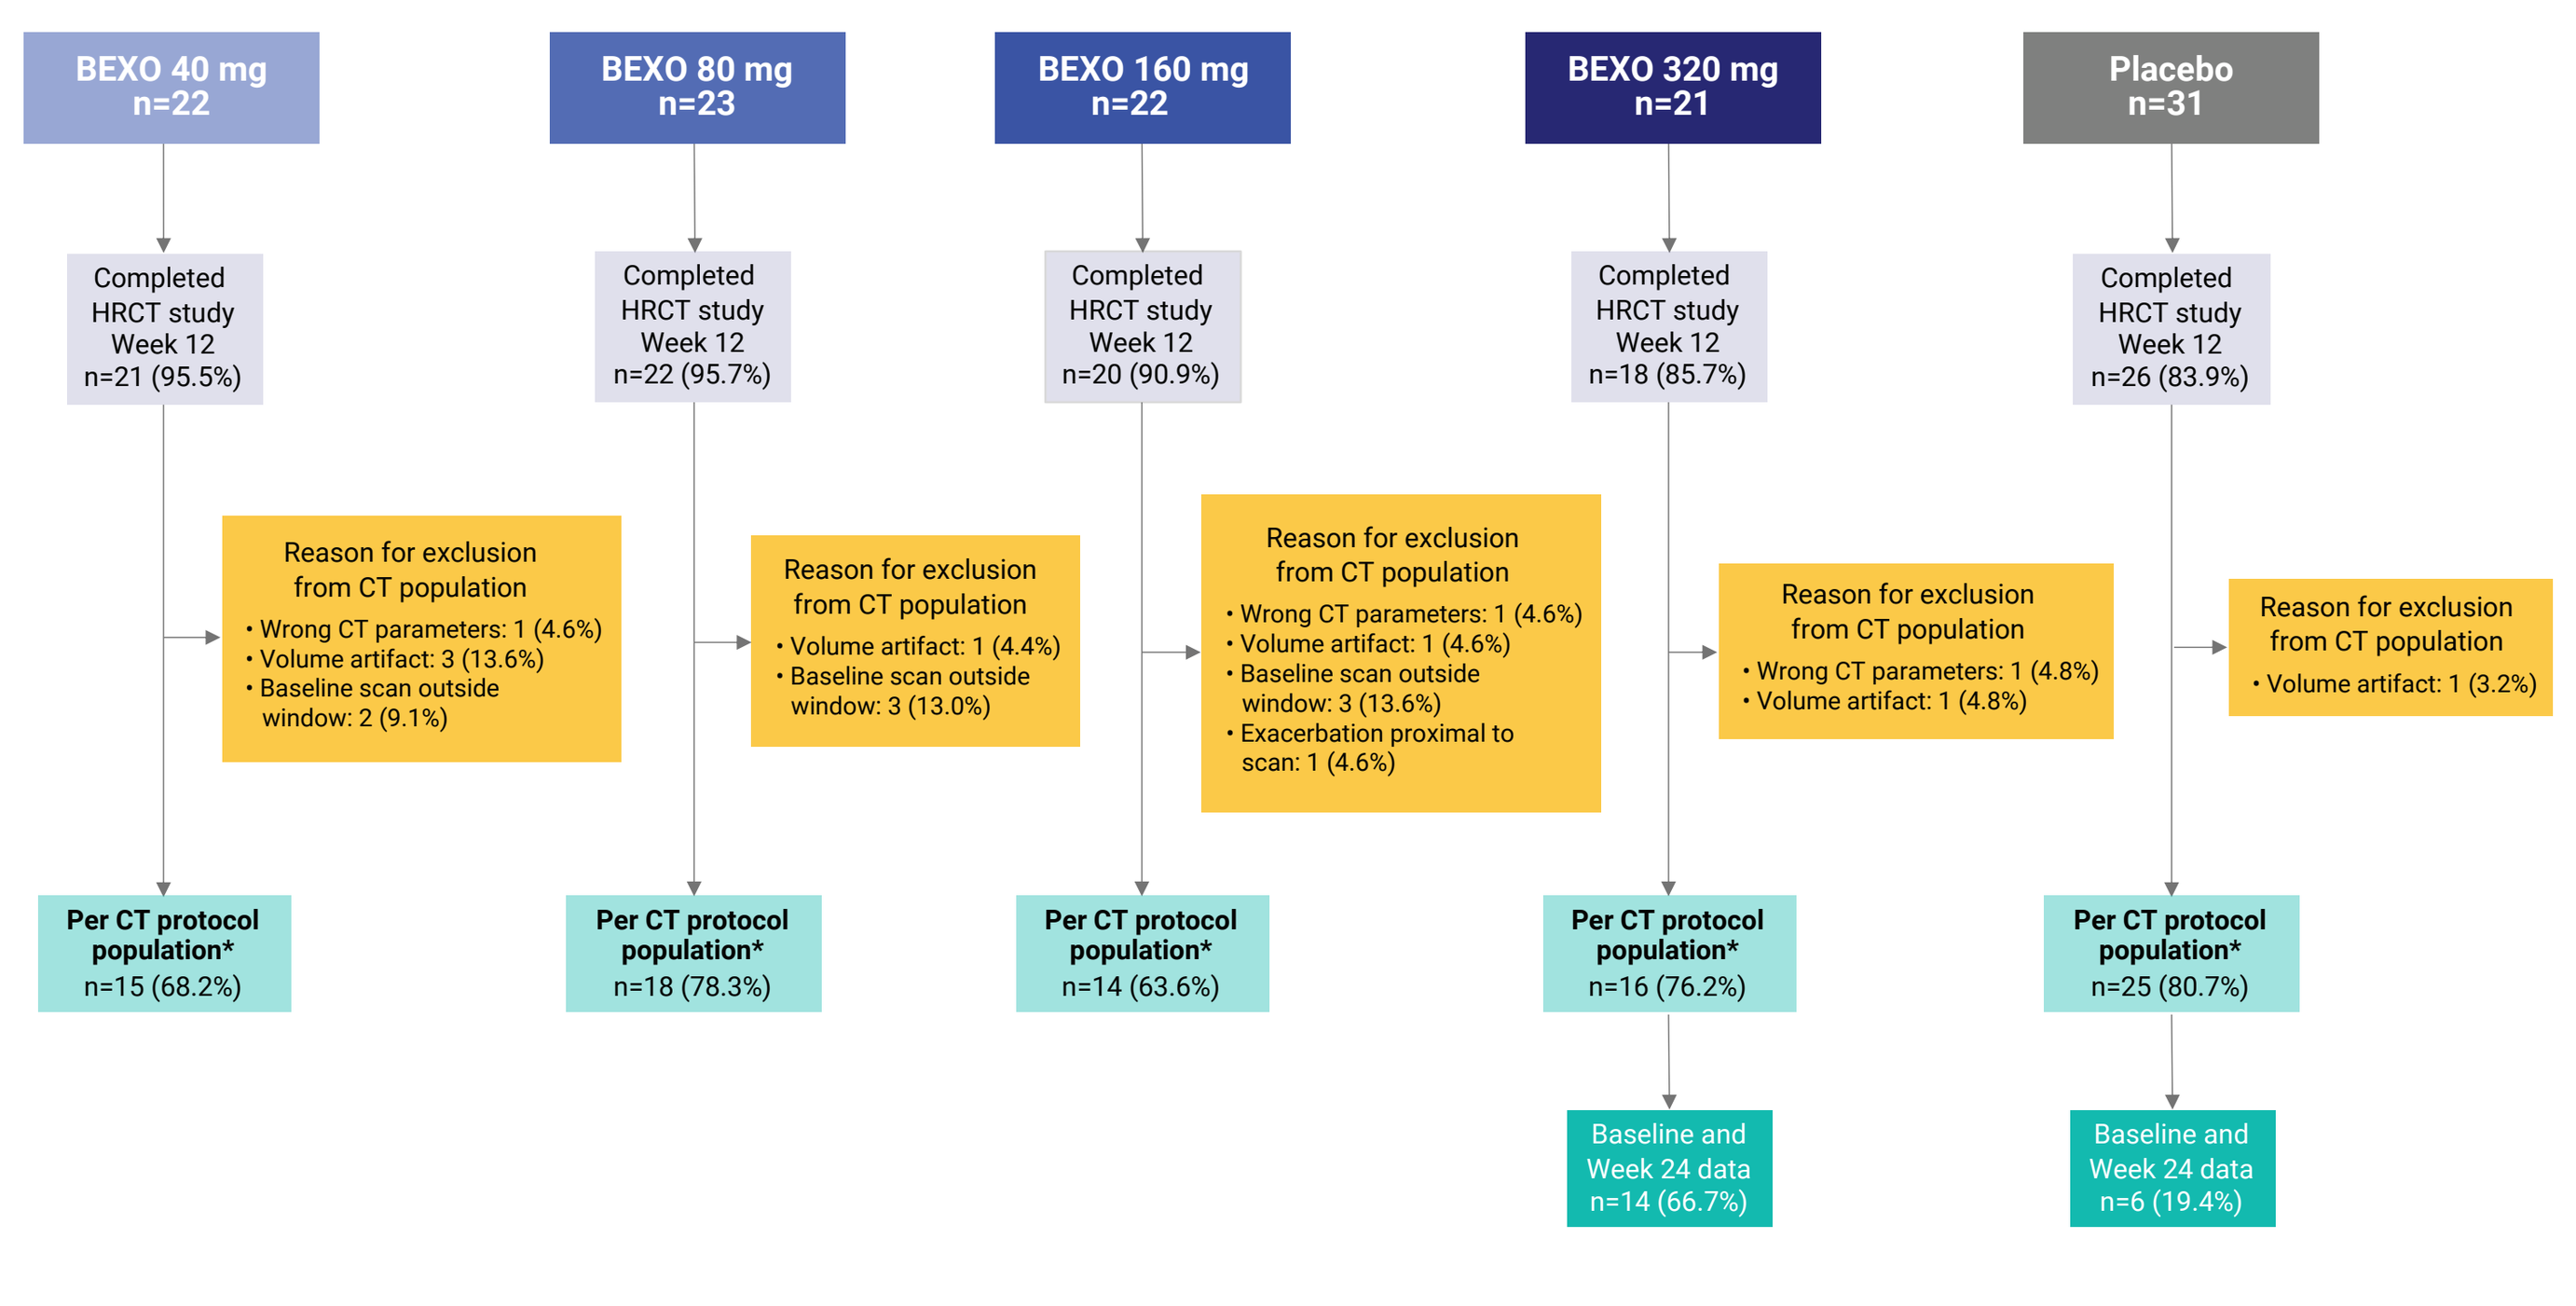
BEXO 40 mg n=22
BEXO 80 mg n=23
BEXO 160 mg n=22
BEXO 320 mg n=21
Placebo n=31
Completed HRCT study Week 12 n=21 (95.5%)
Completed HRCT study Week 12 n=22 (95.7%)
Completed HRCT study Week 12 n=20 (90.9%)
Completed HRCT study Week 12 n=18 (85.7%)
Completed HRCT study Week 12 n=26 (83.9%)
Reason for exclusion from CT population • Wrong CT parameters: 1 (4.6%) • Volume artifact: 3 (13.6%) • Baseline scan outside window: 2 (9.1%)
Reason for exclusion from CT population • Volume artifact: 1 (4.4%) • Baseline scan outside window: 3 (13.0%)
Reason for exclusion from CT population • Wrong CT parameters: 1 (4.6%) • Volume artifact: 1 (4.6%) • Baseline scan outside window: 3 (13.6%) • Exacerbation proximal to scan: 1 (4.6%)
Reason for exclusion from CT population • Wrong CT parameters: 1 (4.8%) • Volume artifact: 1 (4.8%)
Reason for exclusion from CT population • Volume artifact: 1 (3.2%)
Per CT protocol population* n=15 (68.2%)
Per CT protocol population* n=18 (78.3%)
Per CT protocol population* n=14 (63.6%)
Per CT protocol population* n=16 (76.2%)
Per CT protocol population* n=25 (80.7%)
Baseline and Week 24 data n=14 (66.7%)
Baseline and Week 24 data n=6 (19.4%)

BEXO, bexotegrast; HRCT, high-resolution computed tomography.
*Includes participants with both baseline and Week 12 HRCT scans.

- Mean (SD) age was 70.7 (6.9), 73.8 (5.1), 71.4 (6.8), 70.8 (7.7) and 71.8 (6.1) years in the bexotegrast 40 mg, 80 mg, 160 mg, 320 mg and placebo treatment groups, respectively (**Table 2**)
 - Participants in the bexotegrast 320 mg and placebo treatment groups reported longer mean (SD) durations of IPF (38.5 [29.1] vs 35.0 [21.3] months, respectively) and background therapy with nintedanib or pirfenidone (27.1 [21.9] vs 23.4 [17.8] months, respectively) compared with the other bexotegrast treatment groups
 - The majority of participants in all treatment groups were receiving background nintedanib or pirfenidone therapy

Table 2. Demographics and Baseline Characteristics

Parameter	Bexotegrast 40 mg (n=16)	Bexotegrast 80 mg (n=19)	Bexotegrast 160 mg (n=16)	Bexotegrast 320 mg (n=19)	Placebo (n=30)
Age, mean (SD), years	70.7 (6.9)	73.8 (5.1)	71.4 (6.8)	70.8 (7.7)	71.8 (6.1)
Male, n (%)	12 (75.0)	16 (84.2)	10 (62.5)	19 (100)	26 (86.7)
Race, n (%)					
White	16 (100)	17 (89.5)	16 (100)	18 (94.7)	29 (96.7)
Asian	0	1 (5.3)	0	0	1 (3.3)
Not reported/unknown	0	1 (5.3)	0	0	0
Other	0	0	0	1 (5.3)	0
Ethnicity, n (%)					
Hispanic or Latino	1 (6.3)	0	0	0	0
Not Hispanic or Latino	15 (93.8)	19 (100)	16 (100)	19 (100)	30 (100)
BMI, mean (SD), kg/m^2	27.4 (4.6)	27.8 (5.7)	29.4 (4.7)	28.4 (4.1)	27.3 (2.6)
Duration of IPF, mean (SD), months	24.3 (13.0)	27.1 (17.8)	26.3 (11.6)	38.5 (29.1)	35.0 (21.3)
Duration of background therapy, mean (SD), months	20.9 (12.7)	20.8 (11.7)	18.4 (9.7)	27.1 (21.9)	23.4 (17.8)
Background nintedanib or pirfenidone therapy, n (%)	11 (68.8)	15 (78.9)	13 (81.3)	15 (78.9)	23 (76.7)
Nintedanib	7 (43.8)	7 (36.8)	4 (25.0)	8 (42.1)	13 (43.3)
Pirfenidone	4 (25.0)	8 (42.1)	9 (56.3)	7 (36.8)	10 (33.3)

BMI, body mass index; IPF, idiopathic pulmonary fibrosis.

Change in QLF From Baseline

- The percent change in QLF extent decreased in a dose-dependent manner from baseline to Week 12 (**Figure 3A**)
 - At Week 12, mean (SE) change from baseline in QLF extent was 3.2% (1.2), 0.7% (1.0), 0% (1.1), 0.2% (0.7) and 1.5% (1.0) in the bexotegrast 40 mg, 80 mg, 160 mg and 320 mg and placebo treatment groups, respectively
 - Participants randomized to bexotegrast 160 mg or 320 mg demonstrated limited to no progression of fibrosis by Week 12
- At Week 24, mean (SE) change from baseline was 1.3% (0.7) with bexotegrast 320 mg vs 3.7% (1.8) with placebo (**Figure 3B**)

Figure 3. Change From Baseline in QLF Extent at (**A**) Week 12 and (**B**) Week 24

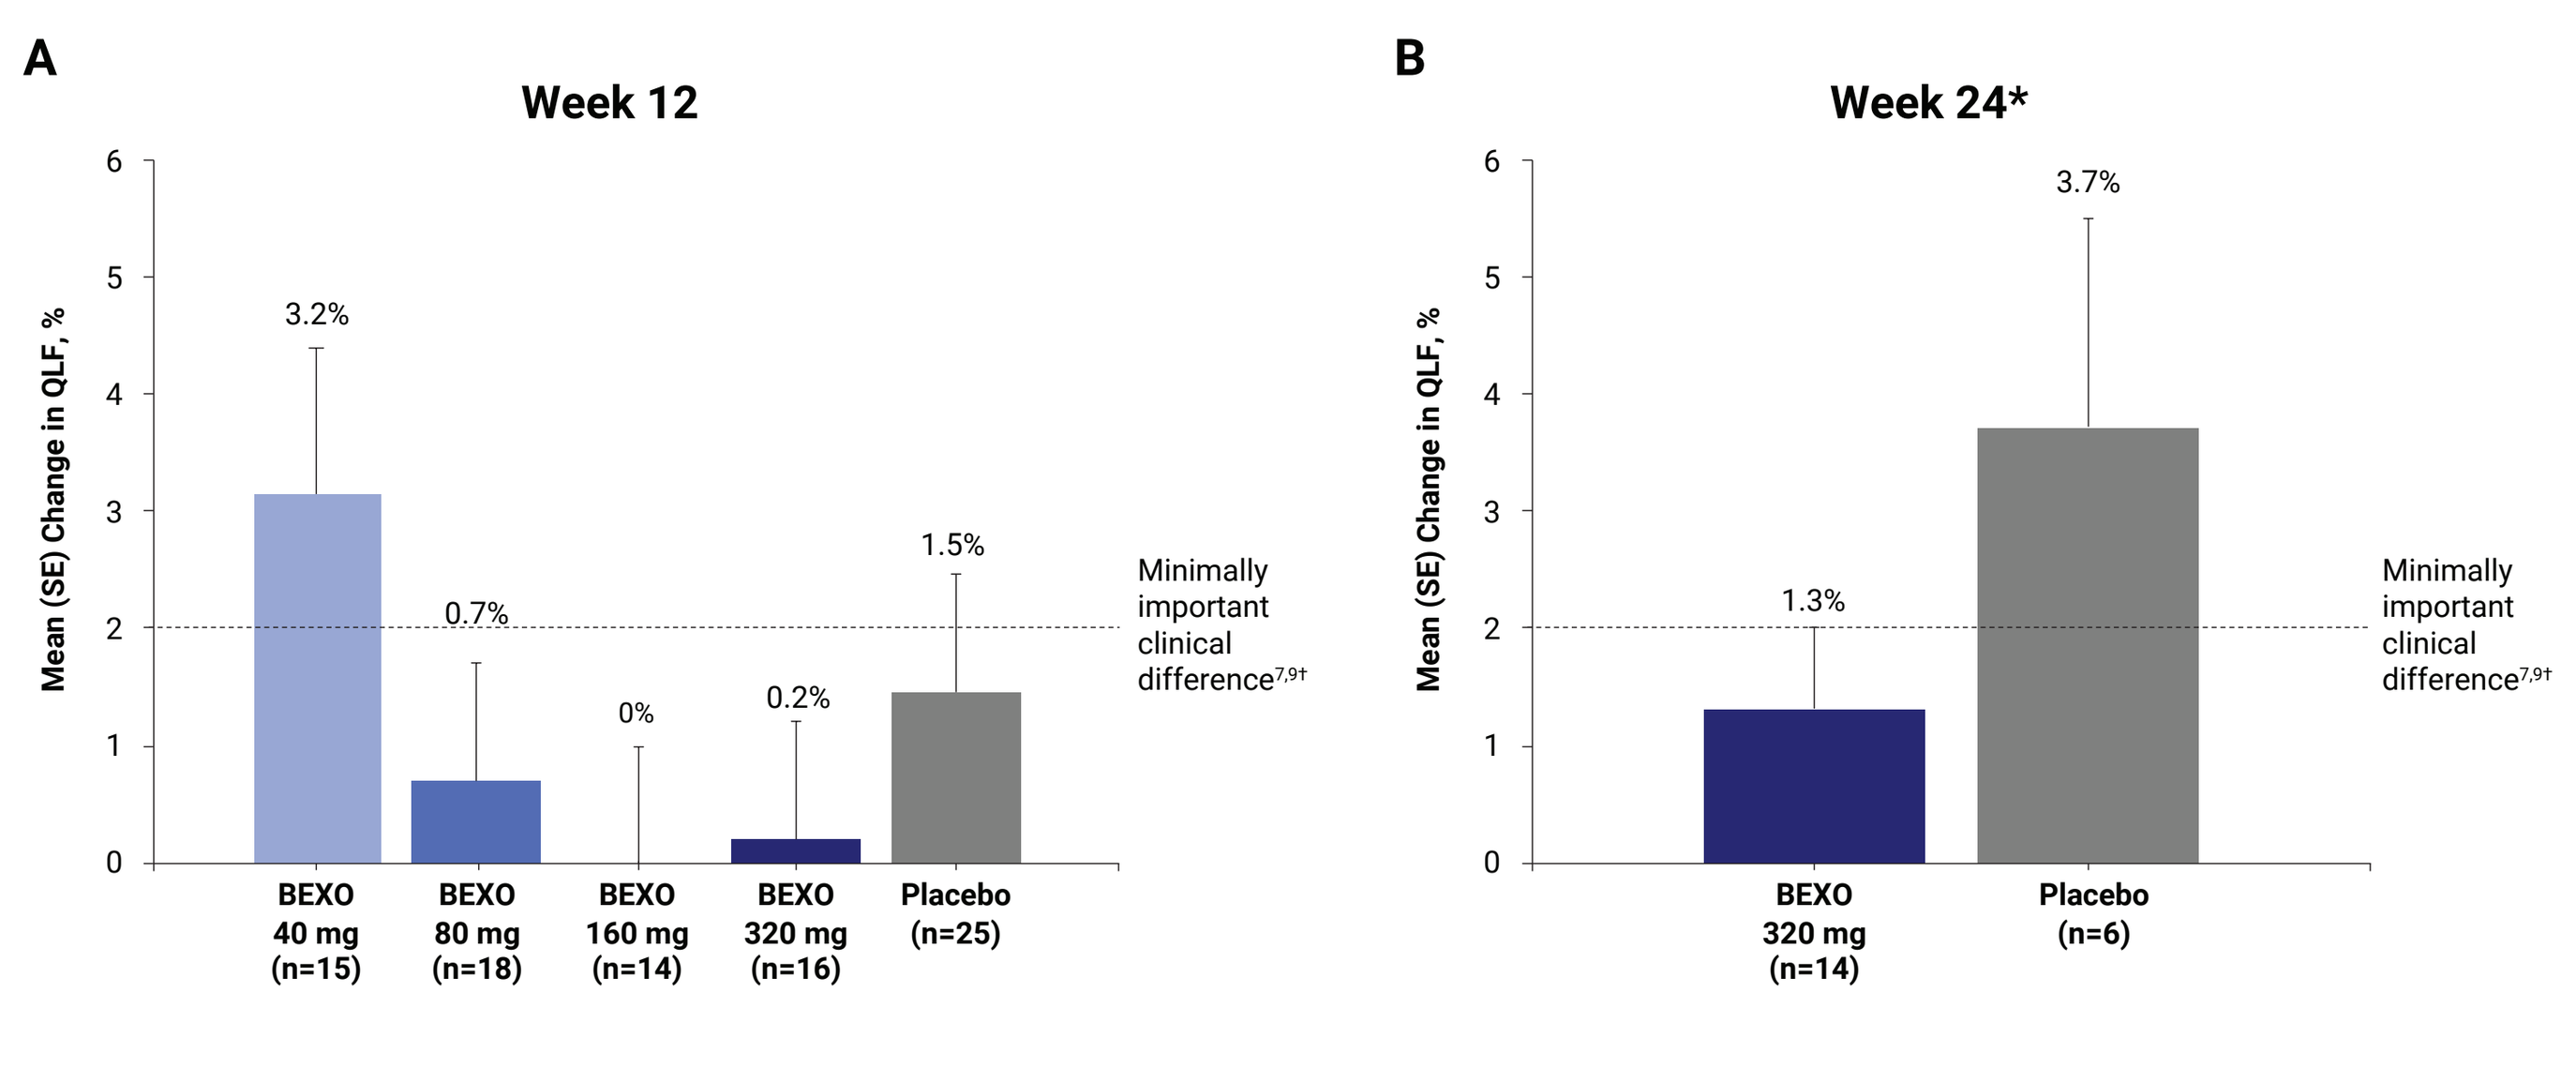
A
Week 12
Mean (SE) Change in QLF, %
3.2%
0.7%
0%
0.2%
1.5%
Minimally important clinical difference[7,9†]
BEXO 40 mg (n=15)
BEXO 80 mg (n=18)
BEXO 160 mg (n=14)
BEXO 320 mg (n=16)
Placebo (n=25)
B
Week 24*
1.3%
3.7%
BEXO 320 mg (n=14)
Placebo (n=6)

BEXO, bexotegrast; QLF, quantitative lung fibrosis.
*Week 24 is cohort 3 only (320 mg or placebo for up to 48 weeks).
†Stable QLF is defined as a range of ≥−2% to ≤2% change from baseline; an increase of >2% is considered worsening fibrosis.[7,9]

- At Week 24, 71.4% of participants treated with bexotegrast 320 mg demonstrated stable or improved QLF extent[7,10] vs 33.3% of participants who received placebo (**Figure 4**)

Figure 4. Proportion of Participants With Stable or Improved QLF* at Week 24

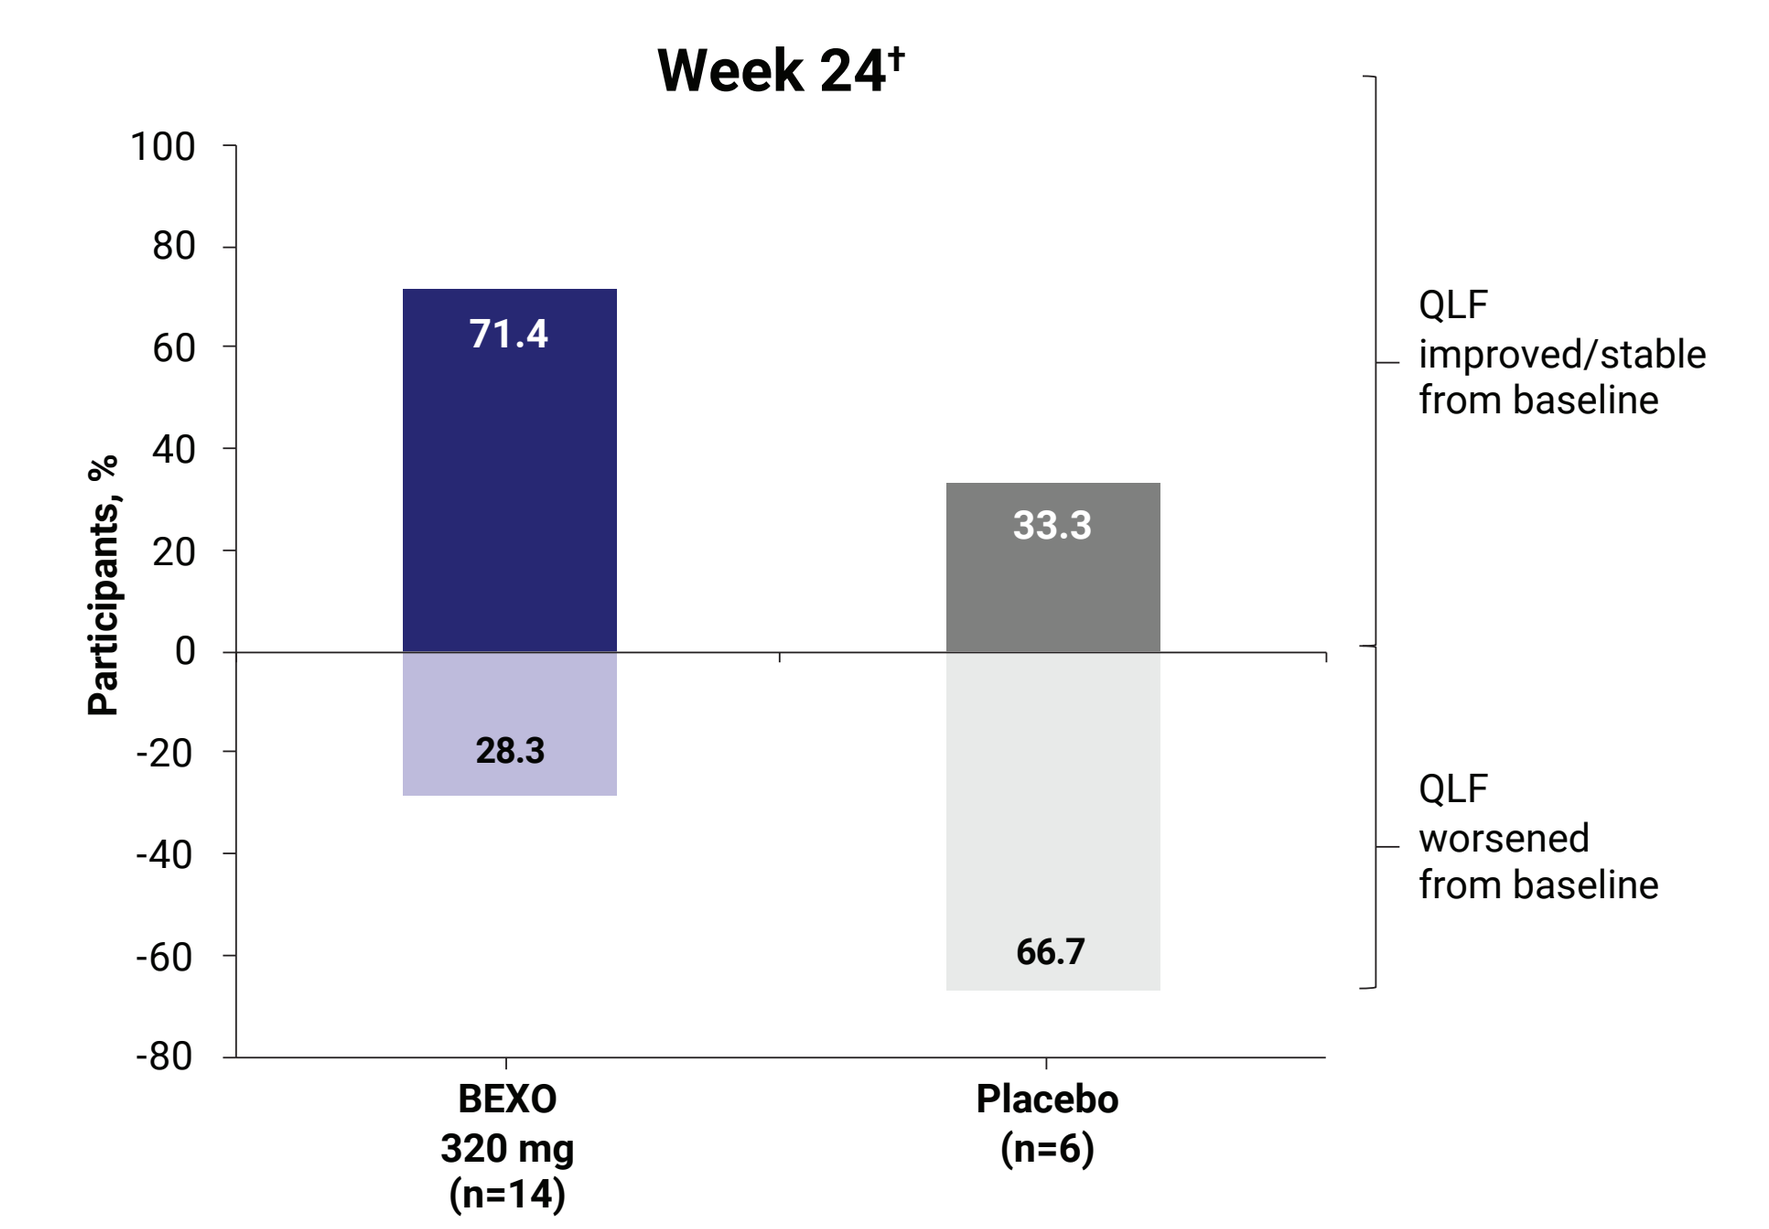
Week 24†
Participants, %
71.4
28.3
33.3
66.7
QLF improved/stable from baseline
QLF worsened from baseline
BEXO 320 mg (n=14)
Placebo (n=6)

BEXO, bexotegrast; QLF, quantitative lung fibrosis.
*Improved QLF is defined as a <2% change from baseline. Stable QLF is defined as a range of ≥−2% to ≤2% change from baseline. Worsened QLF is defined as >−2% change from baseline.[7,9]
†Week 24 is cohort 3 only (320 mg or placebo for up to 48 weeks).

Change in QGG From Baseline

- No notable changes from baseline or differences between bexotegrast and placebo in QGG were identified at Week 12 (**Figure 5A**)
 - At Week 12, mean (SE) percent change from baseline in QGG extent was 1.4% (0.9), −1.1% (1.0), 0% (0.6), −0.2% (0.9) and −0.2% (0.4) in the bexotegrast 40 mg, 80 mg, 160 mg and 320 mg and placebo treatment groups, respectively
- At Week 24, mean (SE) change from baseline was 0.6% (1.0) with bexotegrast 320 mg and 1.6% (1.6) with placebo (**Figure 5B**)
- At Weeks 12 and 24, 75.0% and 64.3% of participants treated with bexotegrast 320 mg demonstrated stable or improved QGG vs 88.0% and 50.0% of participants who received placebo, respectively

Figure 5. Change From Baseline in QGG Extent* at (**A**) Week 12 and (**B**) Week 24

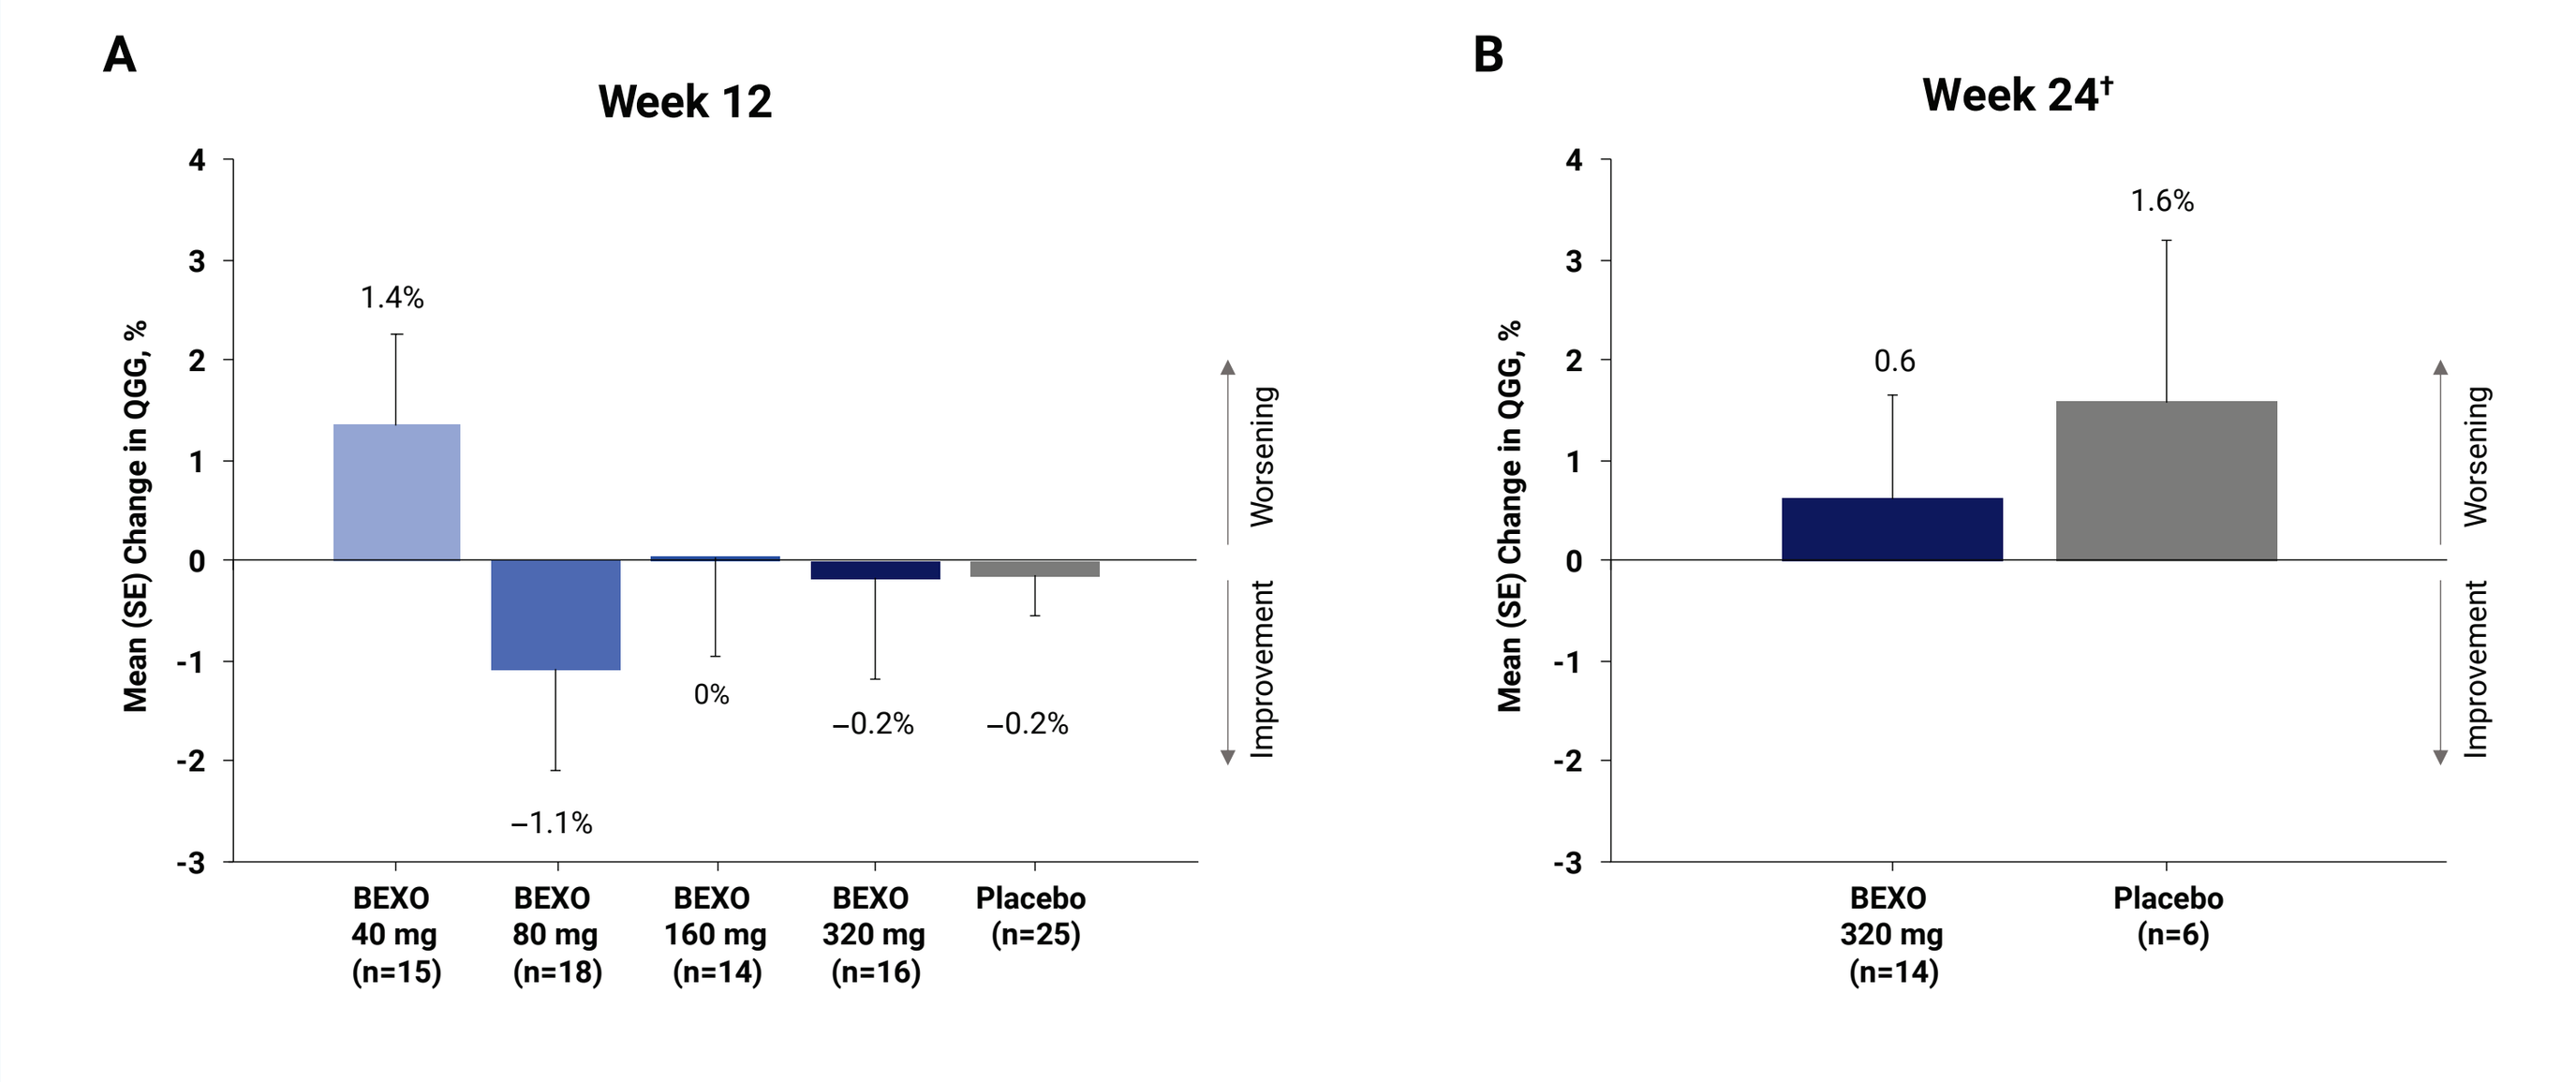
A
Week 12
Mean (SE) Change in QGG, %
1.4%
−1.1%
0%
−0.2%
−0.2%
Worsening
Improvement
BEXO 40 mg (n=15)
BEXO 80 mg (n=18)
BEXO 160 mg (n=14)
BEXO 320 mg (n=16)
Placebo (n=25)
B
Week 24†
0.6
1.6%
BEXO 320 mg (n=14)
Placebo (n=6)

BEXO, bexotegrast; QGG, quantitative ground glass.
*Worsened QGG is defined as a >2% change from baseline.
†Week 24 is cohort 3 only (320 mg or placebo for up to 48 weeks).

LIMITATIONS

- The treatment period in this study was relatively short (≤24 weeks); longer treatment periods will be used to assess the durability of the observed antifibrotic effects
- This was an analysis with a small sample size, which resulted in more variability
- This study was not powered to adequately assess differences in treatment groups; however, it provides evidence for hypothesis generation, to be confirmed in larger and longer studies

CONCLUSIONS

- **Volumetric HCRT scans provide useful metrics to describe fibrotic changes in the lungs of patients with IPF**
- **The potential antifibrotic benefit of bexotegrast, with a majority of participants also receiving background therapy, was observed through the reduced progression of QLF extent vs placebo at Week 24, with no notable increases in QGG in participants with IPF**
- **These results support the need for further studies of bexotegrast for the treatment of IPF; late-stage evaluation, including assessment of QLF, is currently underway in the BEACON-IPF study (NCT06097260)**

References: 1. Lederer DJ, et al. *N Engl J Med*. 2018;379(8):797-798. **2.** Saini G, et al. *Eur Respir J*. 2015;46(2):486-494. **3.** Saito A, et al. *Int J Mol Sci*. 2018;19(8):2460. **4.** Decaris ML, et al. *Respir Res*. 2021;22(1):1-14. **5.** Raghu G, et al. *Am J Respir Crit Care Med*. 2018;198(5):e44-e68. **6.** Rodriguez K, et al. *Sem Respir Crit Care Med*. 2022;43(6):764-779. **7.** Kim GHJ, et al. *Ther Adv Respir Dis*. 2021;15:1-44. **8.** Kim GHJ, et al. *Eur Radiol*. 2020;30(2):726-734. **9.** Kim GJ, et al. *Am J Respir Crit Care Med*. 2022;205:A3426. **10.** Wuyts WA, et al. *Eur Respir J*. 2023;62(67):OA1423.

Presented at the American Thoracic Society International Conference; May 17-22, 2024; San Diego, CA.

Disclosures: JG Goldin was the founder of MedQIA, LLC now Voiant Clinical, LLC; **GJ Kim** is a consultant for Voiant Clinical, LLC; **CN Barnes, M Decaris, S Turner, HE Achneck, ÉA Lefebvre** and **GP Cosgrove** were employed by Pliant Therapeutics Inc. and owned stock at the time of this analysis; **LH Lancaster** received grants from Roche, Pliant Therapeutics Inc., FibroGen, Boehringer Ingelheim, Novartis, Celgene, Galecto, Bristol Myers Squibb, Bridge Biotherapeutics, Horizon, CSL Behring, Nerre; consulting fees from DevPro Biopharma, Pieris, AstraZeneca, Oxygenium, Roche, Bellerophon, Senhwa, Daewoong, Daiichi Sankyo, Nashville Biosciences, Veracyte, United Therapeutics, Structure Therapeutics, Fortress Biotech, Tvardi, SureTech, SynDevRx; honoraria from Boehringer Ingelheim, Genentech, Veracyte, United Therapeutics; has patent pending for Aut02; participated on a data safety monitoring board or advisory board for DevPro Biopharma, Pieris, Oxygenium, Bellerophon, Veracyte, United Therapeutics, Aerami.

Acknowledgments: This study was sponsored by Pliant Therapeutics, Inc. Editorial assistance was provided by Samantha O'Dwyer, PhD, of Nucleus Global and funded by Pliant Therapeutics, Inc. This poster was developed in accordance with Good Publication Practice (GPP 2022) guidelines. Authors had full control of the content and made the final decision on all aspects of this publication.

Contact information: gcosgrove@pliantrx.com

Miscellaneous: Copyright © 2024 Pliant Therapeutics, Inc. | All Rights Reserved